The Bicycle platform: an efficient technology to generate high affinity, high selectivity molecules (*Bicycles®*) with unique drug like properties that are amenable to conjugation

Liuhong Chen†; Rachid Lani†; James Cooke†; Sophie Watcham†; Helen Harrison†; Amy Brown†; Catherine Stace†; Katerine van Rietschoten†; Daniel Teufel†; Silvia Pavan†; Gemma Mudd†; Diane Blakeley†; Spencer Campbell†; Julia Kristensson†; Gavin Bennett†; Michael Skynner†; Gregory Winter ‡

ABSTRACT#

ABSTRACT

- *Bicycles®* are bicyclic peptides constrained *via* trifunctional chemical scaffolds.
- *Bicycles* bind to their targets with high affinity and exquisite selectivity.
- *Bicycles* can deliver any pharmacology to targets that are often considered "undruggable" by conventional small molecules approaches.
- The Bicycle platform uses phage display to rapidly identify and optimise *Bicycle* binders.
- *Bicycles* are being developed to carry and direct cytotoxic agents as *Bicycle Drug Conjugates® (BDC*s) for the treatment of cancers.
- *Bicycles* offer rapid penetration into extravascular space and the ability to bypass liver metabolism by benign renal clearance.
- The Bicycle platform is efficient, flexible and adaptable and lends itself to multiple applications in oncology and beyond.

INTRODUCTION

Bicycles® bind with high affinity & selectivity akin to antibodies due to the peptide being attached at three points to a chemical scaffold. Bi-cyclisation "locks in" conformations that are productive for target binding and reduces the entropy penalty of binding to a target as compared to less constrained peptides.

Binders to targets are identified by phage display – *Bicycles* are fused to the N-terminus of coat protein III and presented on the surface of bacteriophage particles. Neither the presence of the *Bicycle* on protein III nor chemical cyclisation affect the ability of phage to infect and be amplified in *E. coli.*

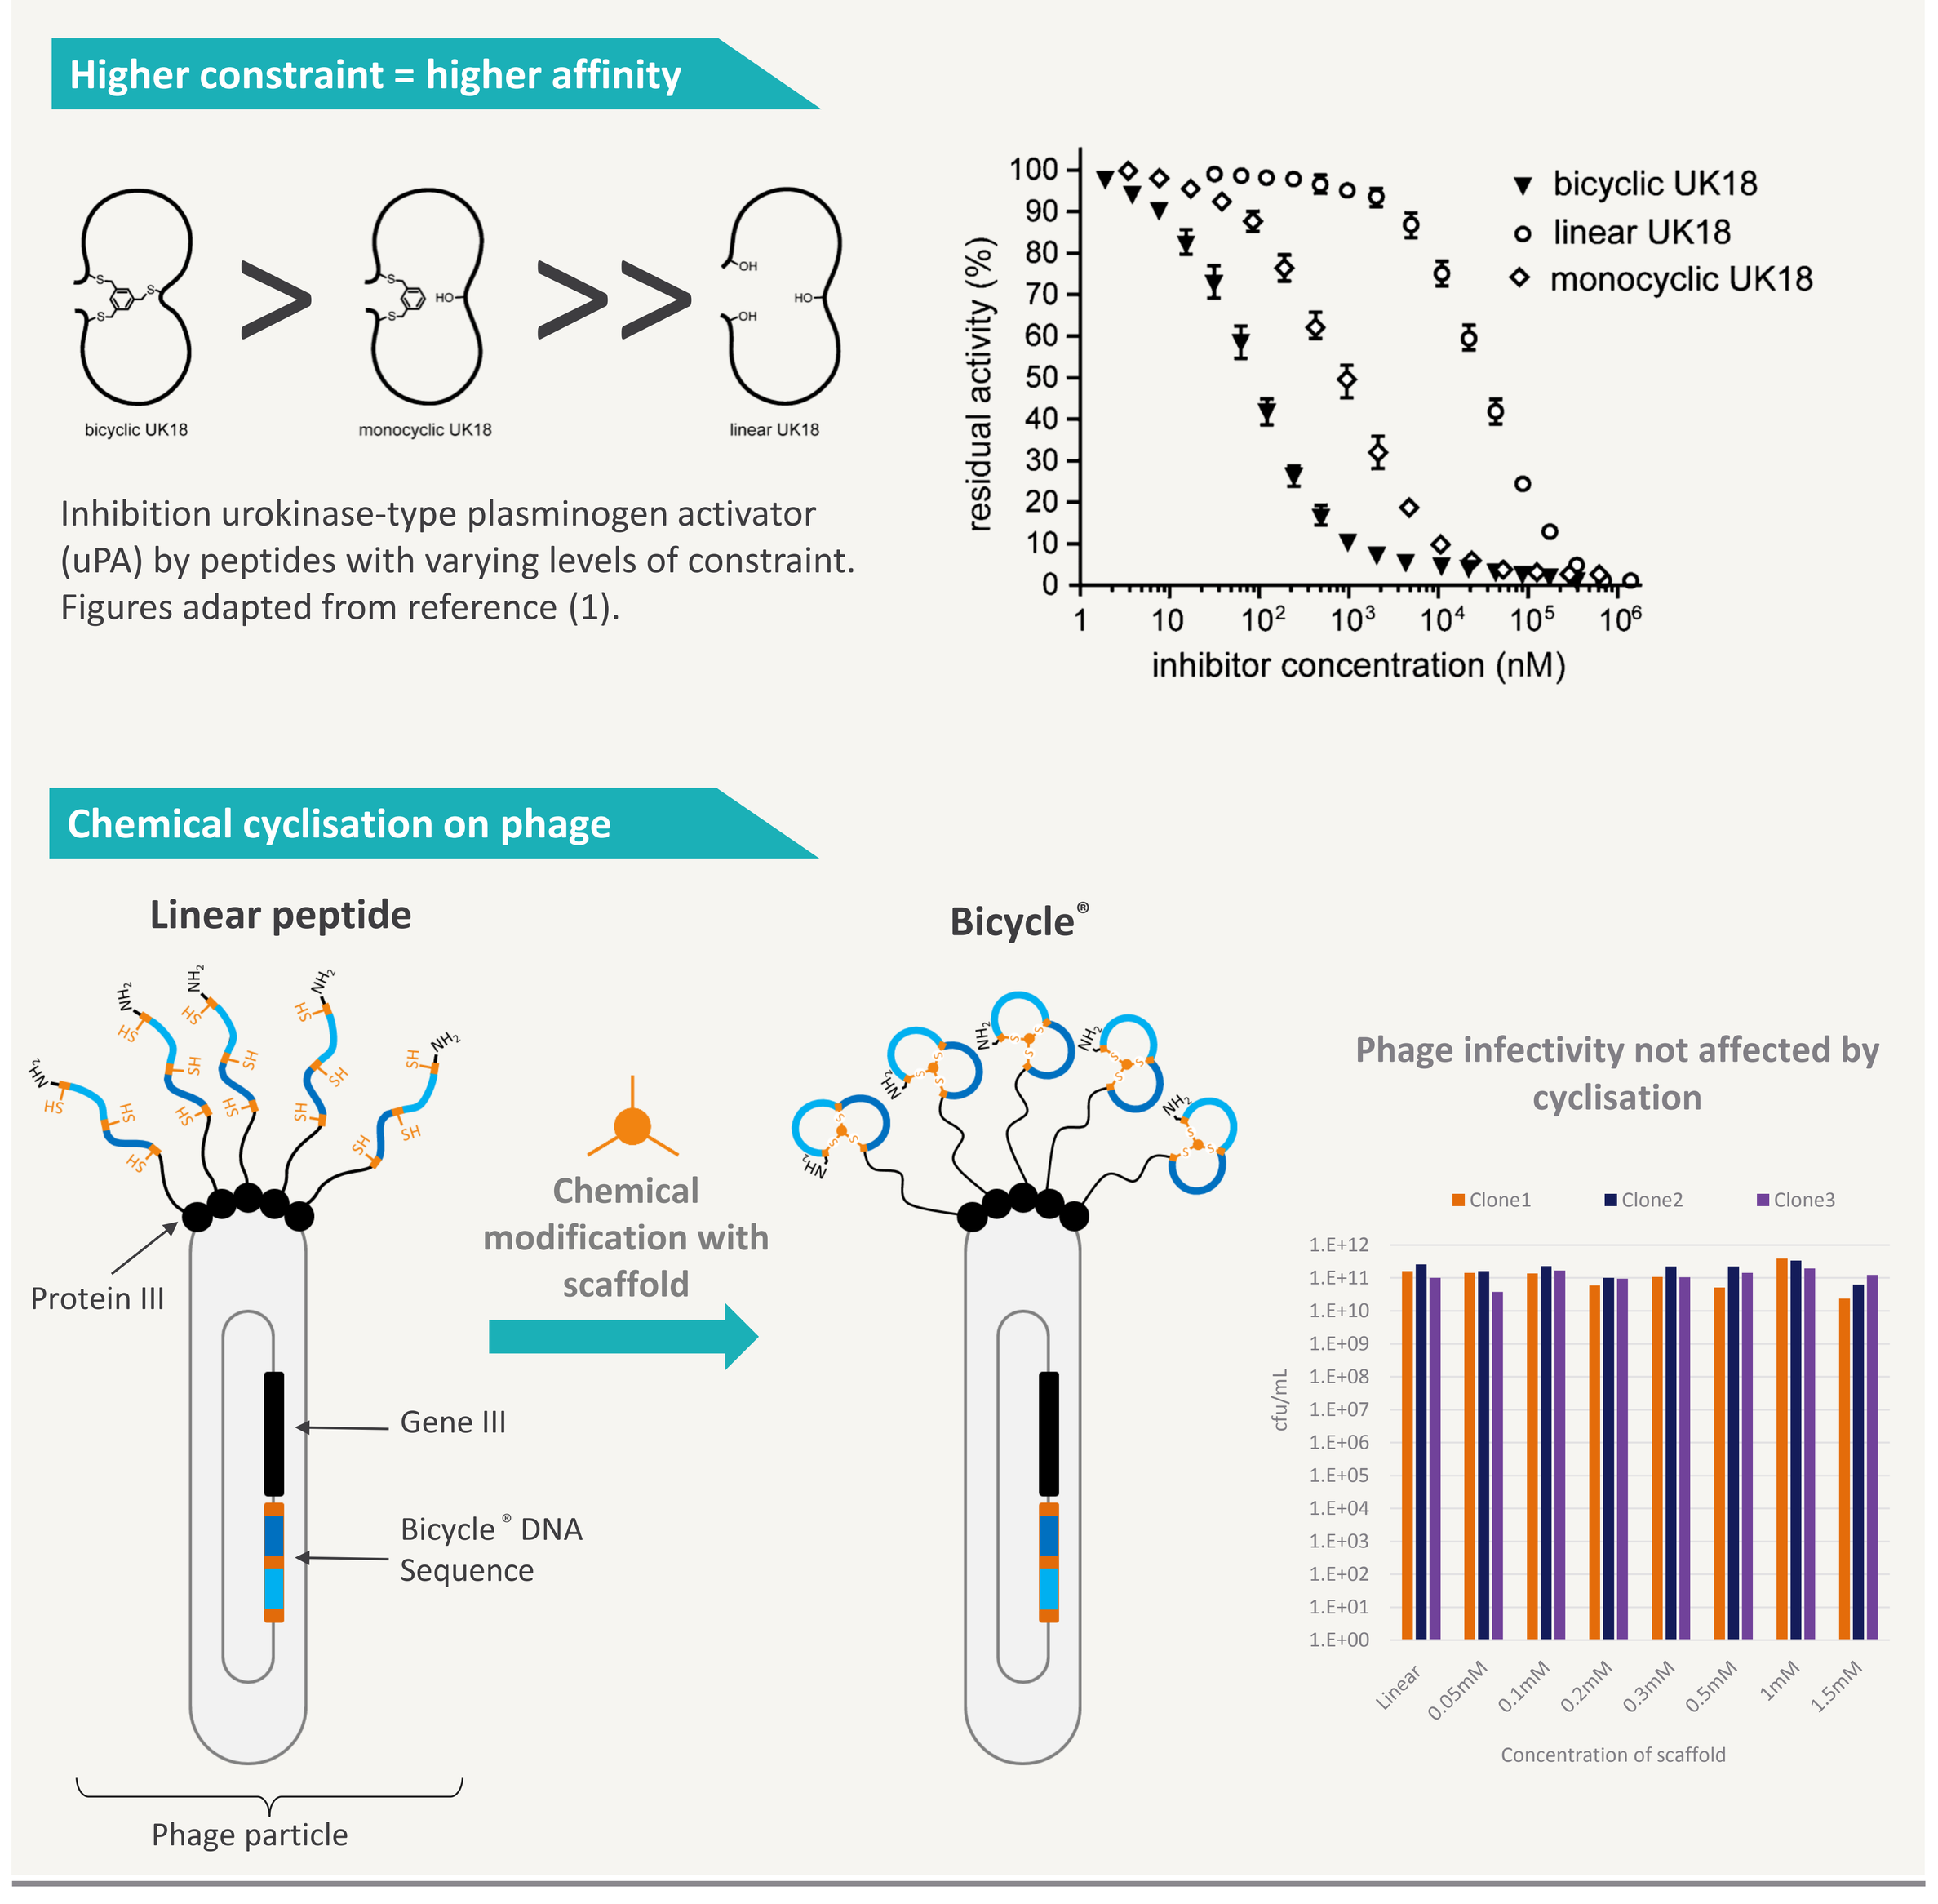

Higher constraint = higher affinity
bicyclic UK18
monocyclic UK18
linear UK18
Inhibition urokinase-type plasminogen activator (uPA) by peptides with varying levels of constraint. Figures adapted from reference (1).
residual activity (%)
inhibitor concentration (nM)
bicyclic UK18
linear UK18
monocyclic UK18
Chemical cyclisation on phage
Linear peptide
Bicycle®
Protein III
Chemical modification with scaffold
Gene III
Bicycle® DNA Sequence
Phage particle
Phage infectivity not affected by cyclisation
Clone1
Clone2
Clone3
cfu/mL
Concentration of scaffold

Figure 1: Bicycle Therapeutics' unique constrained peptide phage display platform (1,2)

The robustness of *Bicycles* and phage allow the use of stringent selection conditions. When combined with a high-throughput phage-based binding assay the best binders can be rapidly identified as opposed to the most abundant. The evolution driven, informed selection process is highly efficient and has low synthetic chemistry requirements.

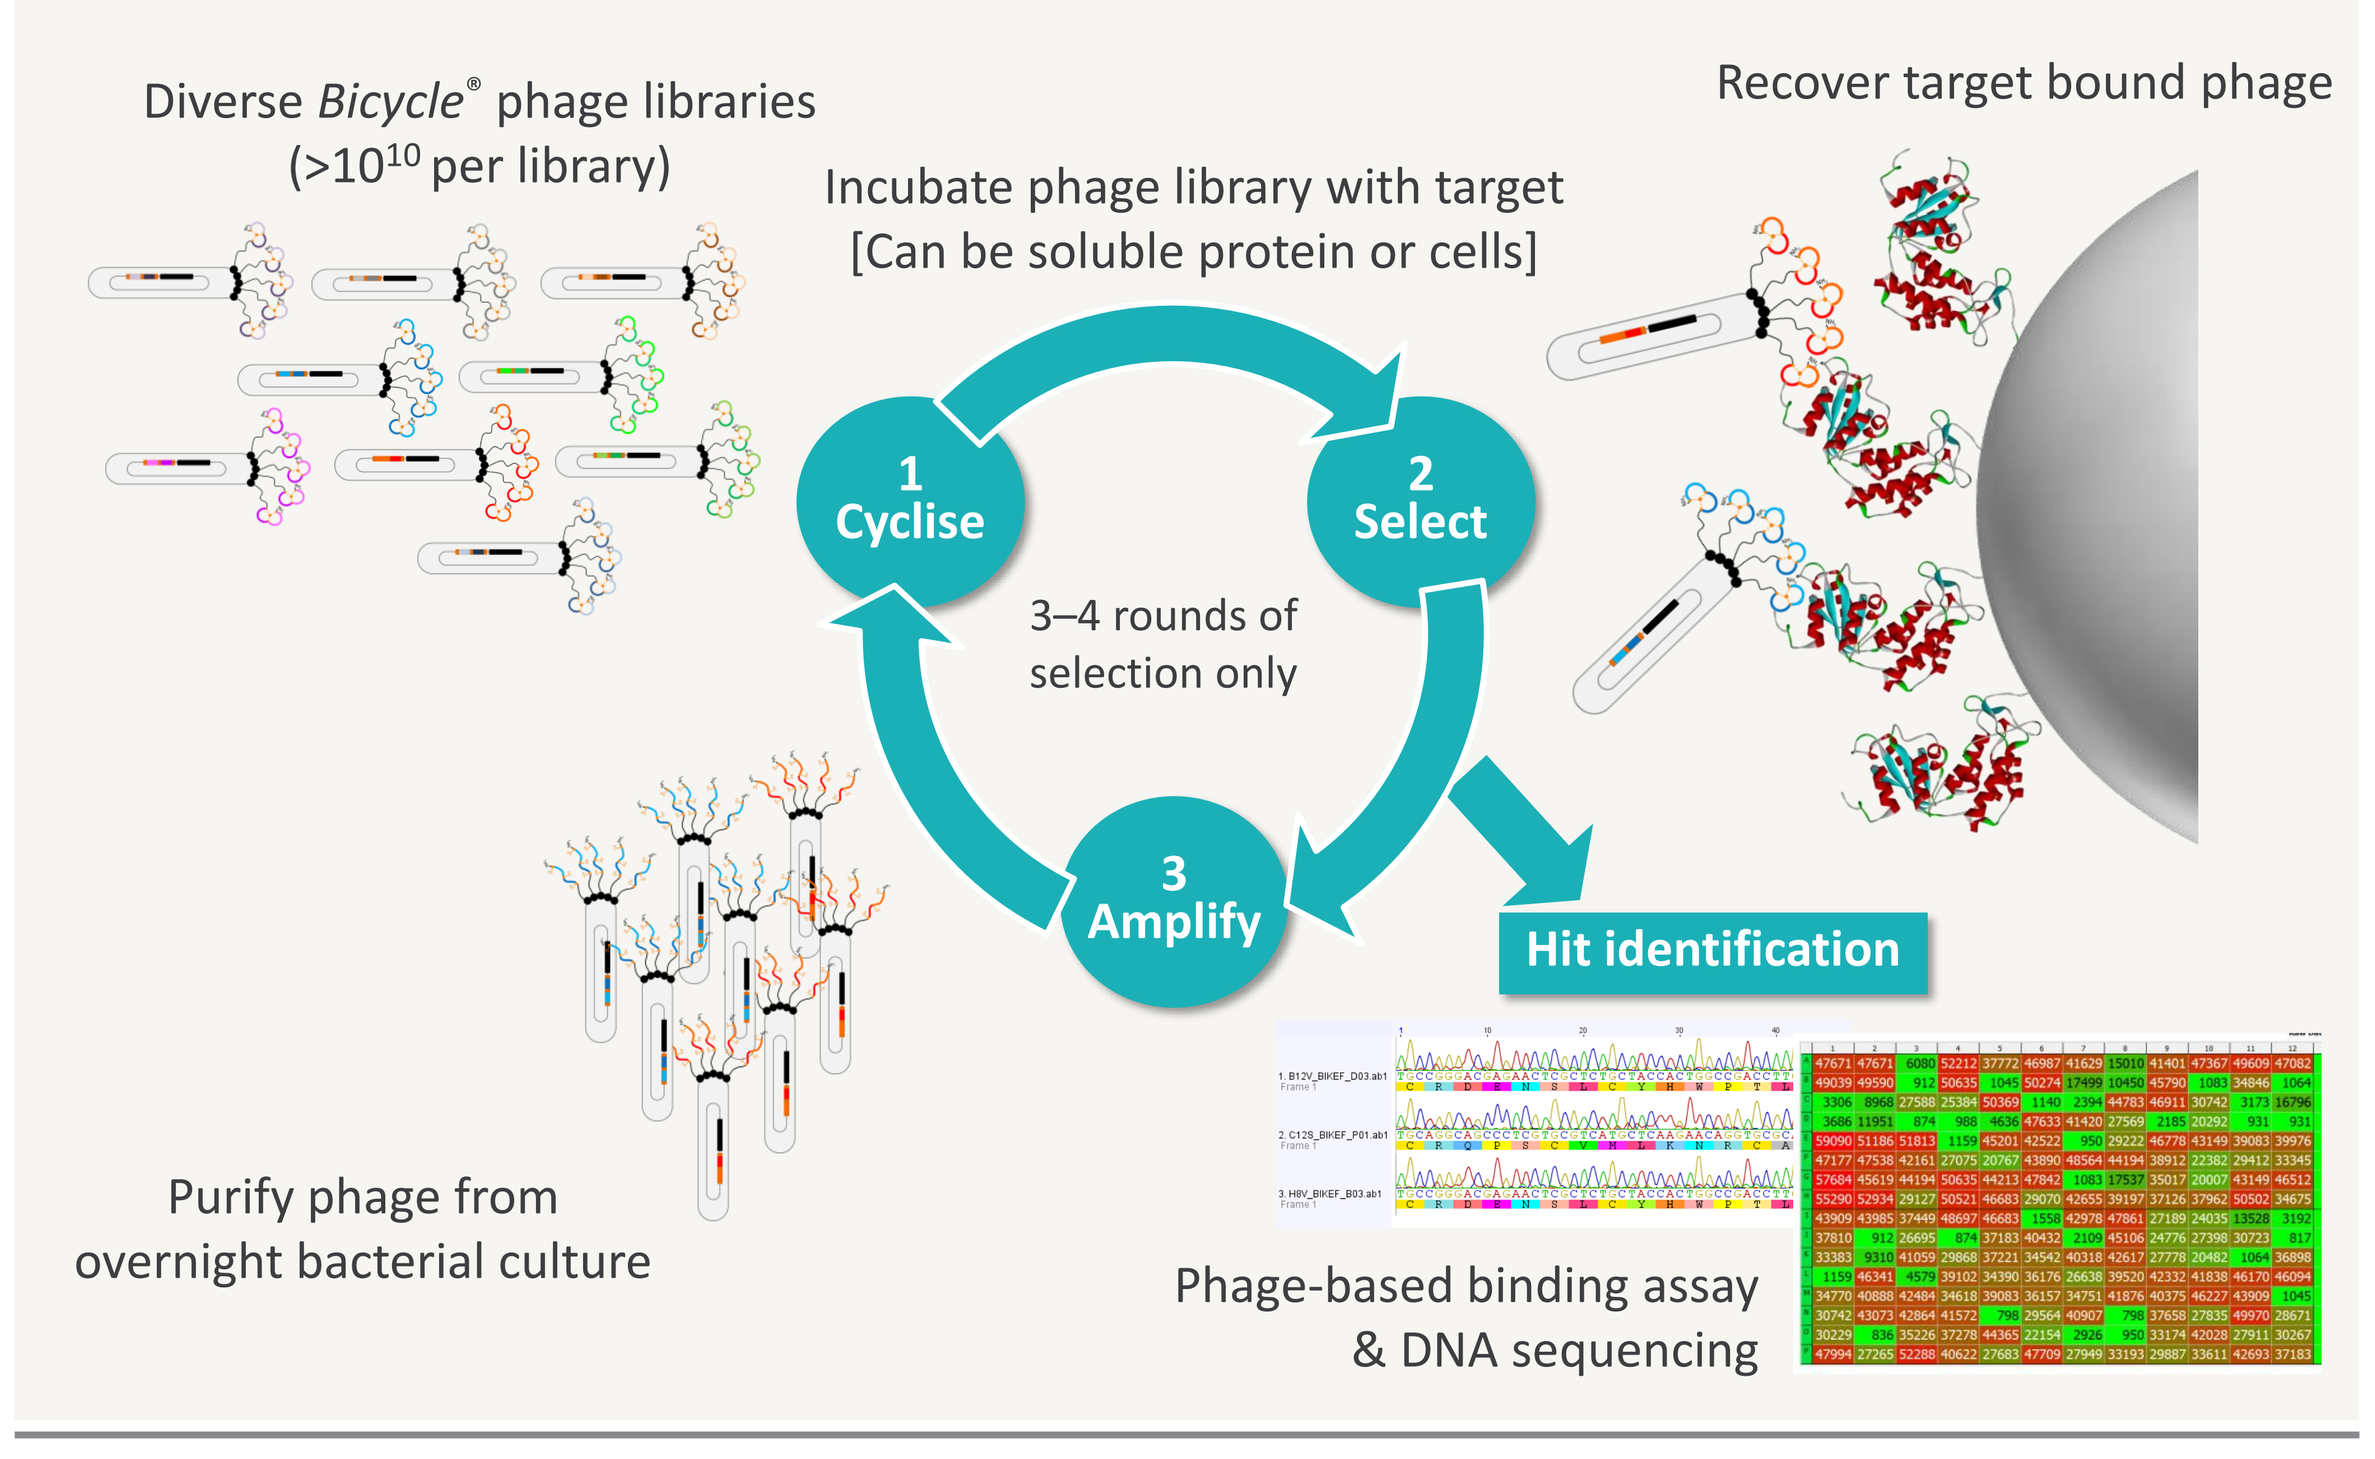

Diverse Bicycle® phage libraries ($>10^{10}$ per library)
Incubate phage library with target [Can be soluble protein or cells]
Recover target bound phage
1 Cyclise
2 Select
3 Amplify
3–4 rounds of selection only
Hit identification
Purify phage from overnight bacterial culture
Phage-based binding assay & DNA sequencing

Figure 2: Phage selection process rapidly identifies high affinity binders

METHODS

Identification of phage *Bicycle* binders – binders were selected from chemically scaffolded "naïve" phage libraries(2) and then optimized using target-specific bespoke phage libraries. **Binding affinity of *Bicycles*** – determined by fluorescence polarization (FP) using either fluorescein-labelled *Bicycles* (direct FP) or by displacement of a fluorescein-labelled ligand (FP competition). **CA IX & CD38 crystallography** – FLAG-tagged human CD38 ecto domain co-crystallised with *Bicycle* binder at 1:1 ratio, structure resolved to 1.7Å; his-tagged CA IX catalytic domain co-crystallised with *Bicycle* inhibitor at ratio of 2 Bicycles:1 CA IX dimer, structure resolved to 2.5Å. **Production of CD38 *Bicycle Drug Conjugate*** – *Bicycle* sequence identified on phage was made using standard solid phase peptide synthesis and conjugated to DM1 (mertansine) via a peptidic spacer and cleavable di-sulfide linker. **Mouse xenograft** – CD38-targeting *BDC* administered *iv* tiw to female CB17-SCID mice bearing MOLP-8 xenografts alongside a vehicle control group (n=3).

HARNESSING THE POWER OF BIOLOGY

Phage libraries produce large sequence diversity. Additional layers of structural and chemical complexity are added by changing the size and (a)symmetry of loops formed by the small molecule scaffold. Together the biological and chemical aspects of the platform come together to yield very high success rates against targets traditionally considered difficult to address with small molecules (e.g. protein-protein interactions) or antibodies (e.g. GPCRs).

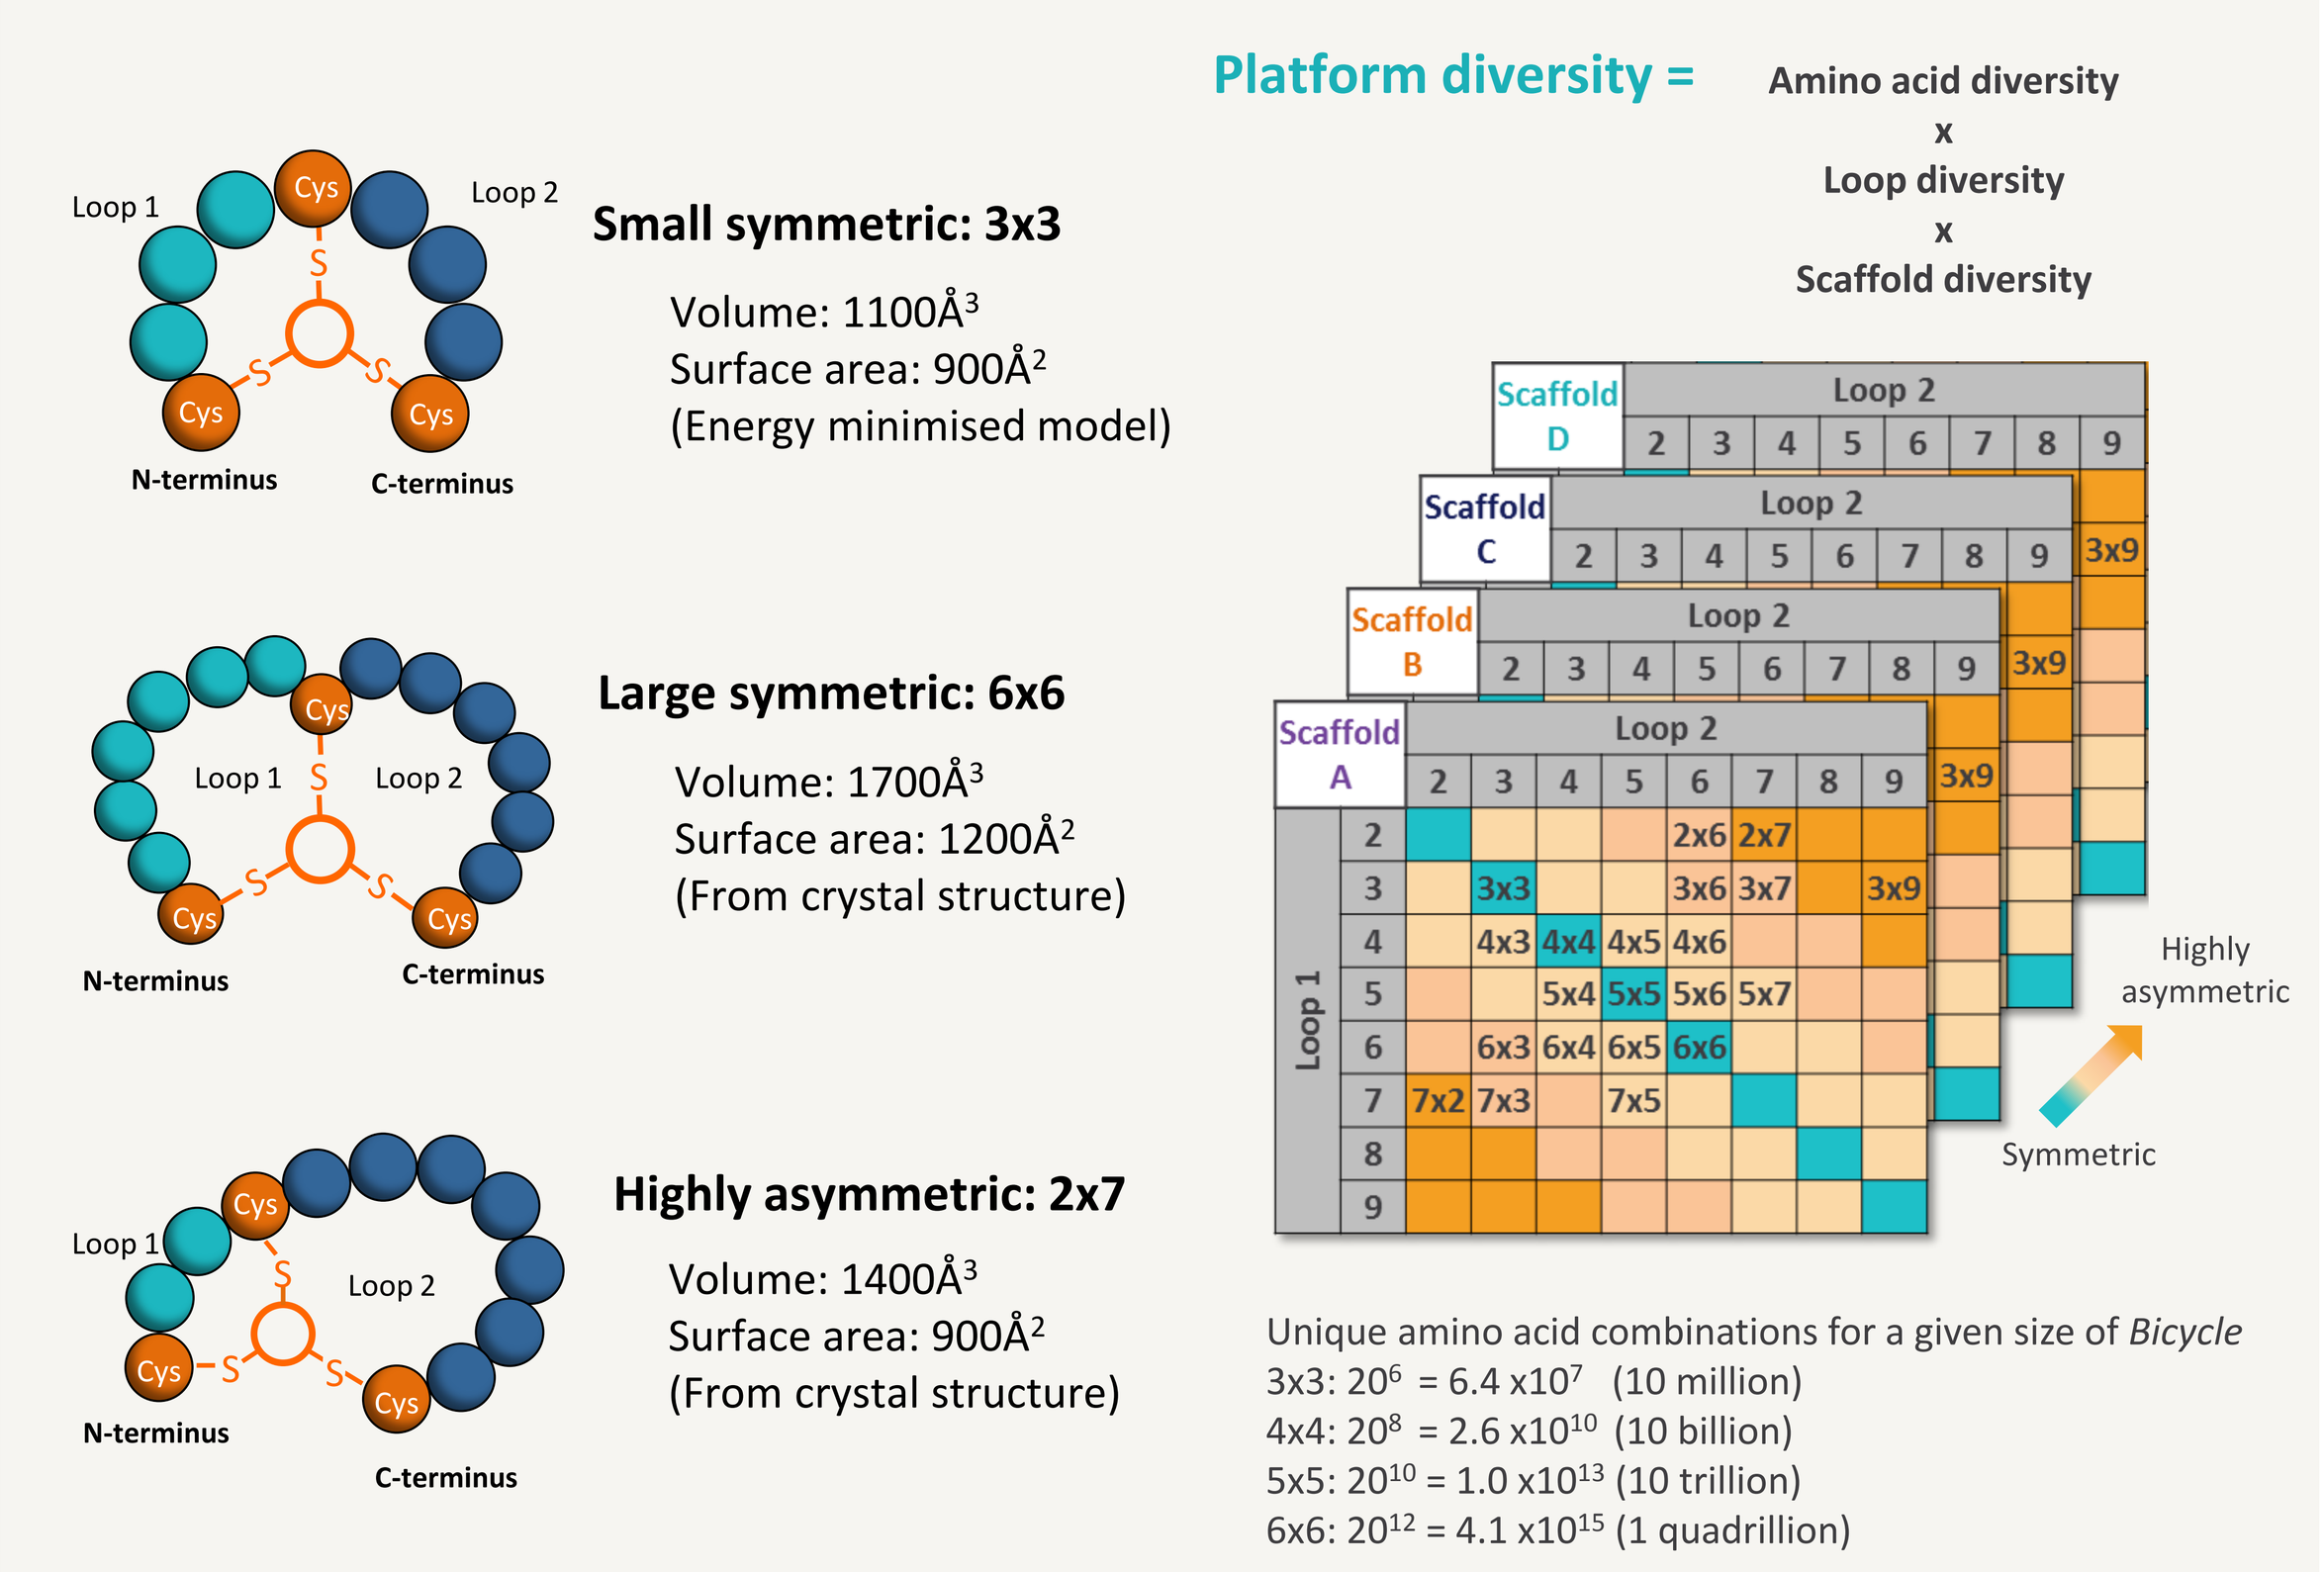

Platform diversity =
Amino acid diversity x Loop diversity x Scaffold diversity
Small symmetric: 3x3
Volume: 1100Å³
Surface area: 900Å²
(Energy minimised model)
Large symmetric: 6x6
Volume: 1700Å³
Surface area: 1200Å²
(From crystal structure)
Highly asymmetric: 2x7
Volume: 1400Å³
Surface area: 900Å²
(From crystal structure)
Loop 1
Loop 2
Cys
N-terminus
C-terminus
Scaffold A
Scaffold B
Scaffold C
Scaffold D
Highly asymmetric
Symmetric
Unique amino acid combinations for a given size of Bicycle
3x3: 20^6 = 6.4 x10^7 (10 million)
4x4: 20^8 = 2.6 x10^{10} (10 billion)
5x5: 20^{10} = 1.0 x10^{13} (10 trillion)
6x6: 20^{12} = 4.1 x10^{15} (1 quadrillion)

Some of the target classes tractable to the Bicycle platform	
Matrix metalloproteinases	Growth factors
Receptor tyrosine kinase	Metalloenzymes
Enzymes	Ig-like domain proteins
Integrins	Coagulation cascade enzymes
Serine proteases	Serum proteins
Cysteine proteases	TNF receptor superfamily members
GPCRs	Glycoproteins
Cytokines	Calcium signalling proteins
Chemokines	Immune-oncology targets
Cancer antigens	Interleukins
Interleukin receptors	Other proteases

Figure 3: Deep coverage of large chemical space translates to high success rate against even difficult protein target classes

AFFINITY & SELECTIVITY OF AN ANTIBODY

Bicycles have large molecular footprints enabling them to show profound selectivity between closely related proteins.

Carbonic anhydrases (CAs) are a large family of highly-homologous metallo-enzymes. Small molecules drugs against carbonic anhydrases have struggled to generate much selectivity between them.

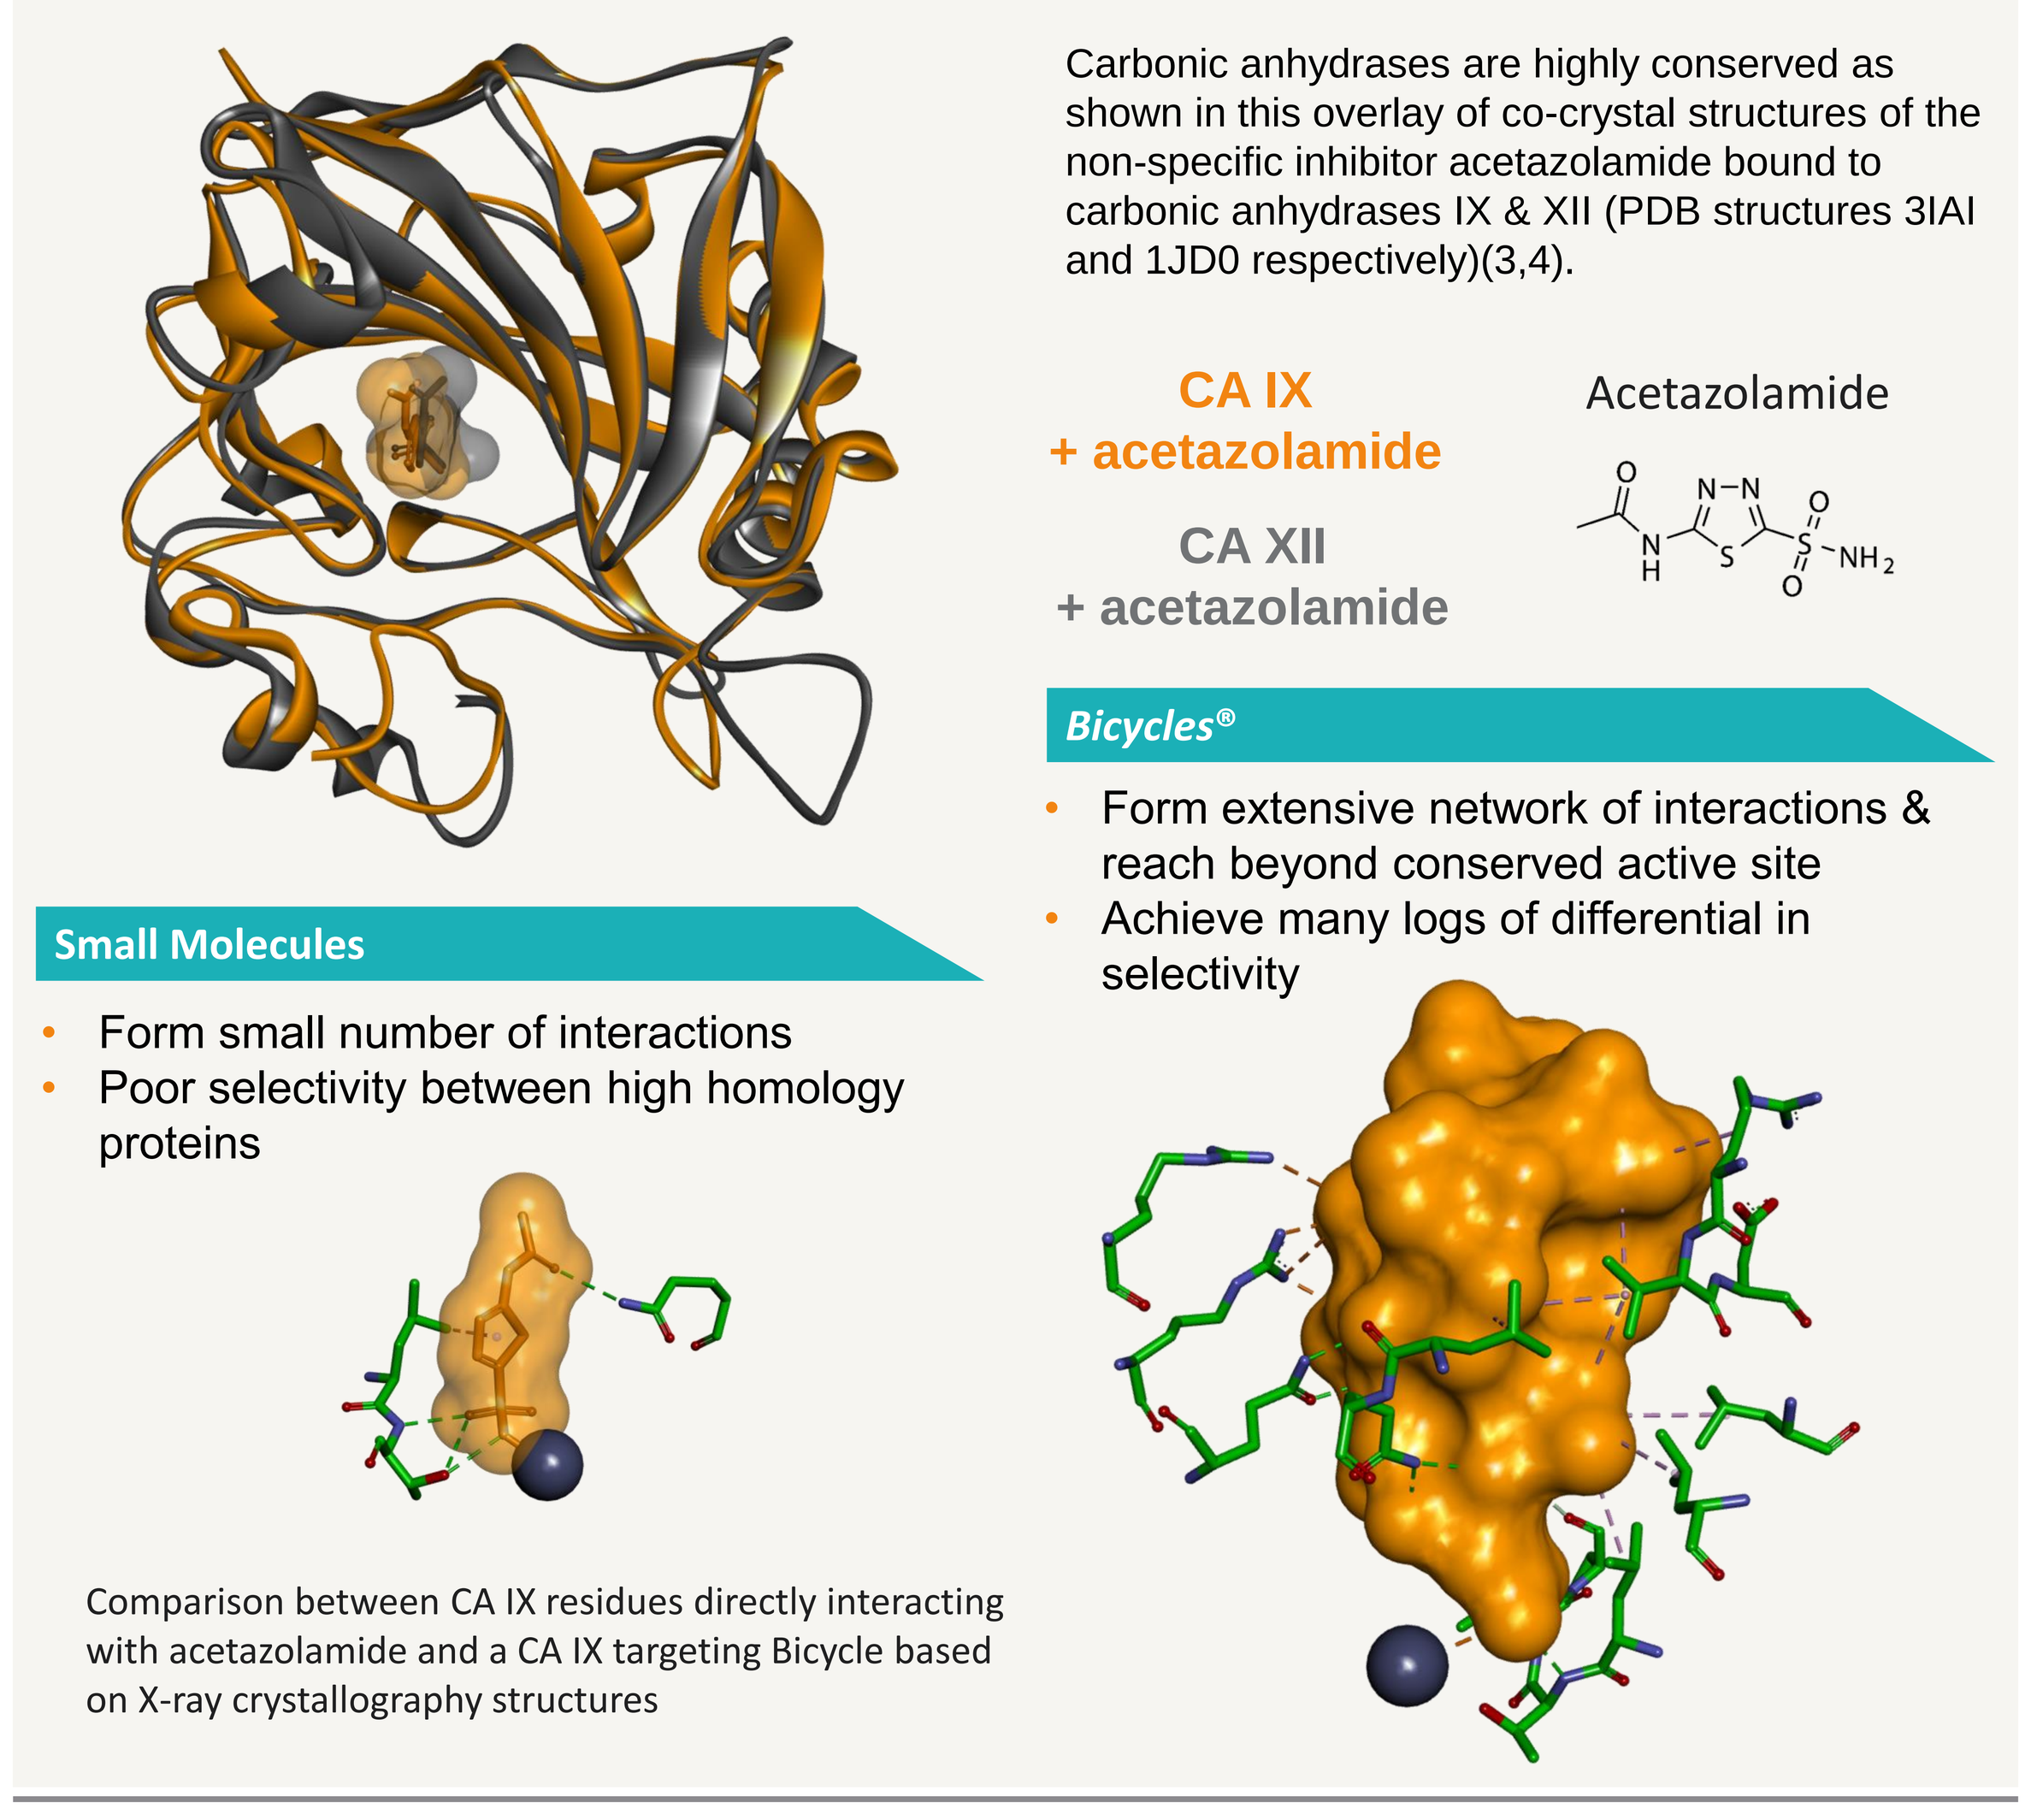

Carbonic anhydrases are highly conserved as shown in this overlay of co-crystal structures of the non-specific inhibitor acetazolamide bound to carbonic anhydrases IX & XII (PDB structures 3IAI and 1JD0 respectively)(3,4).
CA IX + acetazolamide
CA XII + acetazolamide
Acetazolamide
Bicycles®
Form extensive network of interactions & reach beyond conserved active site
Achieve many logs of differential in selectivity
Small Molecules
Form small number of interactions
Poor selectivity between high homology proteins
Comparison between CA IX residues directly interacting with acetazolamide and a CA IX targeting Bicycle based on X-ray crystallography structures

Figure 4: *Bicycle* inhibitor (solid orange surface) showing many more interactions to CA IX (green sticks) compared, a non-selective carbonic anhydrase inhibitor, acetazolamide (transparent orange surface & sticks)(3); catalytic zinc ion shown as grey sphere.

Carbonic anhydrase	*Bicycle* affinity K_d/nM	Acetazolamide affinity K_i/nM(5)	Residues identical vs CA IX active site	Residues identical vs CA IX "pocket"
CA IX	**3**	25	8/8	28/28
CA IV	**>2000**	74	8/8	24/28
CA VI	**>2000**	11	8/8	25/28
CA XII	**>2000**	6	8/8	26/28
CA XIV	**>2000**	41	8/8	26/28

Table 1: *Bicycle* shows no measurable binding (direct FP assay) up to 2µM against CAs other than CA IX (>666 fold selectivity). Acetazolamide on the other hand only shows 4–12 fold selectivity. This is understandable as the small number of residues that define the CA active site where acetazolamide binds are identical in sequence across the family members. It is by binding beyond these conserved residues that the *Bicycle* is able to discriminate between the different CAs.

PLUG AND PLAY VERSATILITY BUILT-IN

Bicycles are inherently amenable to elaboration because the phage display process selects for binders that tolerate significant molecular bulk (phage approx. 3MDa in size).

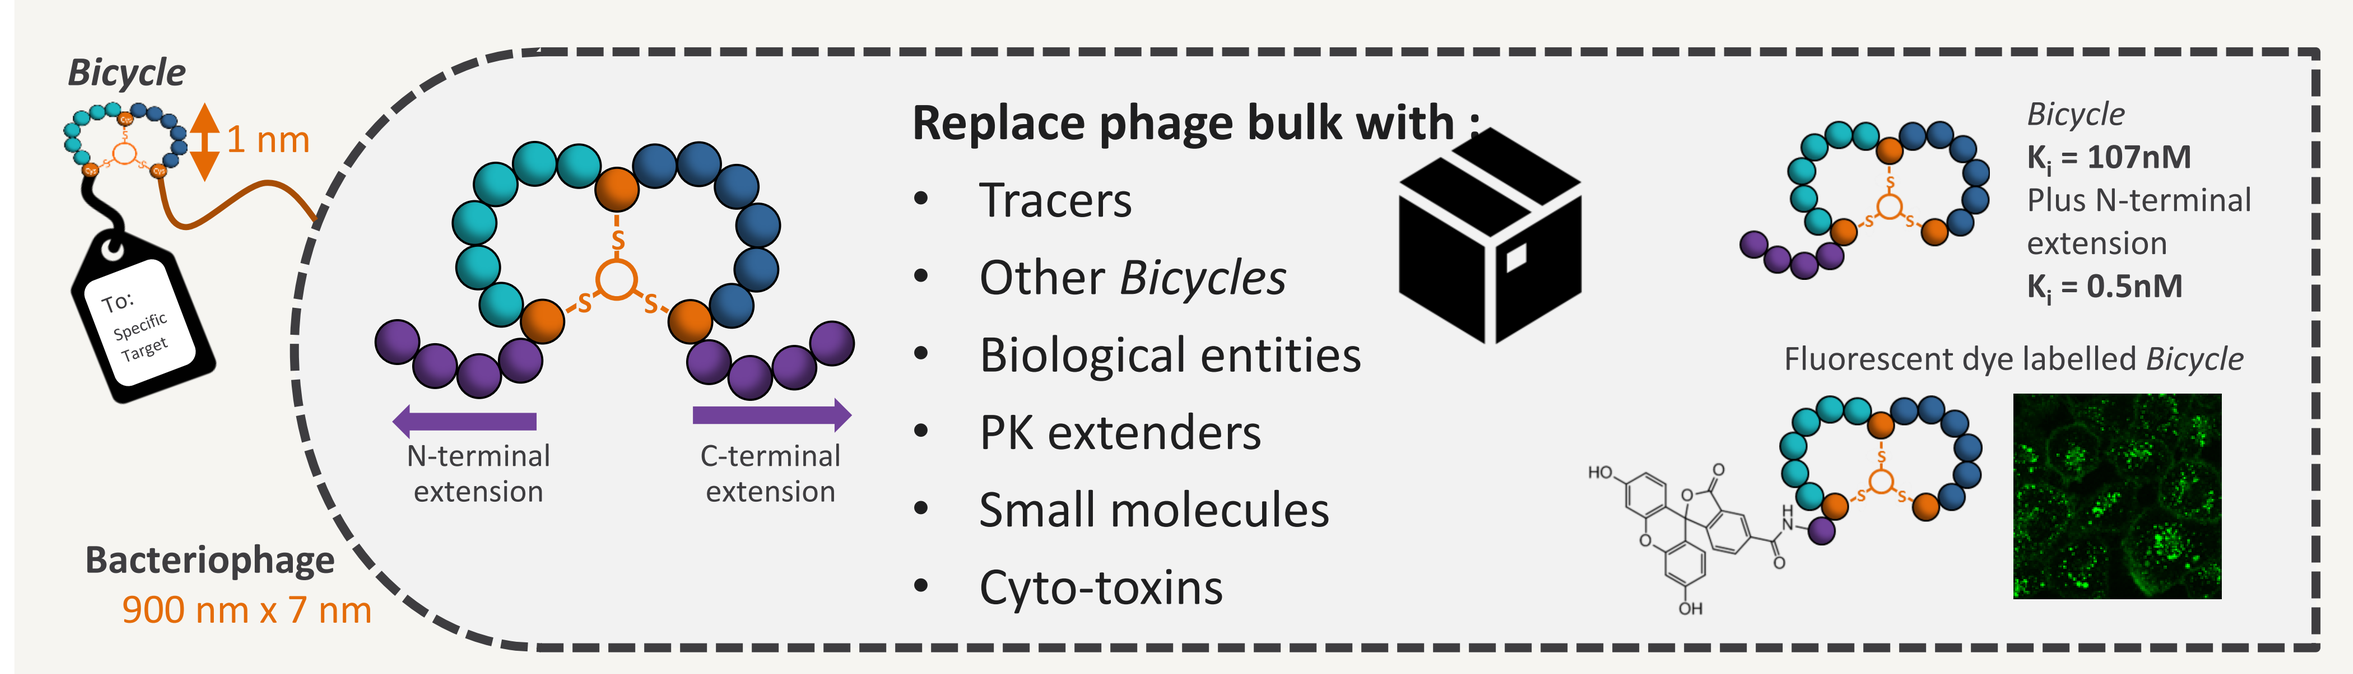

Bicycle
1 nm
To: Specific Target
Bacteriophage
900 nm x 7 nm
N-terminal extension
C-terminal extension
Replace phage bulk with :
Tracers
Other Bicycles
Biological entities
PK extenders
Small molecules
Cyto-toxins
Bicycle
K_i = 107nM
Plus N-terminal extension
K_i = 0.5nM
Fluorescent dye labelled Bicycle

Figure 5: Phage bulk can be replaced with useful chemical/biological entities without adversely affecting binding or function of *Bicycle*

BICYCLE DRUG CONJUGATES®

Bicycle Drug Conjugates® (*BDCs®*) are a new therapeutic modality to treat cancers. They utilise the ability to append payloads to *Bicycles* without negatively impacting target binding. Importantly, *BDCs* overcome the limitations of existing cancer therapies by combining highly-targeted toxin delivery, rapid distribution into tissues and renal route of clearance.

*BDC*s take advantage of a bystander effect to exert their tumour killing effect and as a consequence can be directed against even poorly internalising cell-surface cancer markers such as CD38.

CD38 is a type II transmembrane enzyme and receptor that is overexpressed in a variety of haematological tumours (including multiple myeloma).

Bicycles that bind to, but do not inhibit the normal function of CD38 were identified using our phage display platform. In a mouse xenograft model, a *Bicycle*-DM1 conjugate was able to eradicate CD38 expressing MOLP-8 tumours in just two weeks after only 4 doses (3mg/kg tiw) without a significant effect on body weight.

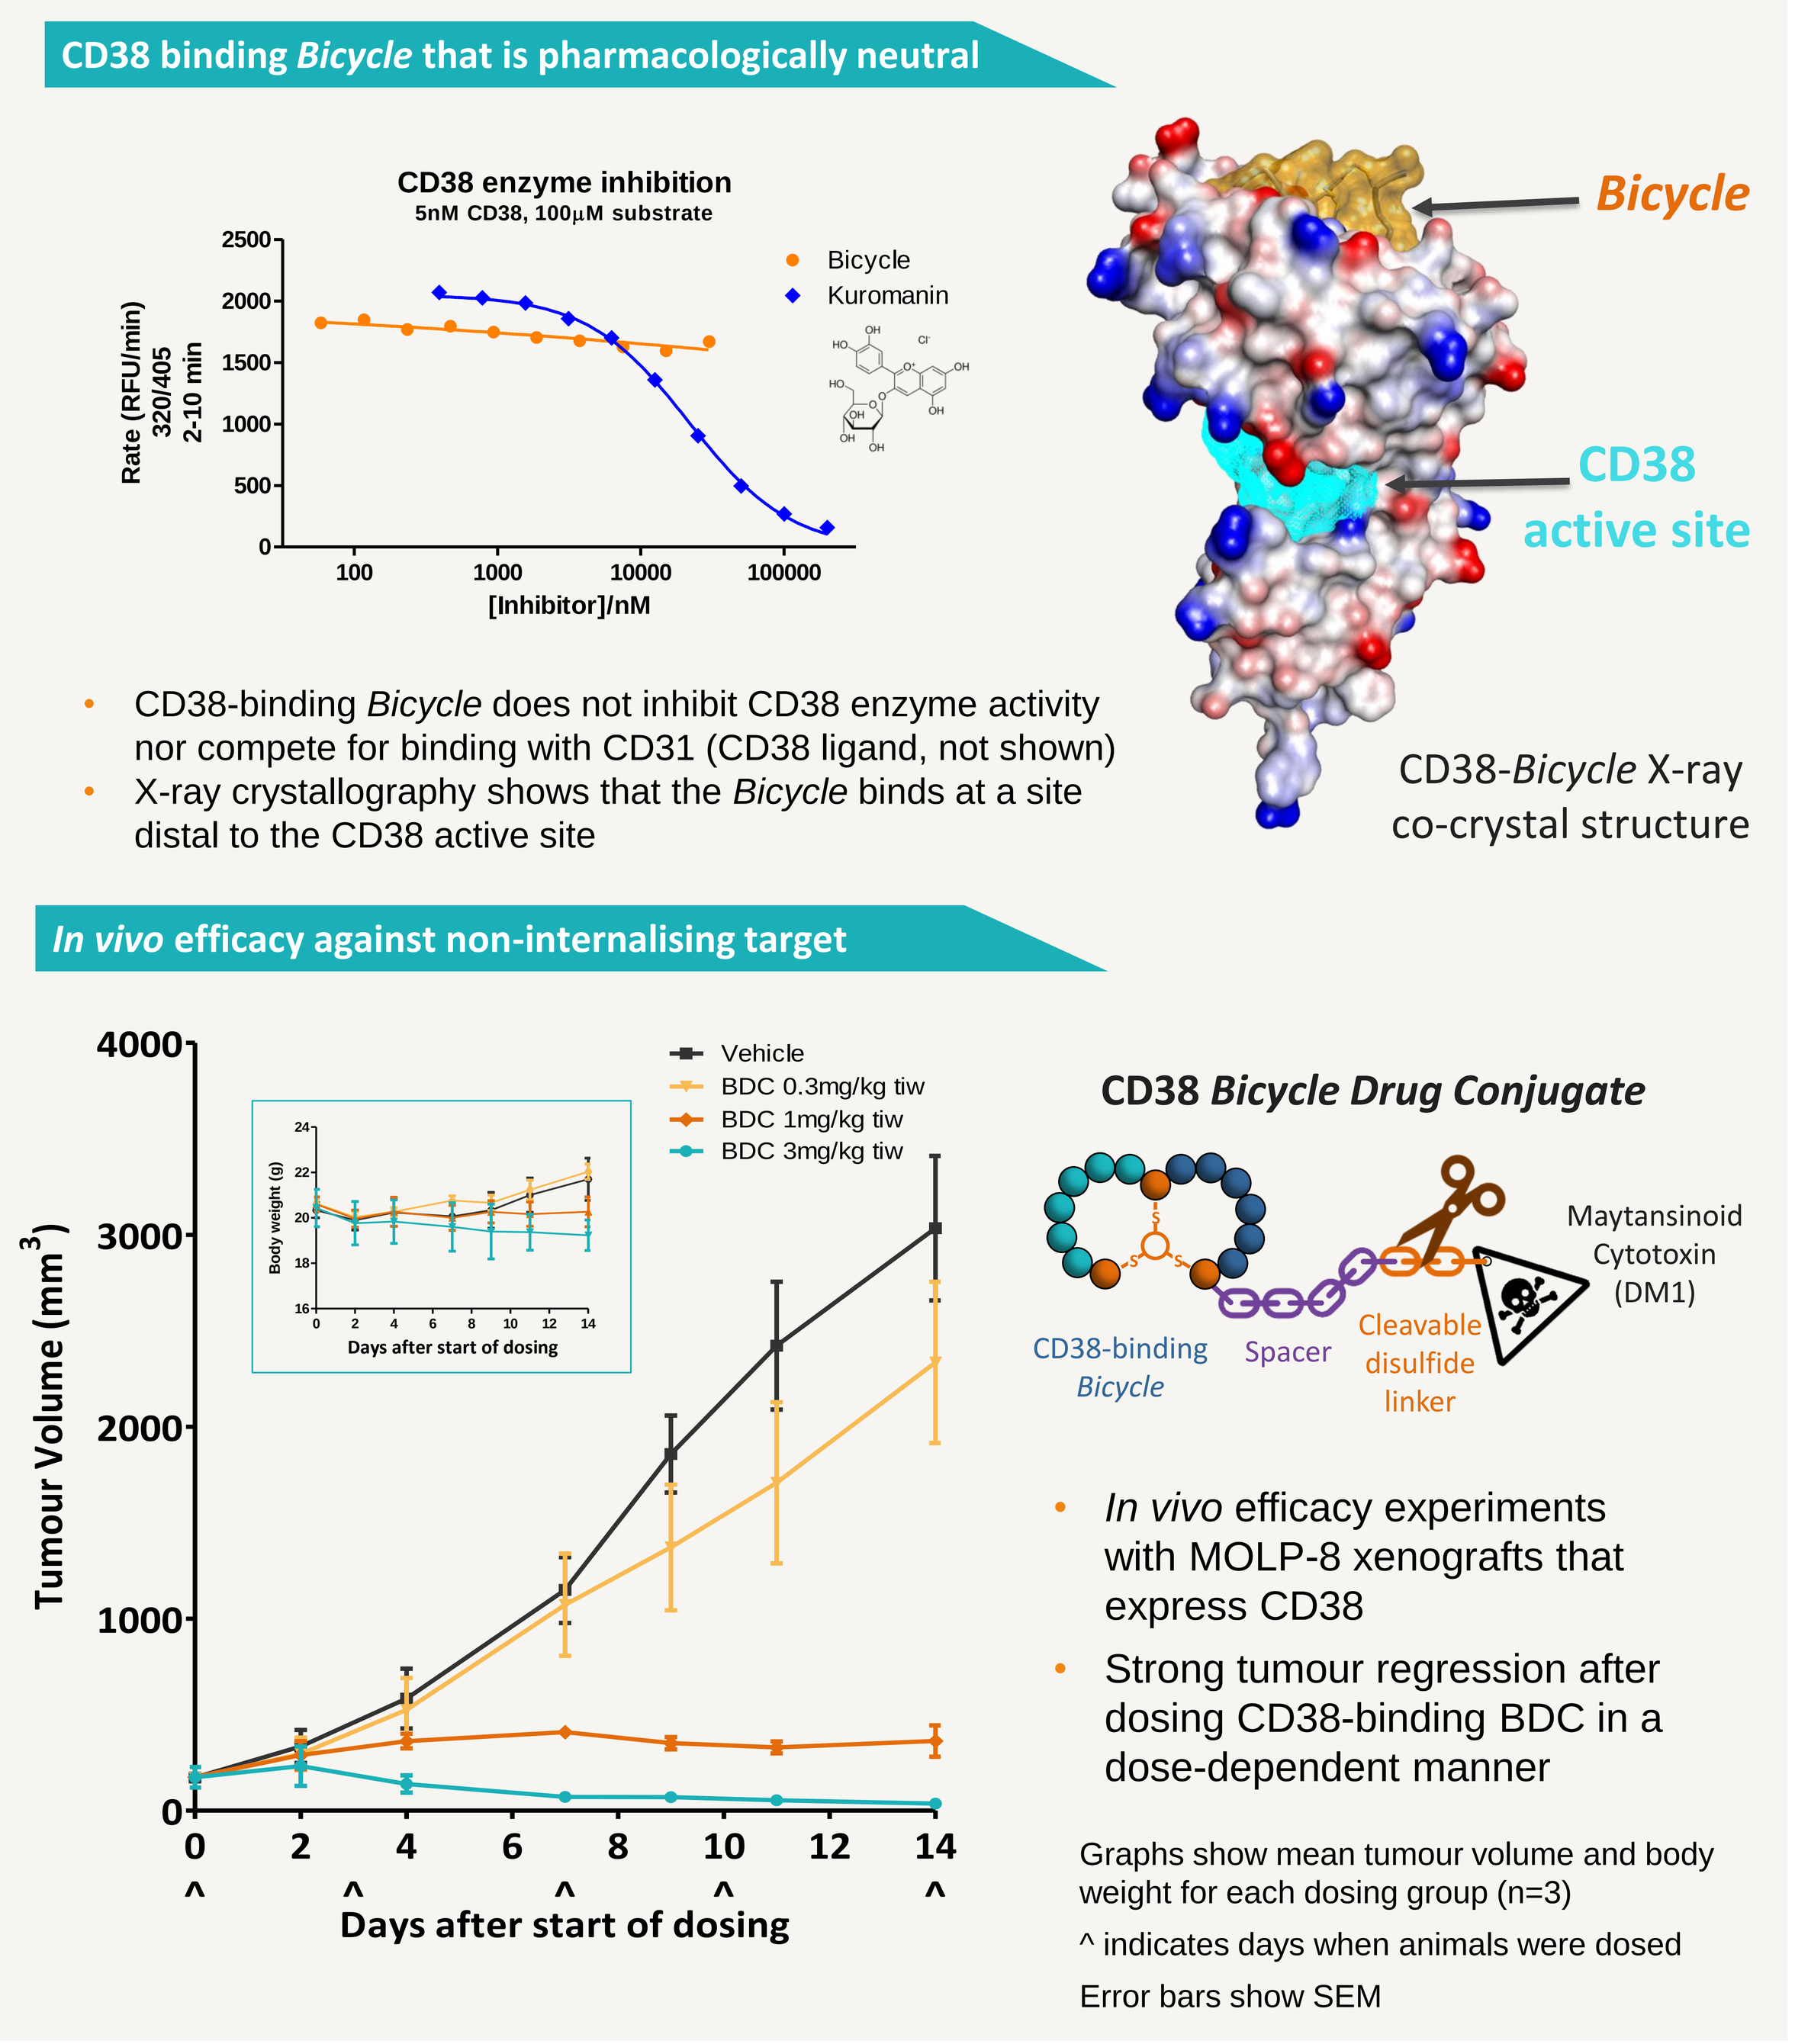

CD38 binding Bicycle that is pharmacologically neutral
CD38 enzyme inhibition
5nM CD38, 100µM substrate
Rate (RFU/min) 320/405 2-10 min
[Inhibitor]/nM
Bicycle
Kuromanin
Bicycle
CD38 active site
CD38-Bicycle X-ray co-crystal structure
CD38-binding Bicycle does not inhibit CD38 enzyme activity nor compete for binding with CD31 (CD38 ligand, not shown)
X-ray crystallography shows that the Bicycle binds at a site distal to the CD38 active site
In vivo efficacy against non-internalising target
Tumour Volume (mm³)
Days after start of dosing
Vehicle
BDC 0.3mg/kg tiw
BDC 1mg/kg tiw
BDC 3mg/kg tiw
Body weight (g)
CD38 Bicycle Drug Conjugate
CD38-binding Bicycle
Spacer
Cleavable disulfide linker
Maytansinoid Cytotoxin (DM1)
In vivo efficacy experiments with MOLP-8 xenografts that express CD38
Strong tumour regression after dosing CD38-binding BDC in a dose-dependent manner
Graphs show mean tumour volume and body weight for each dosing group (n=3)
^ indicates days when animals were dosed
Error bars show SEM

Figure 7: CD38-targeting *Bicycle Drug Conjugate* clears tumour from mice

SUMMARY

- *Bicycles* deliver high affinity and high selectivity by being highly constrained.
- Bicycle's phage platform gives deep coverage of large chemical space that translates to a high success rate against many target classes.
- Bicycles are a fully synthetic, chemically flexible modality that can deliver profound pharmacology.
- Bicycle drug conjugates (BDCs) are highly efficacious against tumours expressing CD38, despite CD38 being a poorly internalising target.
- BDCs limit exposure of non-tumour tissues to cytotoxins by avoiding liver metabolism and being renally excreted.

REFERENCES

1. Heinis *et al., ACS Chem. Biol.,* 7:pp817–821 (2012)
2. Winter *et al., Nature Chem. Biol.,* 5:pp502–507 (2009)
3. De Simone *et al.*, Proc.Natl.Acad.Sci.USA, 106:16233–16238 (2009)
4. Christianson *et al.*, Proc. Natl. Acad. Sci. USA, 98:pp9545–9550 (2001)
5. Supuran, Nature Reviews Drug Discovery. 7:pp168–181 (2008)

–Targets like an antibody **–Performs** like a small molecule **–Excretes** like a peptide

† Bicycle Therapeutics Limited, Cambridge, CB22 3AT, UK
www.bicycletherapeutics.com
‡ Trinity College, University of Cambridge, Cambridge, CB2 1TQ, UK