Preliminary pharmacokinetic assessment of BT1718: A phase I/IIa trial of BT1718 (a first in class Bicycle Toxin Conjugate) in patients with advanced solid tumours

ABSTRACT#
178

Udai Banerji[1], Natalie Cook[2], T.R. Jeffrey Evans[3], Irene Moreno Candilejo[1], Patricia Roxburgh[3], Claire L. S. Kelly[2], Narmatha Sabaratnam[1], Rashmi Passi[1], Sawretse Leslie[4], Sidath Katugampola[4], Lisa Godfrey[4], Gavin Halbert[5], Phillip Jeffrey[6], Mike Rigby[6], Gavin Bennett[6], Maria Koehler[6], Gillian Langford[6], Marc Pittman[4], Stefan N. Symeonides[7]

INTRODUCTION

BT1718 is a novel first in class bicyclic targeting peptide that selectively binds MT1-MMP (MMP-14) and is linked to the maytansinoid tubulin inhibitor DM1 by a cleavable disulfide linker. Bicycle Toxin Conjugates have a low molecular weight compared to other conjugated toxin approaches, enabling rapid tumour penetration and a short systemic half-life (up to 40 minutes in non-human primates) potentially reducing toxicity.

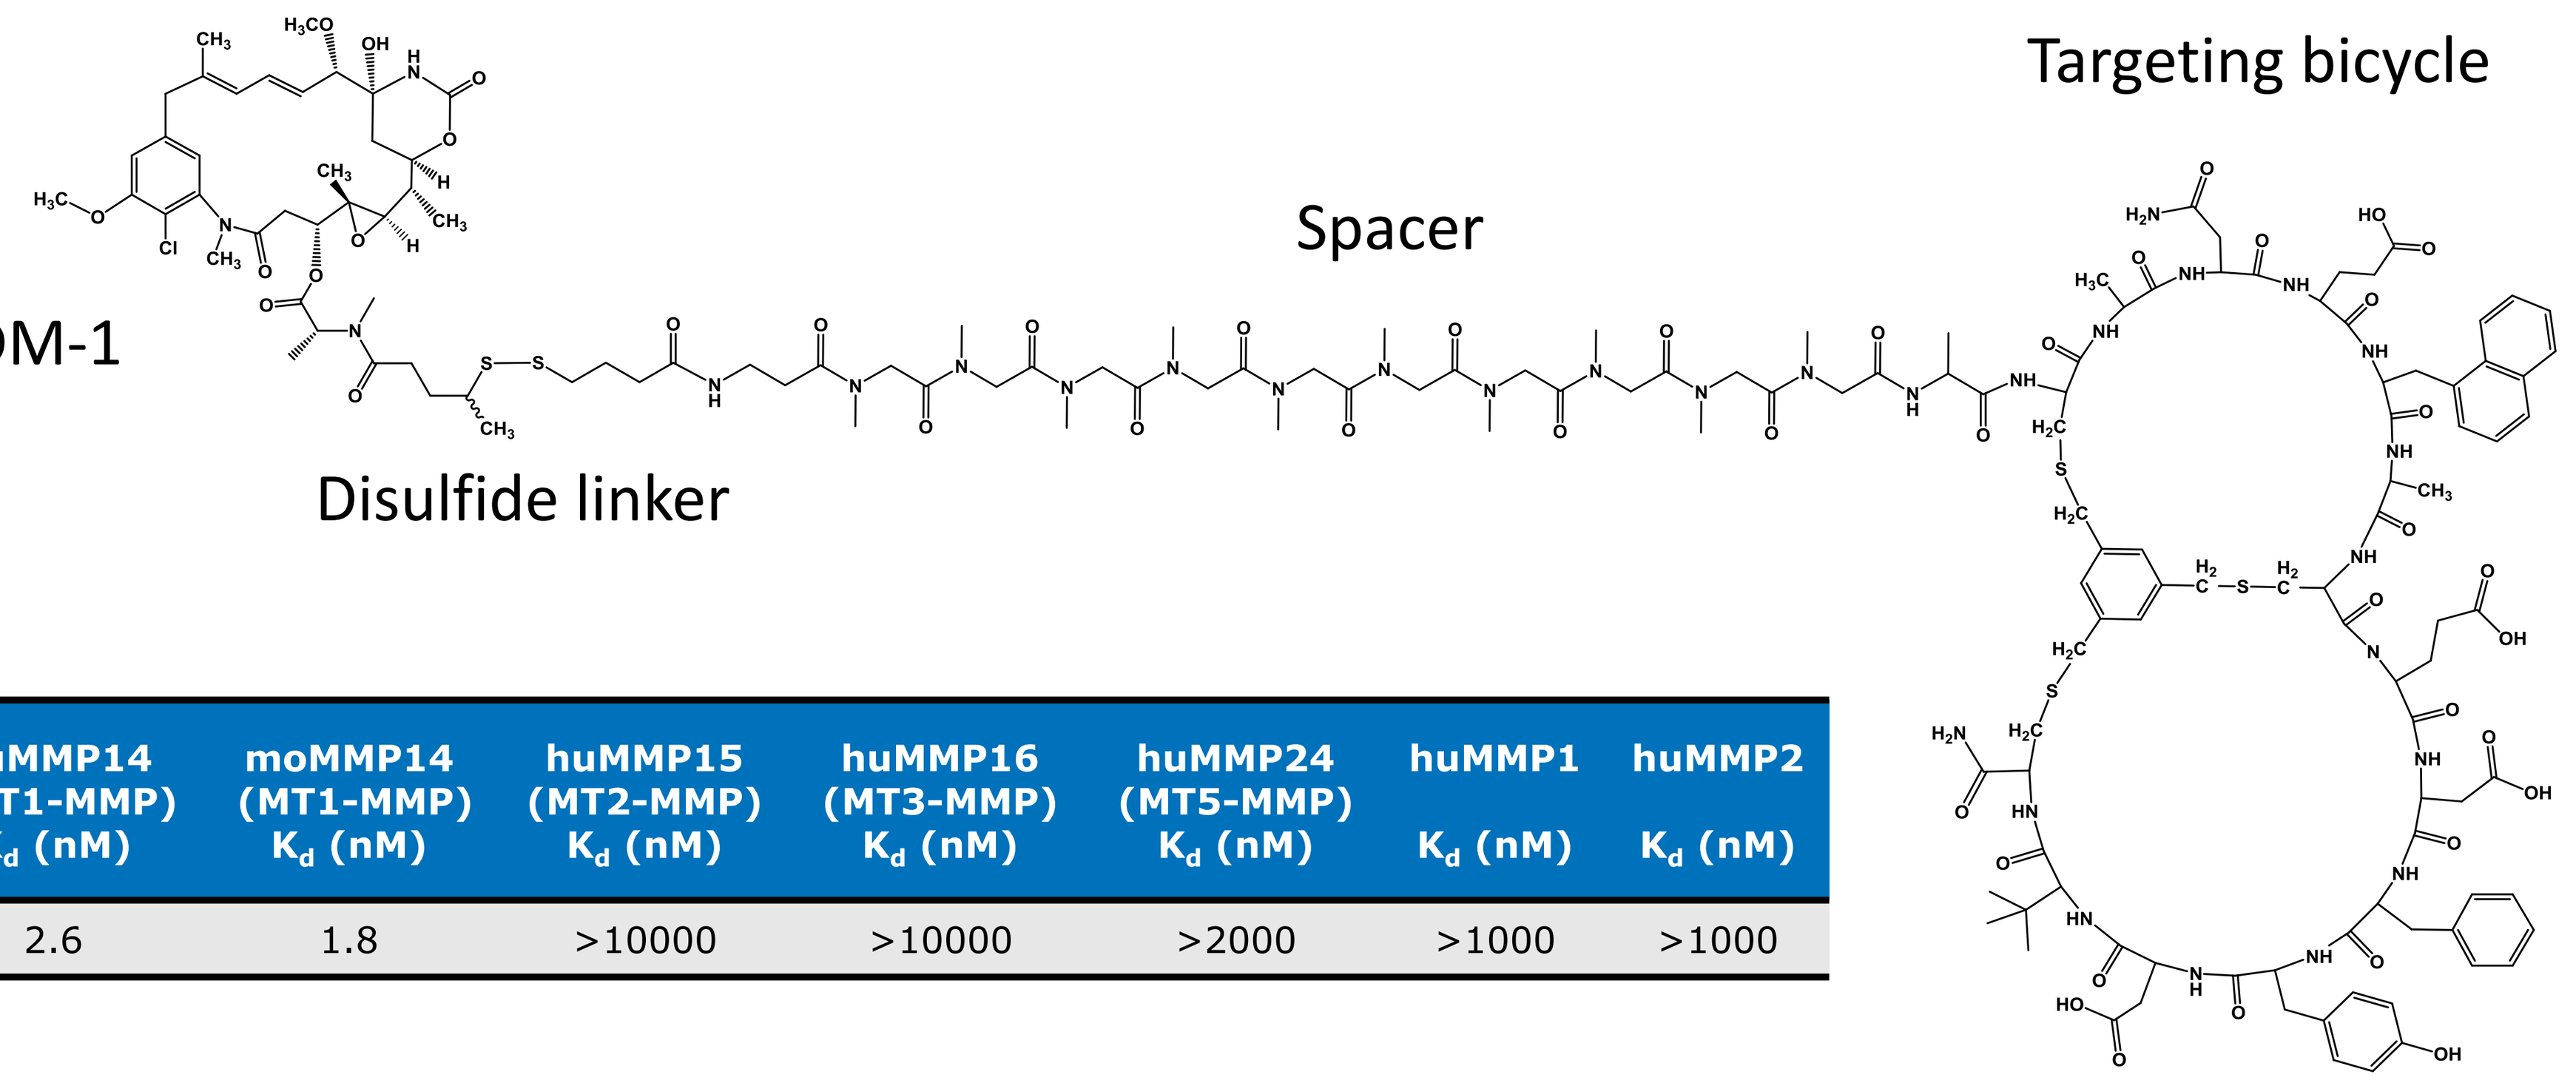
DM-1
Spacer
Targeting bicycle
Disulfide linker

huMMP14 (MT1-MMP) K_d (nM)	moMMP14 (MT1-MMP) K_d (nM)	huMMP15 (MT2-MMP) K_d (nM)	huMMP16 (MT3-MMP) K_d (nM)	huMMP24 (MT5-MMP) K_d (nM)	huMMP1 K_d (nM)	huMMP2 K_d (nM)
2.6	1.8	>10000	>10000	>2000	>1000	>1000

The target MT1-MMP (MT1) is a surface metalloproteinase involved in tissue remodelling through proteolysis of extracellular matrix components**:**

- Highly expressed in tumours with unmet medical need, such as triple negative breast cancer, non small cell lung cancer and ovarian cancer
- Strong link with cell invasion and metastasis
- High tumour MT1 expression is correlated with poor outcomes in multiple tumour types
- High adjacent stromal expression and low expression in adult normal tissue

BT1718 has shown potent anti-tumour preclinical activity, causing complete regressions in CDX and PDX models at doses ranging from 3 – 10 mg/kg.

Regression in MT1-MMP expressing xenografts

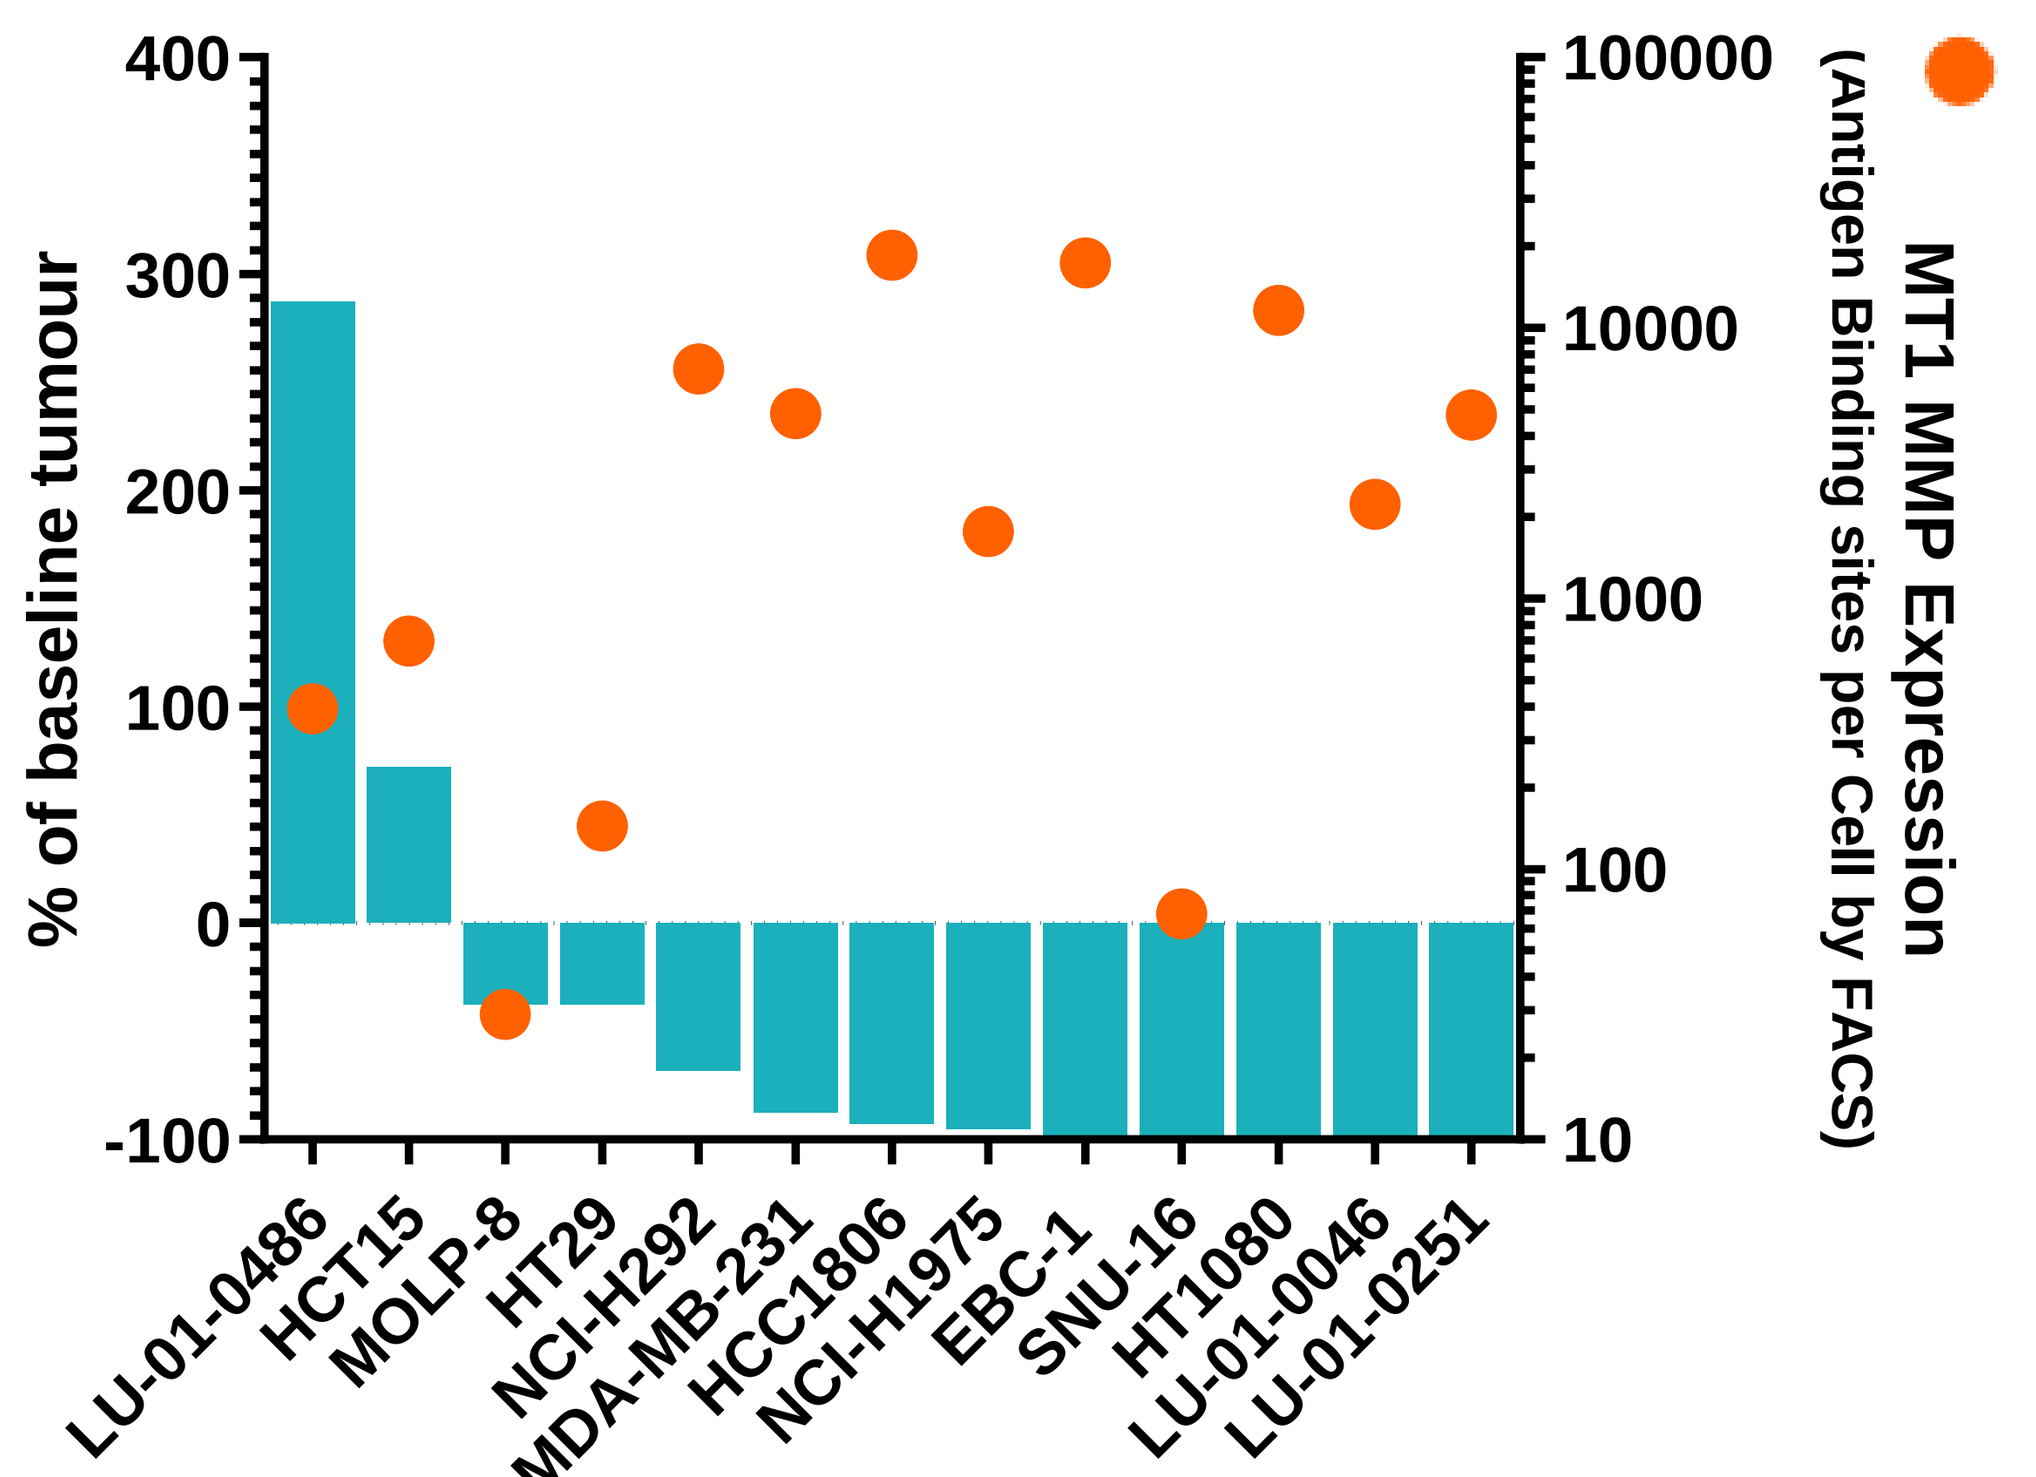
400
300
200
100
0
-100
% of baseline tumour
100000
10000
1000
100
10
MT1 MMP Expression
(Antigen Binding sites per Cell by FACS)
LU-01-0486
HCT15
MOLP-8
HT29
NCI-H292
MDA-MB-231
HCC1806
NCI-H1975
EBC-1
SNU-16
HT1080
LU-01-0046
LU-01-0251

STUDY DESIGN

Open label, first in human phase I/IIa study: primary objective to determine a recommended phase 2 dose (RP2D) and schedule of BT1718. Secondary objectives: pharmacokinetic (PK) parameters, and preliminary efficacy in biomarker pre-defined cohorts. Tertiary objectives: correlative blood and tissue biomarker questions.

- Accelerated dose escalation design with single patient cohorts until grade 2 drug related toxicity, then a 3+3 design to maximum tolerated dose and RP2D
- Two schedules being evaluated; twice a week and a once a week dosing (1 hour intravenous (IV) infusion)
- Parallel expansions in patients with high MT1 expression, exploring clinical & biological activity, to refine schedule, biomarkers & population for final efficacy expansions

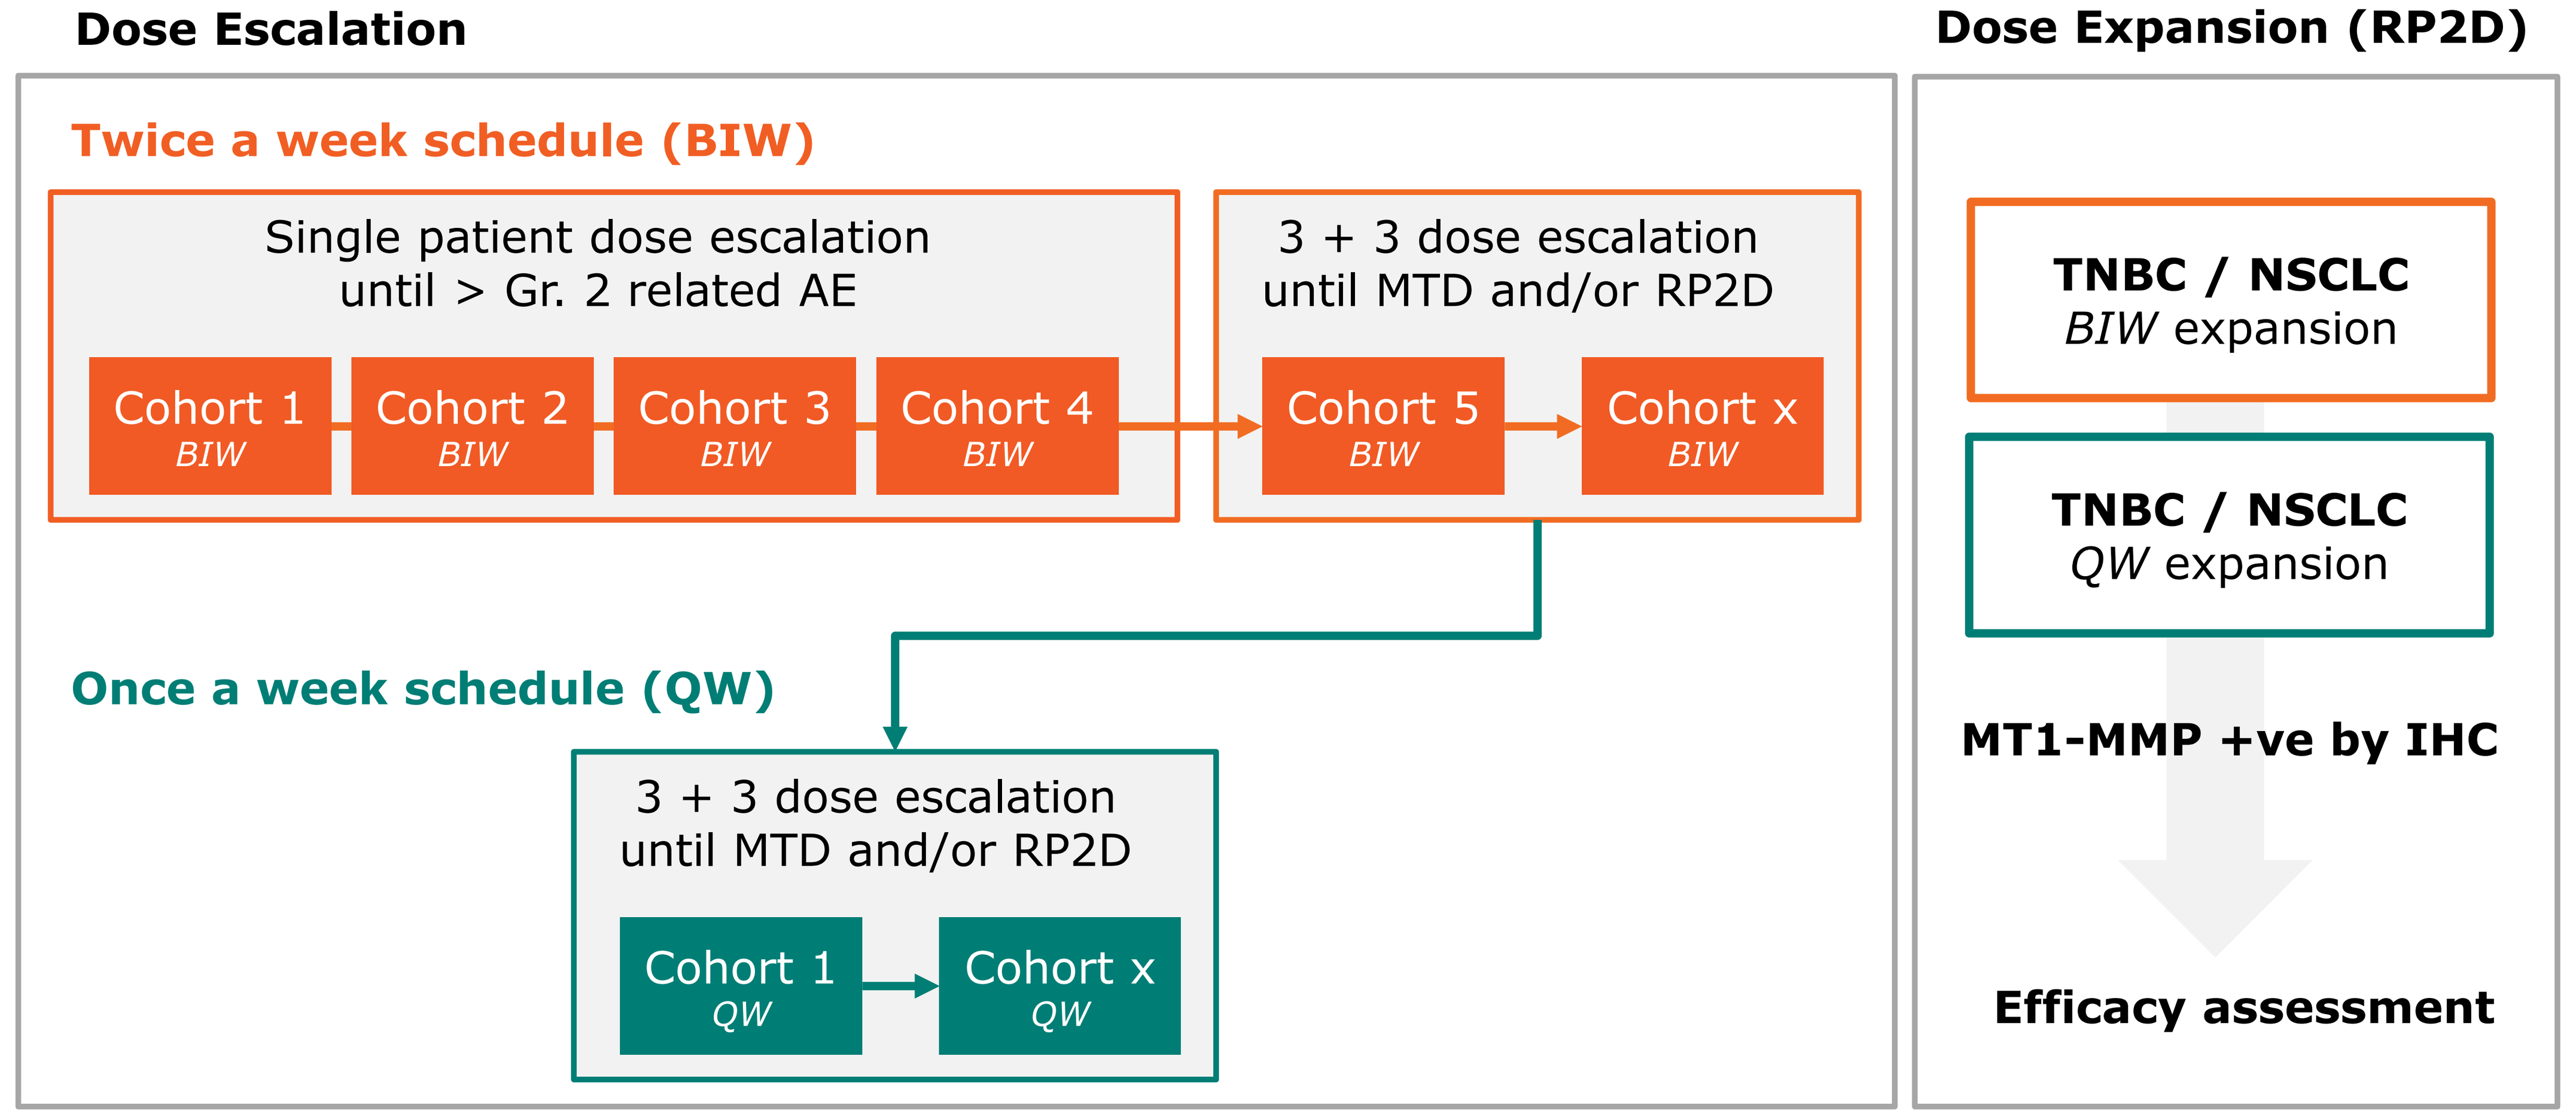
Dose Escalation
Dose Expansion (RP2D)
Twice a week schedule (BIW)
Single patient dose escalation until > Gr. 2 related AE
Cohort 1 BIW
Cohort 2 BIW
Cohort 3 BIW
Cohort 4 BIW
3 + 3 dose escalation until MTD and/or RP2D
Cohort 5 BIW
Cohort x BIW
Once a week schedule (QW)
3 + 3 dose escalation until MTD and/or RP2D
Cohort 1 QW
Cohort x QW
TNBC / NSCLC BIW expansion
TNBC / NSCLC QW expansion
MT1-MMP +ve by IHC
Efficacy assessment

STUDY STATUS: Patient characteristics and dose levels

Recruitment into cohorts 1-5 complete, MTD for twice weekly schedule identified, recruitment into additional twice weekly and once weekly cohorts ongoing.

Characteristics	Patients
Number of patients (cohorts 1 – 5)	8
Male/Female	3/5
Median age (range)	54 (27 – 67)
Tumour types	• 2 CRC • 3 Sarcoma • 1 oesophageal • 1 ovarian • 1 endometrial

Cohort	Dose (mg/m²)	# of patients	# of cycles received	DLTs
1	0.6	1	2	0
2	1.2	1	1	0
3	2.4	1	6	0
4	4.8	1	2	0
5	9.6	4	1 – 2, ongoing*	2

*Dose in cycle 2 reduced to 4.8mg/m² or 7.2mg/m²
Data cut-off 11 Oct 2018

RESULTS: Pharmacokinetics and Safety

BT1718 PK profile, 1 hour IV dosing; cohorts 1 to 5

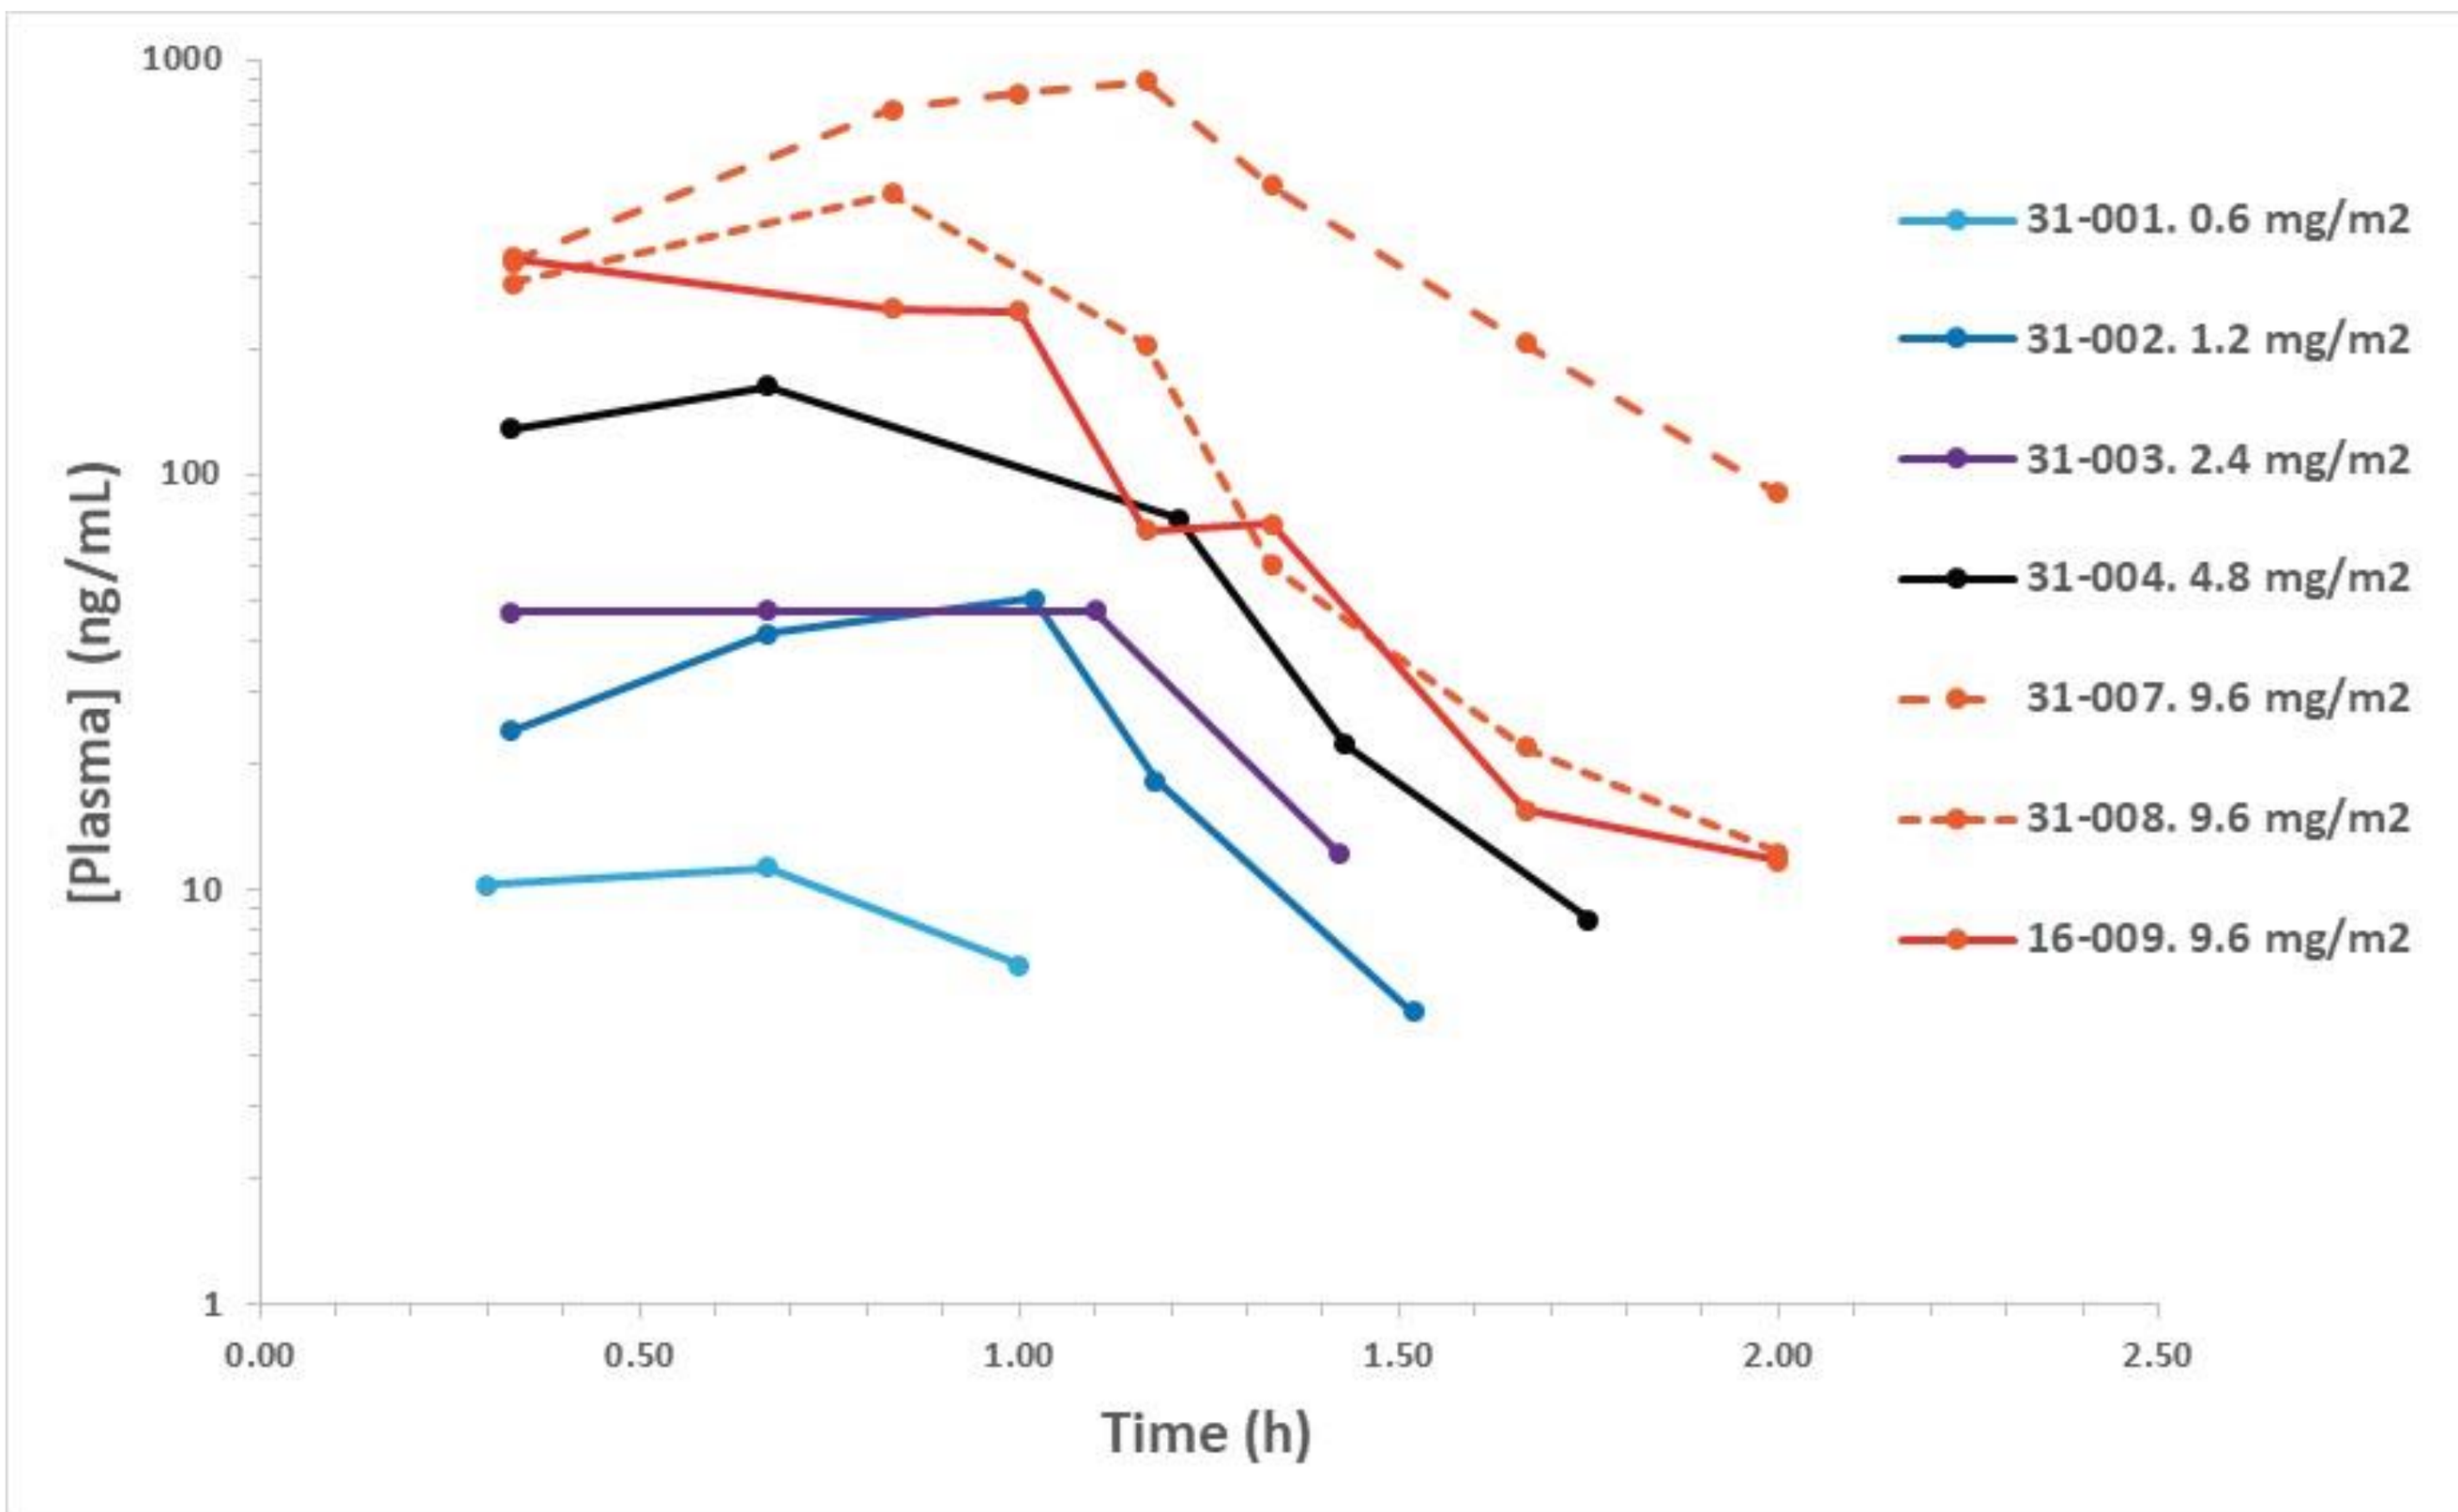
1000
100
10
1
[Plasma] (ng/mL)
0.00
0.50
1.00
1.50
2.00
2.50
Time (h)
31-001. 0.6 mg/m2
31-002. 1.2 mg/m2
31-003. 2.4 mg/m2
31-004. 4.8 mg/m2
31-007. 9.6 mg/m2
31-008. 9.6 mg/m2
16-009. 9.6 mg/m2

- BT1718 AUC increased with dose following a 1h IV administration
- Plasma concentrations were in line with preclinical data
- Plasma clearance (CLp) ranged from 4-17mL/min/kg, with a volume of distribution (Vss) of 0.15-0.30 L/kg, resulting in a terminal half-life ($t_{1/2}$) of 6 to 17 minutes

Safety summary

- Two DLTs were reported in cohort 5, increased GGT (grade 4) and fatigue (grade 3) both of which resolved following cessation or interruption of treatment with BT1718.
- The first DLT occurred in a patient with a history of prior GGT elevation and who had the highest drug exposure (31-007 – see above). Other causes were explored and only concomitant use of paracetamol was noted. This patient had SD at the time of withdrawal. The patient who had the second DLT unfortunately had progressive disease at the time of stopping the study.

CONCLUSIONS/SUMMARY

- The clinical pharmacokinetic profile of BT1718 is consistent with preclinical rodent and primate data (not shown). The AUC of BT1718 increased proportionally with dose following a 1h IV infusion.
- Additional cohorts are being recruited to determine the RP2D for once or twice weekly dosing. Once the RP2D has been determined, efficacy will be assessed in patients selected for high levels of MT1 expression.

AUTHOR AFFILIATIONS

[1]Institute of Cancer Research & Royal Marsden NHS Foundation Trust, Sutton, UK; [2]University of Manchester & Christie NHS Foundation Trust, Manchester, UK; [3]University of Glasgow & Beatson West of Scotland Cancer Centre, Glasgow, UK; [4]Cancer Research UK Centre for Drug Development, London, UK; [5]Cancer Research UK Formulation Unit, University of Strathclyde, Glasgow, UK; [6]Bicycle Therapeutics, Cambridge, UK; [7]University of Edinburgh, Edinburgh UK & Cancer Research UK Centre for Drug Development, London, UK

Thank you to all the patients who have kindly participated in this trial.