BT7480, a fully synthetic tumor-targeted immune cell agonist (TICA™) induces tumor localized CD137 agonism and modulation of tumor immune microenvironment

Johanna Lahdenranta, Punit Upadhyaya, Kristen Hurov, Jessica Kublin, Jun Ma, Elizabeth Repash, Marianna Kleyman, Julia Kristensson, Drasti Kanakia, Fanglei You, Liuhong Chen, Eric Haines, Sailaja Battula, Kevin McDonnell, Philip E. Brandish, and Nicholas Keen

POSTER#
706

ABSTRACT

- After disappointing first clinical experiences with agonistic anti-CD137 (4-1BB) antibodies, a new generation of both systemic and targeted CD137 agonists is entering clinical development (1-3). These strategies rely on biologic agents with suboptimal properties for CD137 agonism due to their relatively large sizes and long circulating half-lives. These properties may limit their tissue penetration and cause sustained agonism resulting in overstimulation and activation-induced cell death of lymphocytes due to continuous exposure.
- Fully synthetic constrained bicyclic peptides (*Bicycles*®) with antibody-like affinities and target selectivity are uniquely suited to circumvent the above barriers to optimal targeted CD137 agonistic therapeutics. BT7480 is a tumor-targeted immune cell agonist (TICA™) designed to deliver a highly potent CD137 agonist to Nectin-4 overexpressing tumor tissue with a flexible dosing schedule maximizing anti-tumor activity while circumventing the need for continuous systemic exposure.
- BT7480 is a fully synthetic TICA that activates CD137 targeting the highly expressed tumor cell antigen Nectin-4.
- BT7480 demonstrates extremely potent Nectin-4 dependent CD137 agonism in primary human PBMC/tumor cell co-culture assays *in vitro* and anti-tumor activity *in vivo* leading to CD8+ T cell dependent immunogenic memory.
- BT7480 induces significant modulation of the tumor immune microenvironment leading to significant increase in the cytotoxic cell population. BT7480 co-stimulatory agonism also induces the transcription of immune checkpoints such as PD-1 and CTLA-4, supporting the concept of combining BT7480 with checkpoint inhibitors for further increased durable activity.
- BT7480 appears well tolerated in non-human primates at exposures in excess of the predicted efficacious exposure in humans. IND enabling activities are underway.

INTRODUCTION

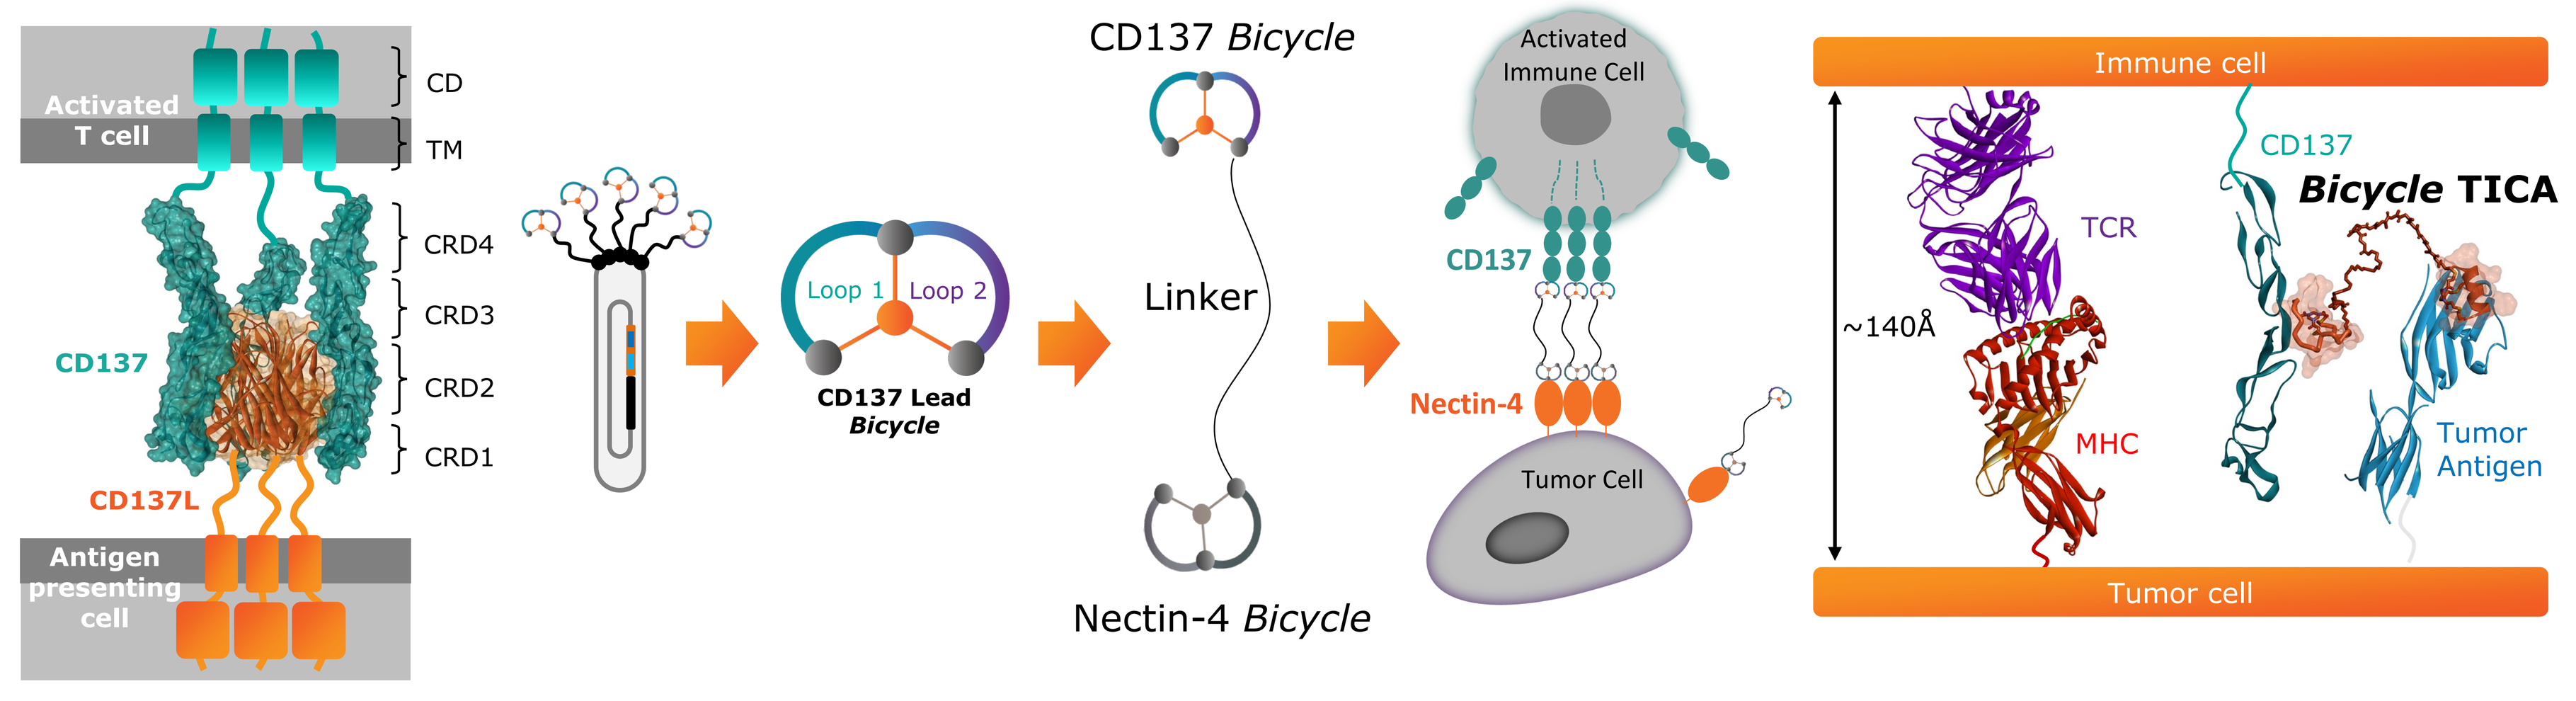
Activated T cell
CD
TM
CRD4
CRD3
CRD2
CRD1
CD137
CD137L
Antigen presenting cell
Loop 1
Loop 2
CD137 Lead Bicycle
CD137 Bicycle
Linker
Nectin-4 Bicycle
Activated Immune Cell
CD137
Nectin-4
Tumor Cell
Immune cell
~140Å
TCR
MHC
CD137
Bicycle TICA
Tumor Antigen
Tumor cell

Figure 1: CD137 is a highly validated immunotherapy target that is expressed on T and NK cells, as well as several other immune cells. CD137 requires trimerization and clustering for its activation. Phage screening, affinity maturation, and chemical optimization resulted in the lead CD137-binding *Bicycle* that was then linked to a Nectin-4-binding *Bicycle*. Nectin-4/PVRL4 is a cell adhesion molecule that is highly expressed in multiple tumor types.Further chemical optimization yielded the development candidate BT7480. TICAs enable optimum spacing at an immune synapse as compared to the spacing formed through interaction of the TCR and MHC.

RESULTS

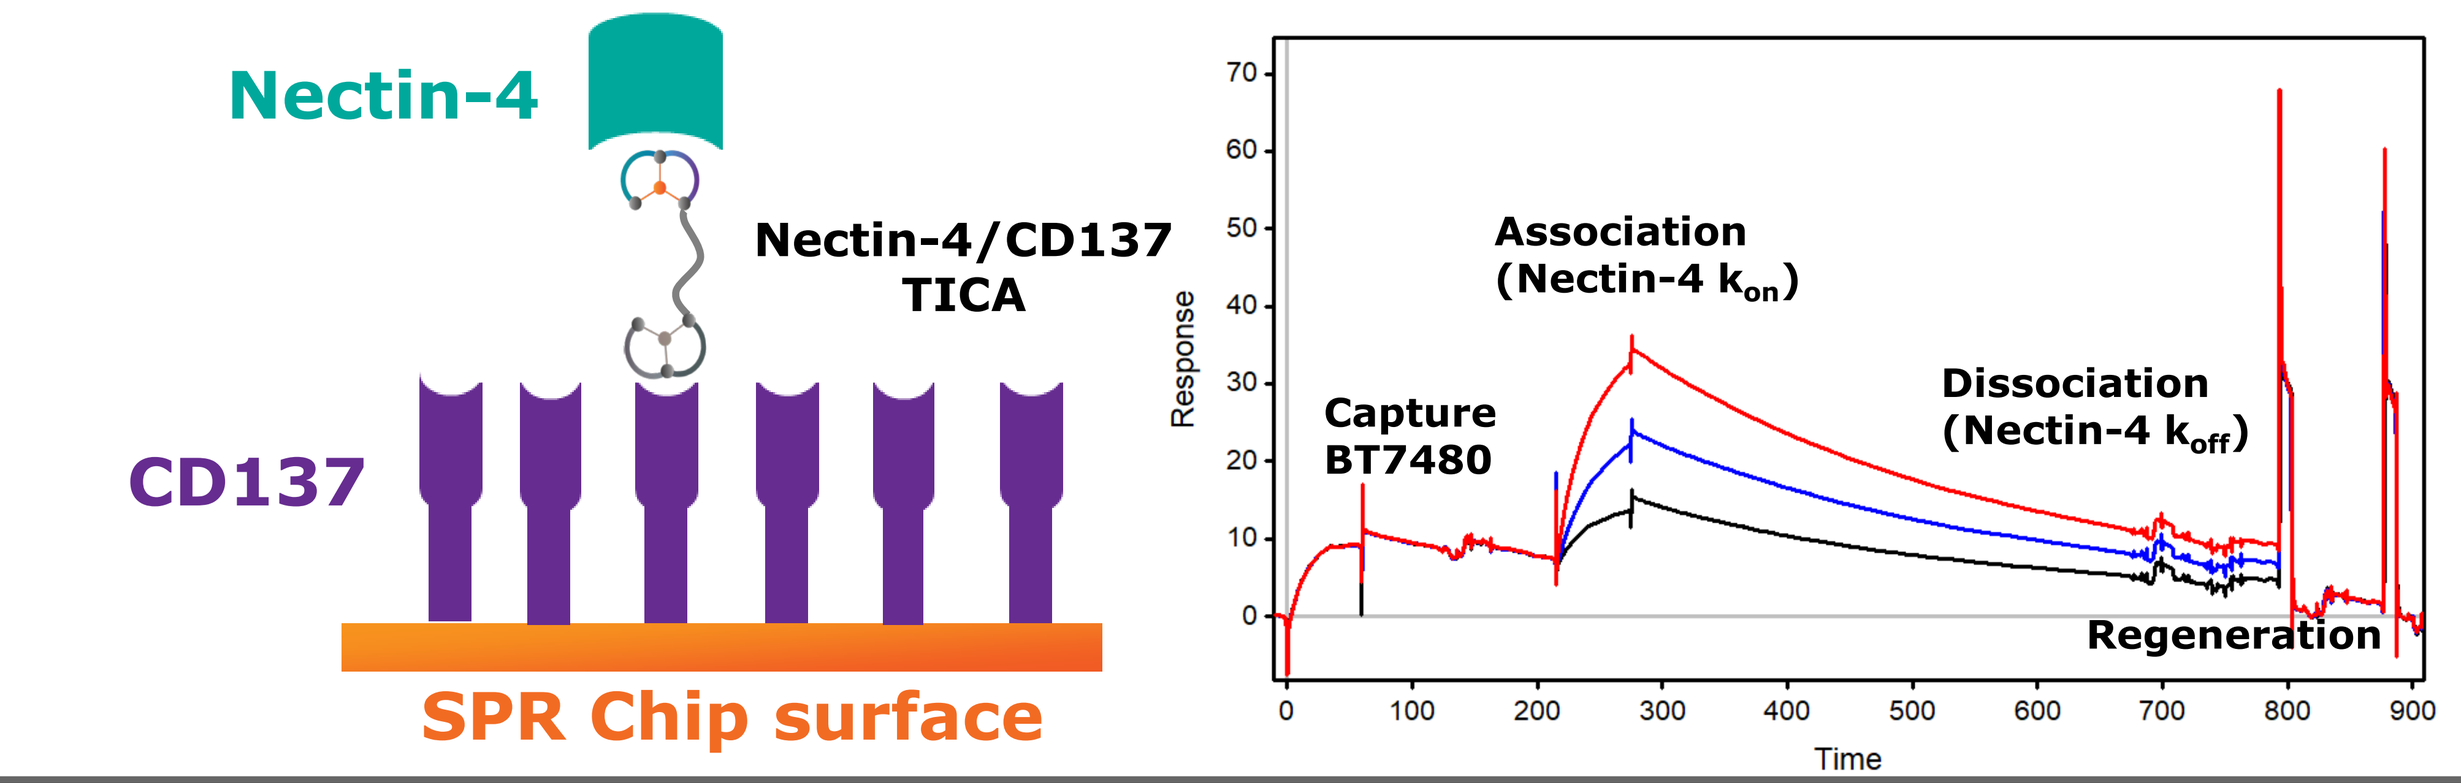
Nectin-4
Nectin-4/CD137 TICA
CD137
SPR Chip surface
Association (Nectin-4 k_{on})
Capture BT7480
Dissociation (Nectin-4 k_{off})
Regeneration
Response
Time

Surface	Analyte	KD (nM)
Nectin-4: BT7480	CD137	6.3 ± 0.7
CD137: BT7480	Nectin-4	12 ± 2

Figure 2: BT7480 can simultaneously bind CD137 and Nectin-4. Biotinylated hCD137 or hNectin-4 was immobilized on SPR (Surface Plasmon Resonance) chip. Each cycle was setup to capture BT7480 with the immobilized proteins followed by injection of the second protein (2-3 fold dilution series) and regeneration of the surface. SPR analysis was run on a Biacore T200 at 25°C at a flow rate of 50µl/min with association time of 60 s and dissociation time of 500 s. Data processing and kinetic fitting were performed using Scrubber software (v2.0c, BioLogic Software).

RESULTS

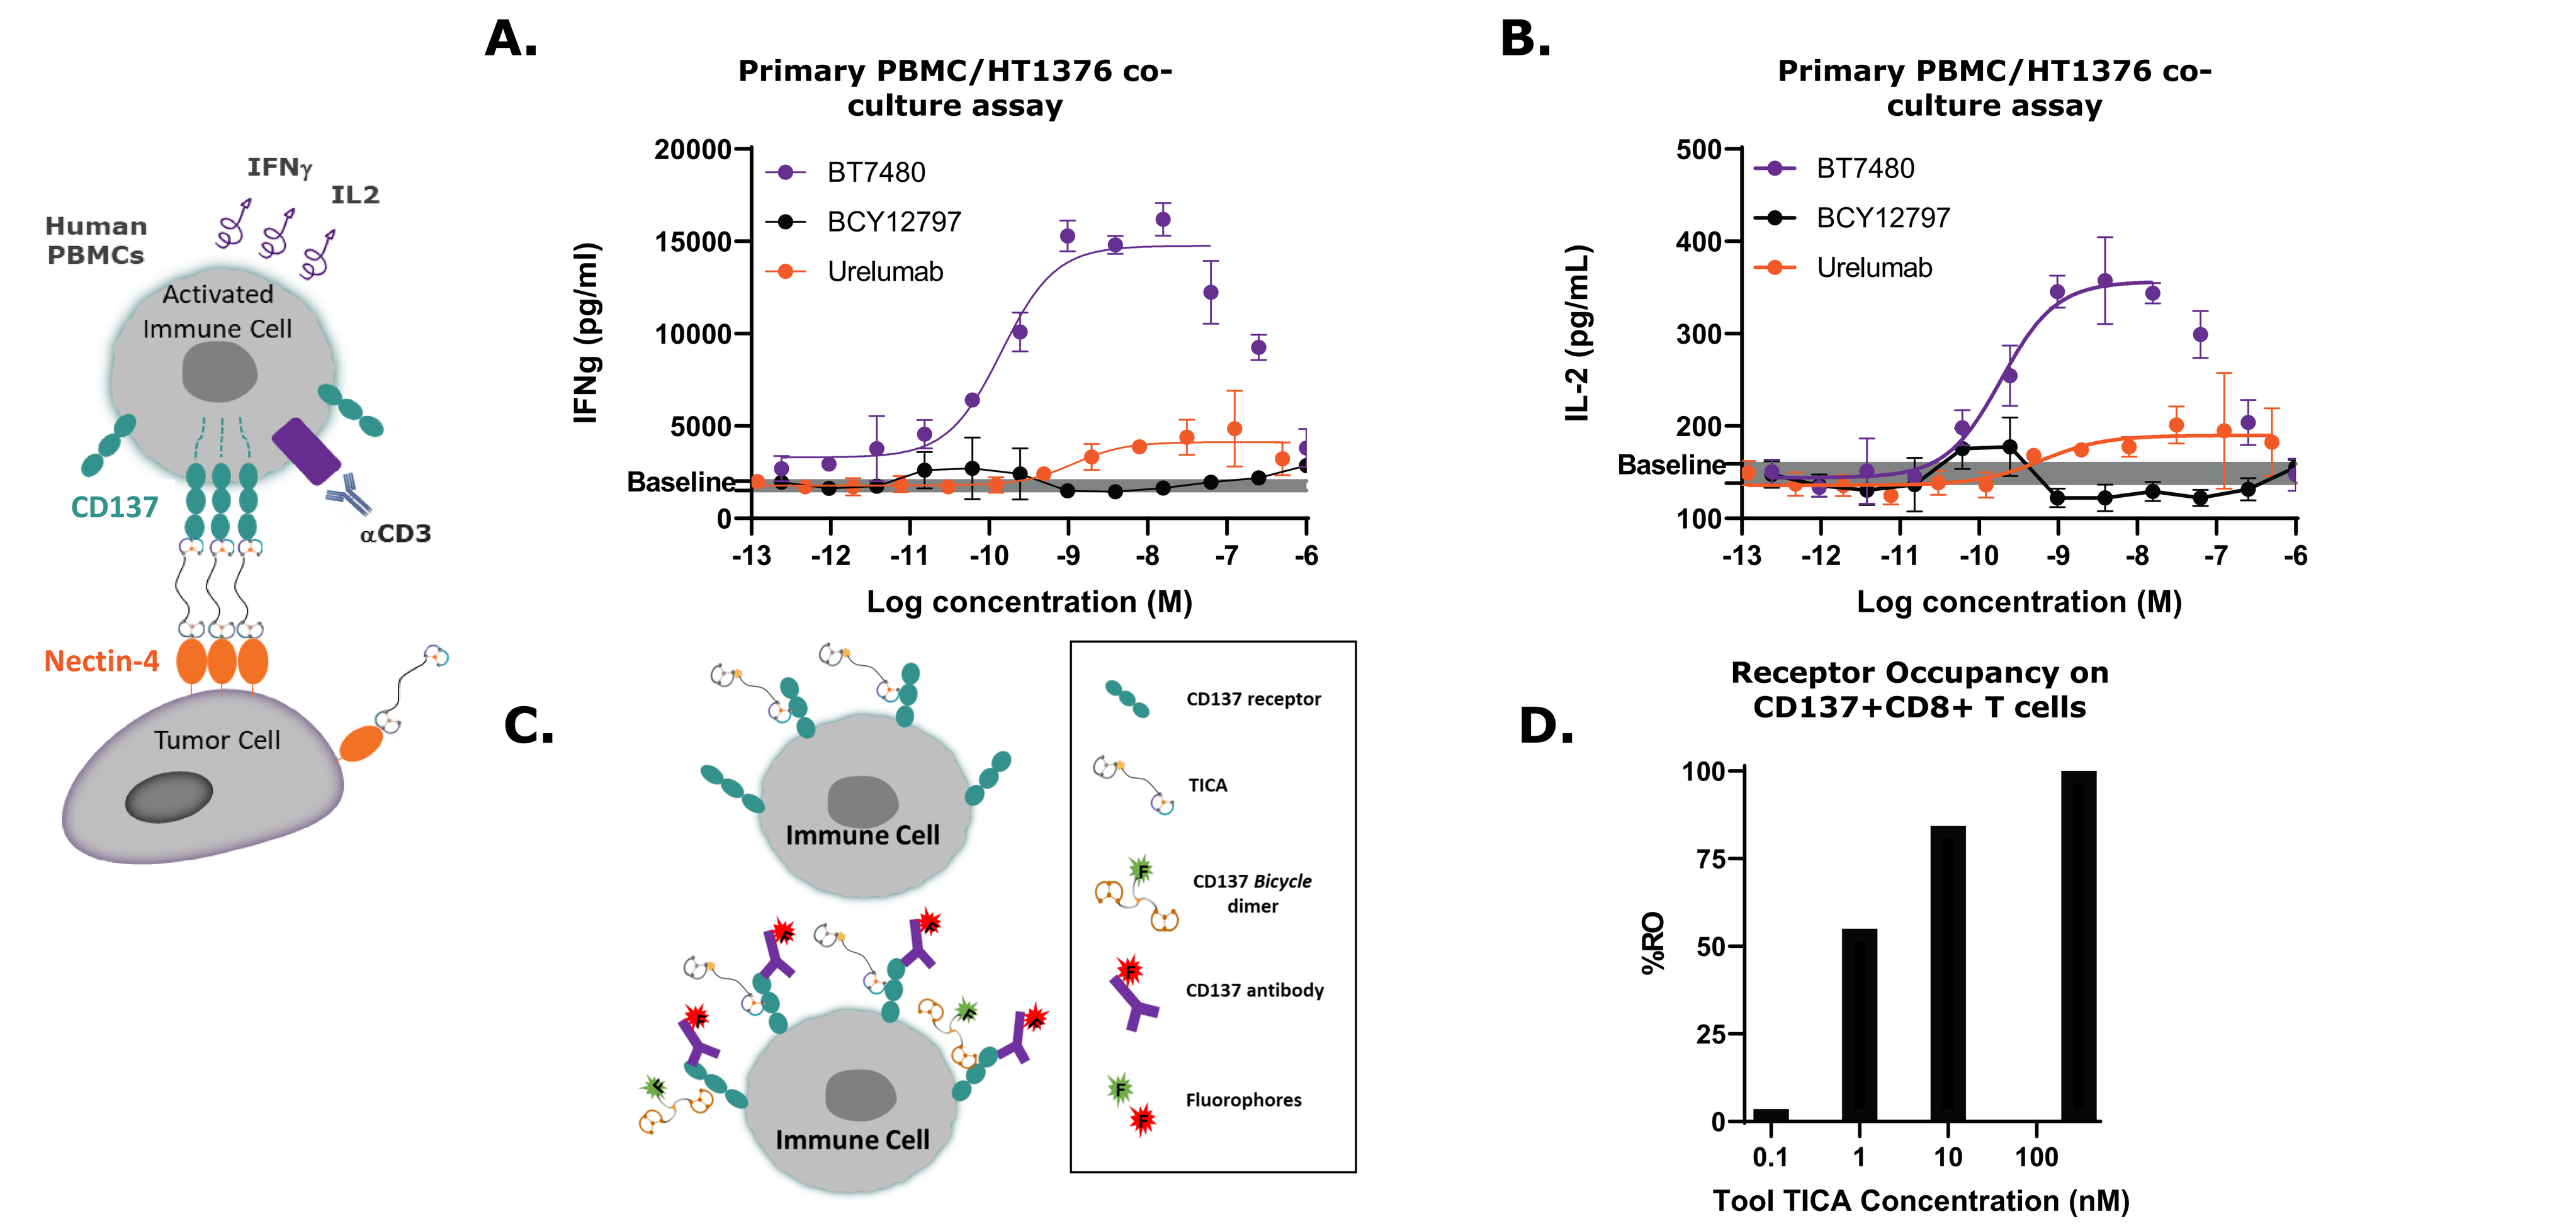
A.
B.
C.
D.
Human PBMCs
IFNγ
IL2
Activated Immune Cell
CD137
αCD3
Nectin-4
Tumor Cell
Primary PBMC/HT1376 co-culture assay
IFNg (pg/ml)
IL-2 (pg/mL)
BT7480
BCY12797
Urelumab
Baseline
Log concentration (M)
Immune Cell
CD137 receptor
TICA
CD137 Bicycle dimer
CD137 antibody
Fluorophores
Receptor Occupancy on CD137+CD8+ T cells
%RO
Tool TICA Concentration (nM)

Figure 3: TICAs bound specifically to CD137-expressing immune cells and BT7480 led to a Nectin-4-dependent increase in cytokine release in human PBMC/tumor cell co-culture assays. A) PBMCs were stimulated with anti-CD3 and co-cultured with the urothelial cancer cell line HT1376 that overexpress Nectin-4. IFNγ and IL-2 levels in the media were measured at 48 h by Luminex (n=3, +/- SD). BT7480 led to increased cytokine release whereas a non-binding analog of BT7480, BCY12797, was not active. The cytokine release induced by αCD137 (Urelumab analogue) is shown for comparison. Similar results were observed with multiple independent PBMC donors. C) We have developed a receptor occupancy assay on immune cells using a fluorescent CD137 Bicycle dimer as a probe for the available TICA (such as BT7480) binding sites. D) CD137 receptor occupancy was measured on activated PBMCs. The CD137 Bicycle dimer selectively bound to CD137 positive cell populations in CD3 –stimulated PBMCs: Following a 1-hour treatment with tool TICA (BCY12491), the level of labelled CD137 dimer was measured within the both the CD137 positive and negative populations and receptor occupancy was calculated as $\%RO=(1-(MFI-Emin)/(Emax-Emin))*100$.

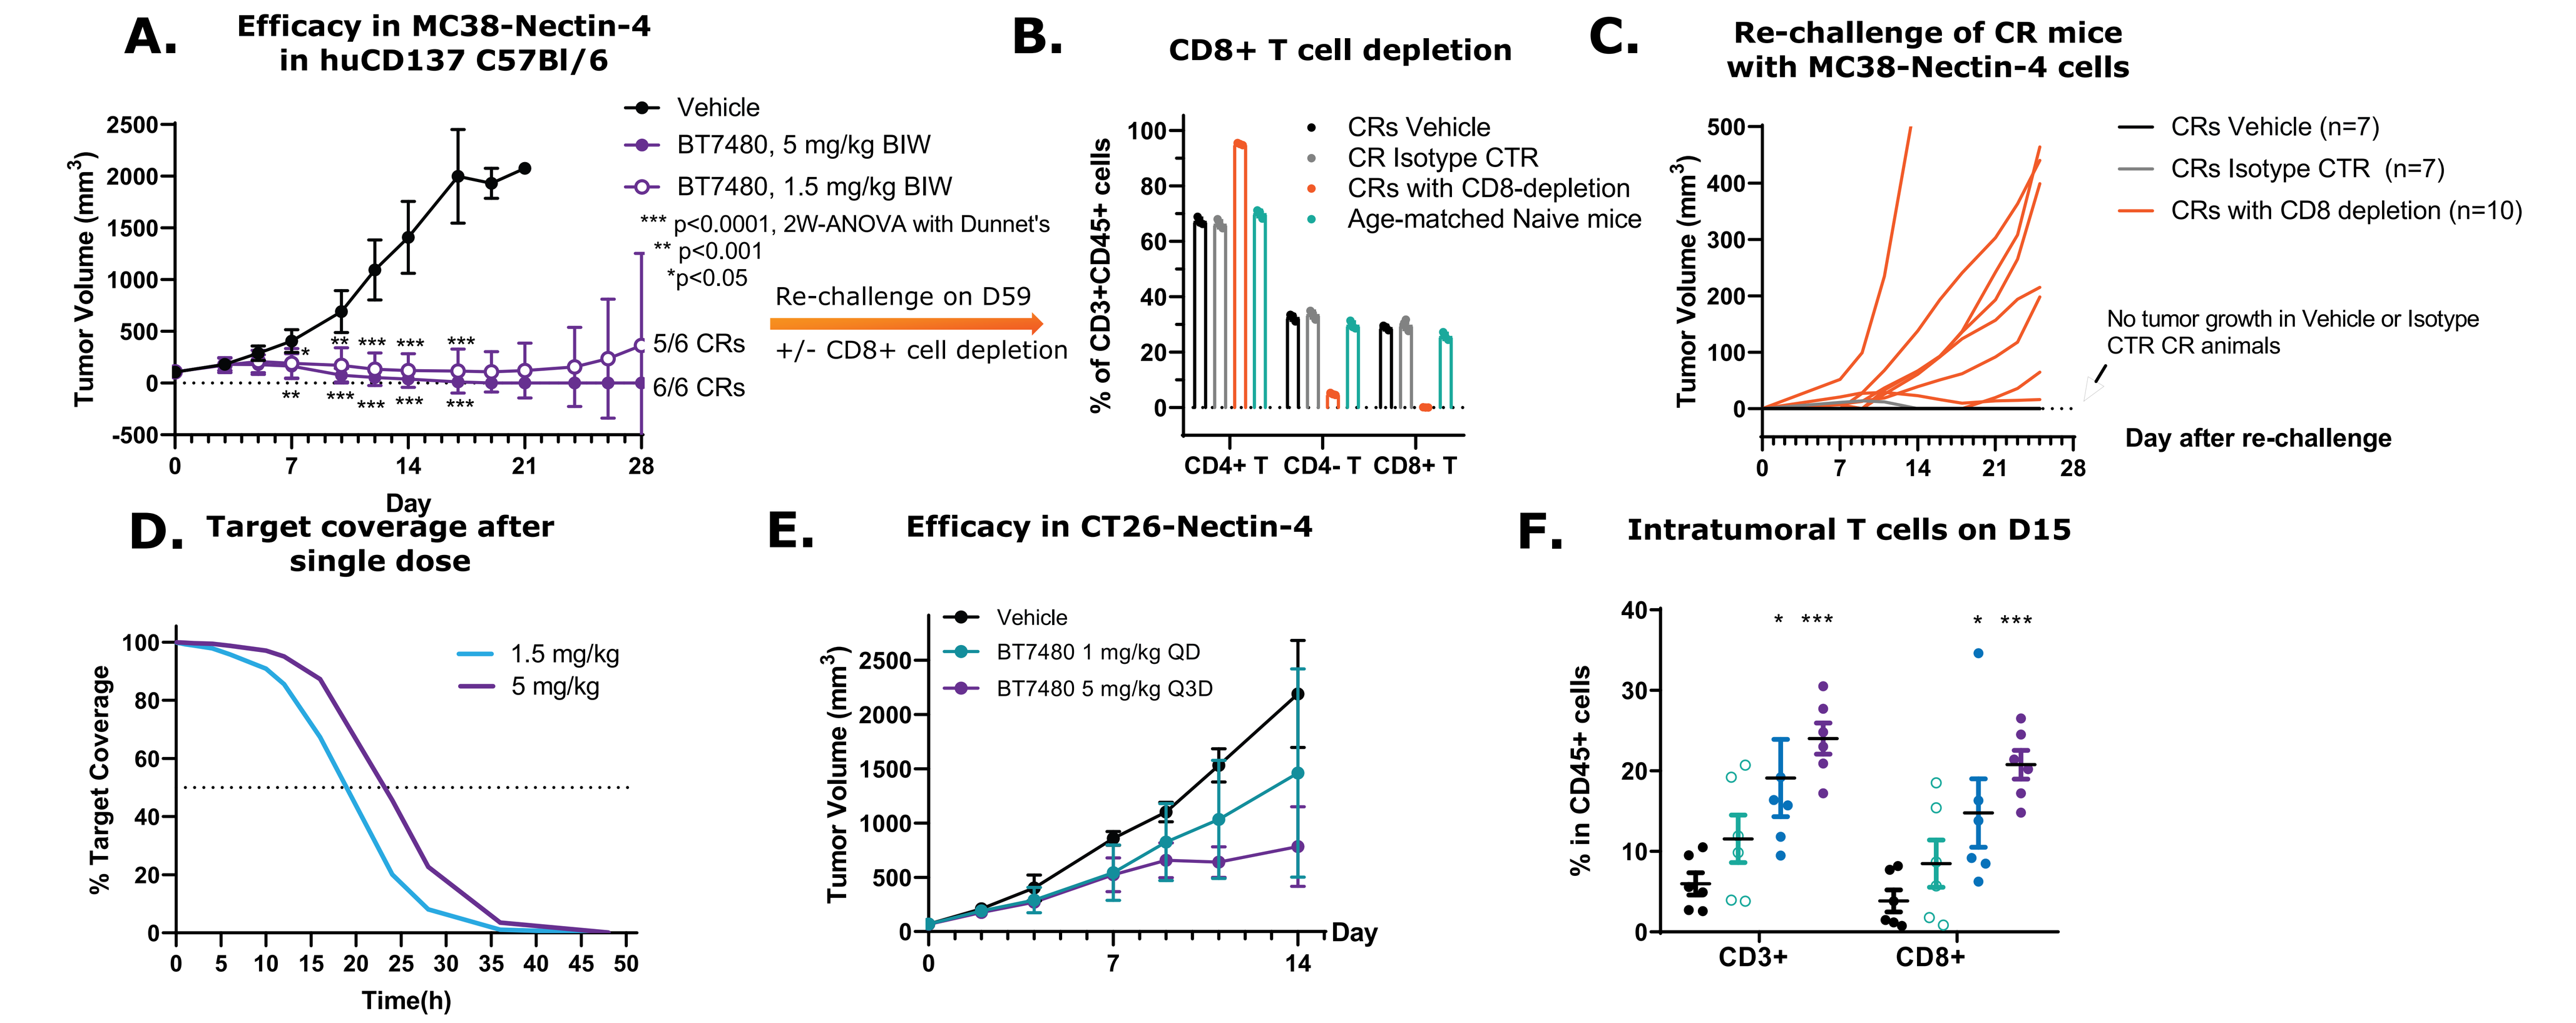
A.
Efficacy in MC38-Nectin-4 in huCD137 C57Bl/6
Vehicle
BT7480, 5 mg/kg BIW
BT7480, 1.5 mg/kg BIW
*** p<0.0001, 2W-ANOVA with Dunnet's
** p<0.001
*p<0.05
5/6 CRs
6/6 CRs
Tumor Volume (mm³)
Day
Re-challenge on D59
+/- CD8+ cell depletion
B.
CD8+ T cell depletion
CRs Vehicle
CR Isotype CTR
CRs with CD8-depletion
Age-matched Naive mice
% of CD3+CD45+ cells
CD4+ T
CD4- T
CD8+ T
C.
Re-challenge of CR mice with MC38-Nectin-4 cells
CRs Vehicle (n=7)
CRs Isotype CTR (n=7)
CRs with CD8 depletion (n=10)
No tumor growth in Vehicle or Isotype CTR CR animals
Day after re-challenge
D.
Target coverage after single dose
1.5 mg/kg
5 mg/kg
% Target Coverage
Time(h)
E.
Efficacy in CT26-Nectin-4
Vehicle
BT7480 1 mg/kg QD
BT7480 5 mg/kg Q3D
F.
Intratumoral T cells on D15
% in CD45+ cells
CD3+
CD8+

Figure 4: Intermittent dosing of BT7480 led to T cell infiltration and robust anti-tumor activity and resistance to re-challenge in a syngeneic mouse model. A) In syngeneic huCD137 mice carrying MC38-Nectin-4 tumors (MC38 engineered to overexpress Nectin-4), BT7480 led to complete responses (CRs) in 11/12 mice dosed twice a week (BIW). B) CD8+ T cells in CR mice were depleted prior to re-challenge, as indicated by flow cytometry. G) CR mice (with or w/o CD8-depletion) were re-implanted with MC38-Nectin-4 cells. All tumors were rejected in animals w/o CD8-depletion, indicating an established memory response. Memory response was dependent CD8+ T cells since most tumors (8/10) grew in CD8-depleted mice. D) 5 mg/kg dose of BT7480 maintains ≥50% target coverage for ~25 hours (simulated from PK data). Dosed BIW, plasma exposure of BT7480 is intermittent with ≤50% target coverage for at least 47 hours. E) Anti-tumor activity was observed also in syngeneic huCD137 mice carrying CT26-Nectin-4 tumors. F) Anti-tumor activity concurred with increased infiltration of CD8+ T cells into the tumor tissue.

RESULTS

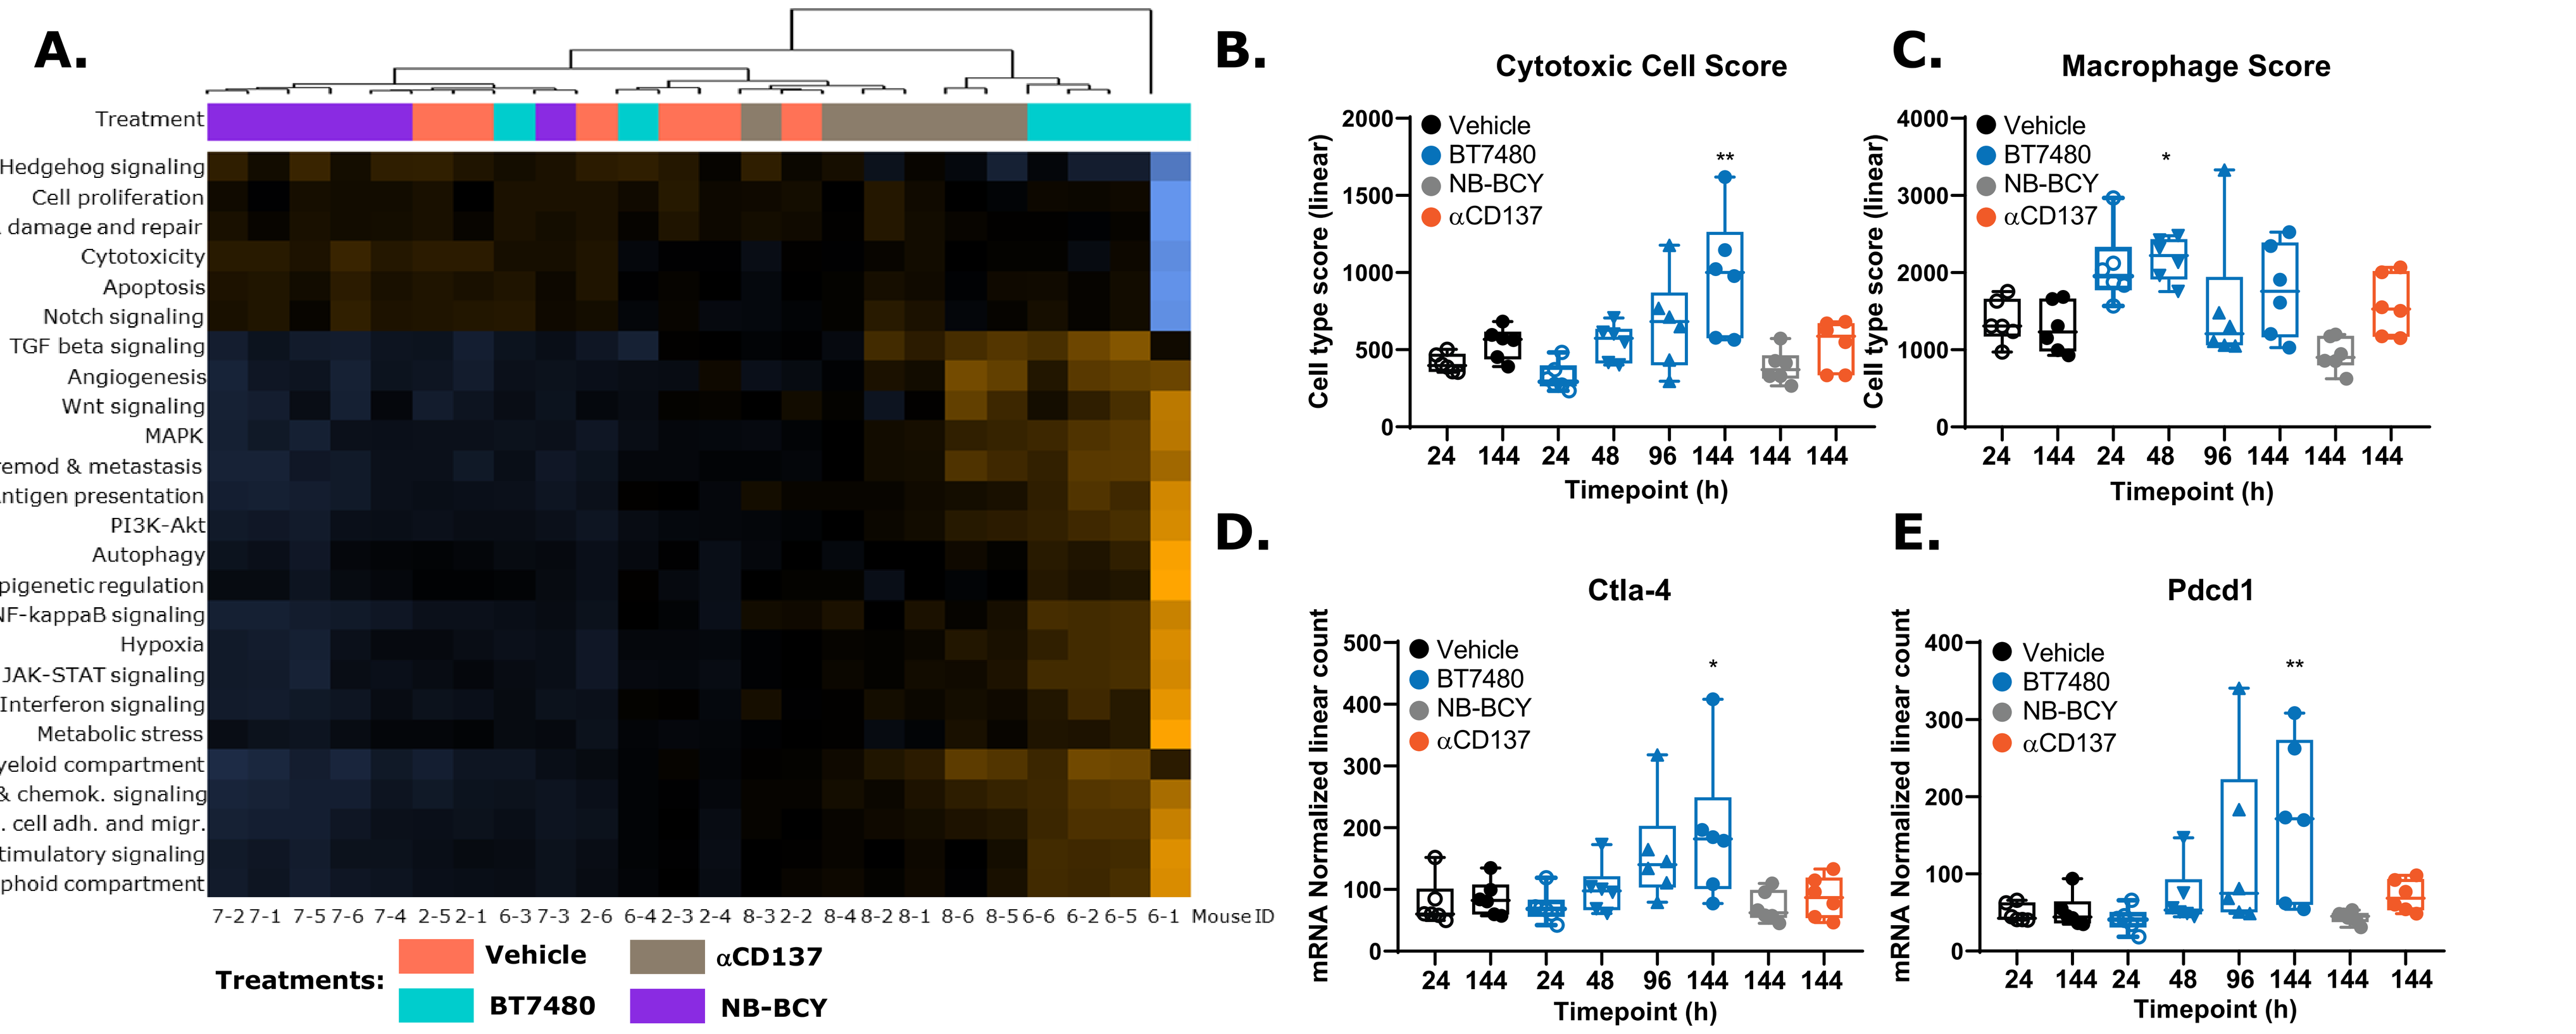
A.
Treatment
Hedgehog signaling
Cell proliferation
DNA damage and repair
Cytotoxicity
Apoptosis
Notch signaling
TGF beta signaling
Angiogenesis
Wnt signaling
MAPK
Matrix remod & metastasis
Antigen presentation
PI3K-Akt
Autophagy
Epigenetic regulation
NF-kappaB signaling
Hypoxia
JAK-STAT signaling
Interferon signaling
Metabolic stress
Myeloid compartment
Cytok. & chemok. signaling
Imm. cell adh. and migr.
Costimulatory signaling
Lymphoid compartment
Mouse ID
Treatments:
Vehicle
αCD137
BT7480
NB-BCY
B.
Cytotoxic Cell Score
C.
Macrophage Score
Cell type score (linear)
Timepoint (h)
D.
Ctla-4
E.
Pdcd1
mRNA Normalized linear count

Figure 5: BT7480 treatment led to significant immunomodulation in tumor tissue.
MC38-Nectin-4 tumor bearing mice (huCD137-C57Bl/6) were treated with Vehicle, 5 mg/kg BT7480 or Non-Binding control Bicycle (NB-BCY; BCY12797) iv at 0h and 24h or 2 mg/kg αCD137 antibody (Urelumab analogue) BIW. Tumors were harvested at 24, 48, 96 or 144h as indicated, RNA was isolated and transcriptional analysis performed by Nanostring. Nanostring data was analyzed using nSolver software. (A) Heatmap of pathway scores across MC38-Nectin-4 tumors 144 hours after treatment initiation. Orange indicates high scores; blue indicates low scores. Scores are displayed on the same scale via a Z-transformation. (B) Increase in cytotoxic cell score, (C) macrophage score, and immune checkpoint molecules (D) Ctla-4 and (E) Pdcd1 (PD-1) in tumor tissue after BT7480 treatment over time. (B-E) *p<0.05, **p<0.01, 1wayANOVA with Dunnett's.

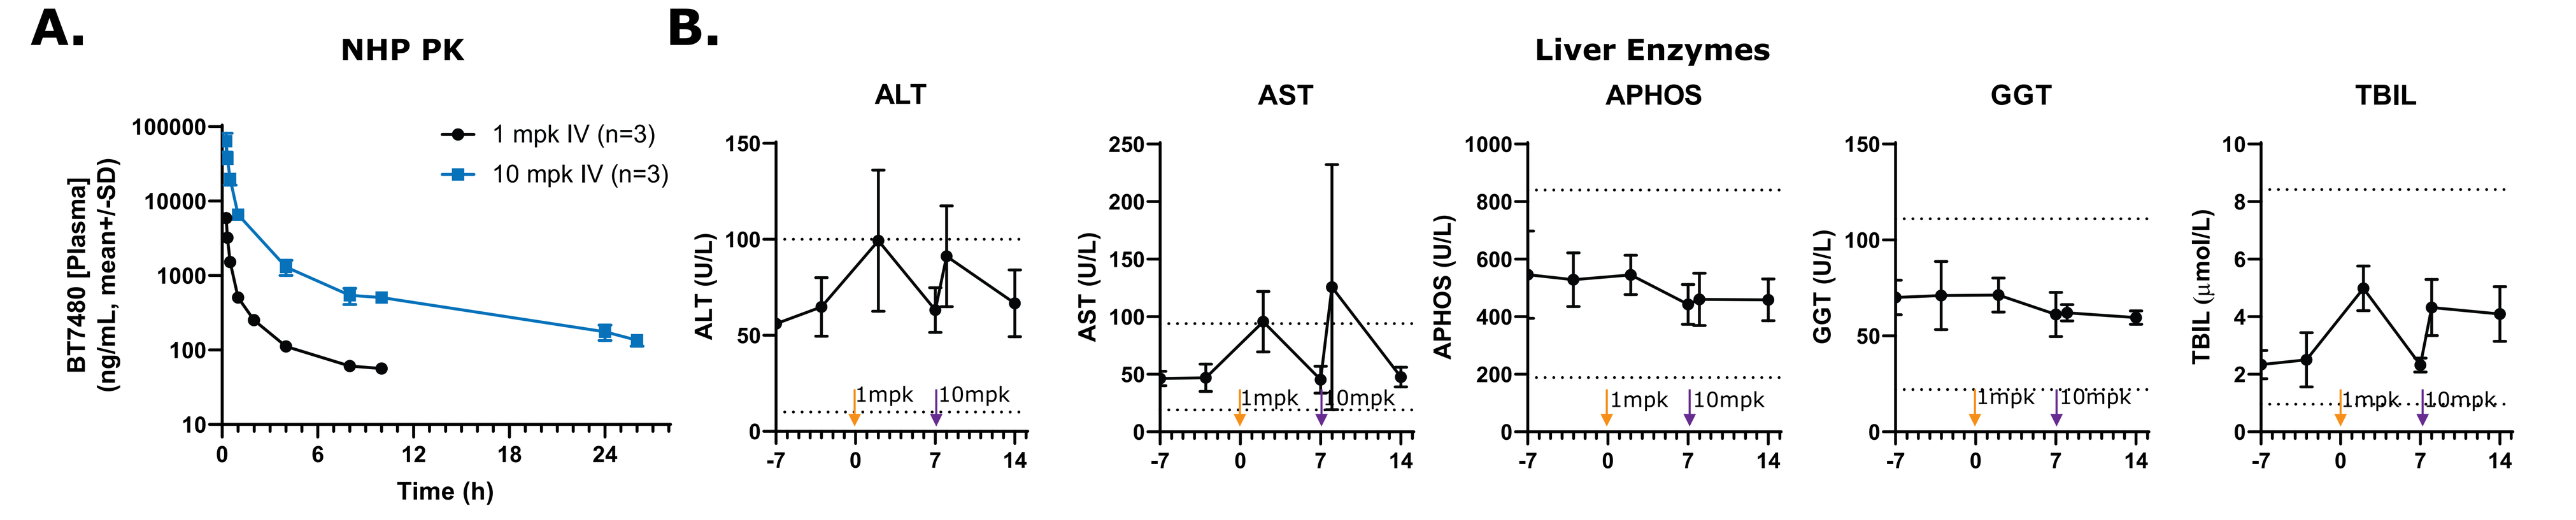
A.
NHP PK
1 mpk IV (n=3)
10 mpk IV (n=3)
BT7480 [Plasma] (ng/mL, mean+/-SD)
Time (h)
B.
Liver Enzymes
ALT
AST
APHOS
GGT
TBIL
ALT (U/L)
AST (U/L)
APHOS (U/L)
GGT (U/L)
TBIL (µmol/L)
1mpk
10mpk

Figure 6: BT7480 demonstrated linear pharmacokinetics and appears well tolerated in NHPs up to 10 mg/kg. A) Animals (n=3 +/- SD) were dosed 1 mg/kg IV on Day 0 and 10 mg/kg on Day 7. Exposures at 10 mg/kg are higher than those predicted to be required for a human efficacious dose. B) Clinical chemistry panel indicated that liver enzymes were generally well within the normal range (indicated by the dotted horizontal lines). The AST level in 1 out of 3 animals rose just above the normal range after the 10 mg/kg dose was administered and quickly recovered. Circulating cytokines were also monitored at 1h and 24h post-dose and were not significantly elevated in response to BT7480.

CONCLUSIONS/SUMMARY

- BT7480 is a Nectin-4/CD137 TICA that represents a new generation of chemically synthetic tumor antigen targeted CD137 agonists.
- BT7480 led to highly potent Nectin-4 dependent activity *in vitro* and *in vivo* inducing complete tumor regressions and resistance to tumor re-challenge with intermittent dosing. BT7480 induced significant modulation of the tumor immune microenvironment including cytotoxic cell infiltration in tumor tissue.
- BT7480 demonstrated linear pharmacokinetics in non-human primates and appears well tolerated at exposures in excess of the predicted efficacious exposure in humans. Further IND-enabling safety studies are ongoing.

References: [1] Hinner et al, Clin Cancer Res 25(19): 5878-89 (2019); [2] Claus et al, *Sci Transl Med* 11(496): eaav5989 (2019); [3] Eskiocak et al, JCI Insight 12; 5(5):e133467 (2020)

Questions? Contact maren.killackey@bicycletx.com

Bicycle Therapeutics
4 Hartwell Place, Lexington, MA 02421
www.bicycletherapeutics.com