In-depth characterization of cDC1 ex vivo and in vitro models: development and comparison of conventional dendritic cell culture systems for industry

Abstract #

▸980

Anna Licht, Hannah Gardner, Tamera Ashworth, Laura L. Goodfield, Johanna Lahdenranta, Kevin McDonnell, Philip E. Brandish, Nicholas Keen

BACKGROUND

Conventional dendritic cells (cDC) are innate immune cells specialized in antigen sampling and subsequent cross-presentation and are critical for an effective anti-tumor immune response. The relatively rare subset cDC type I (cDC1) has been associated with human cancer patient survival, and strategies aimed at increasing their abundance and activation in the tumor microenvironment (TME) are of scientific and therapeutic interest. However, cDC1 constitute less than 0.05% of cells in the blood and are difficult to isolate with high viability and purity, making it a challenge to use these cells in screening assays.

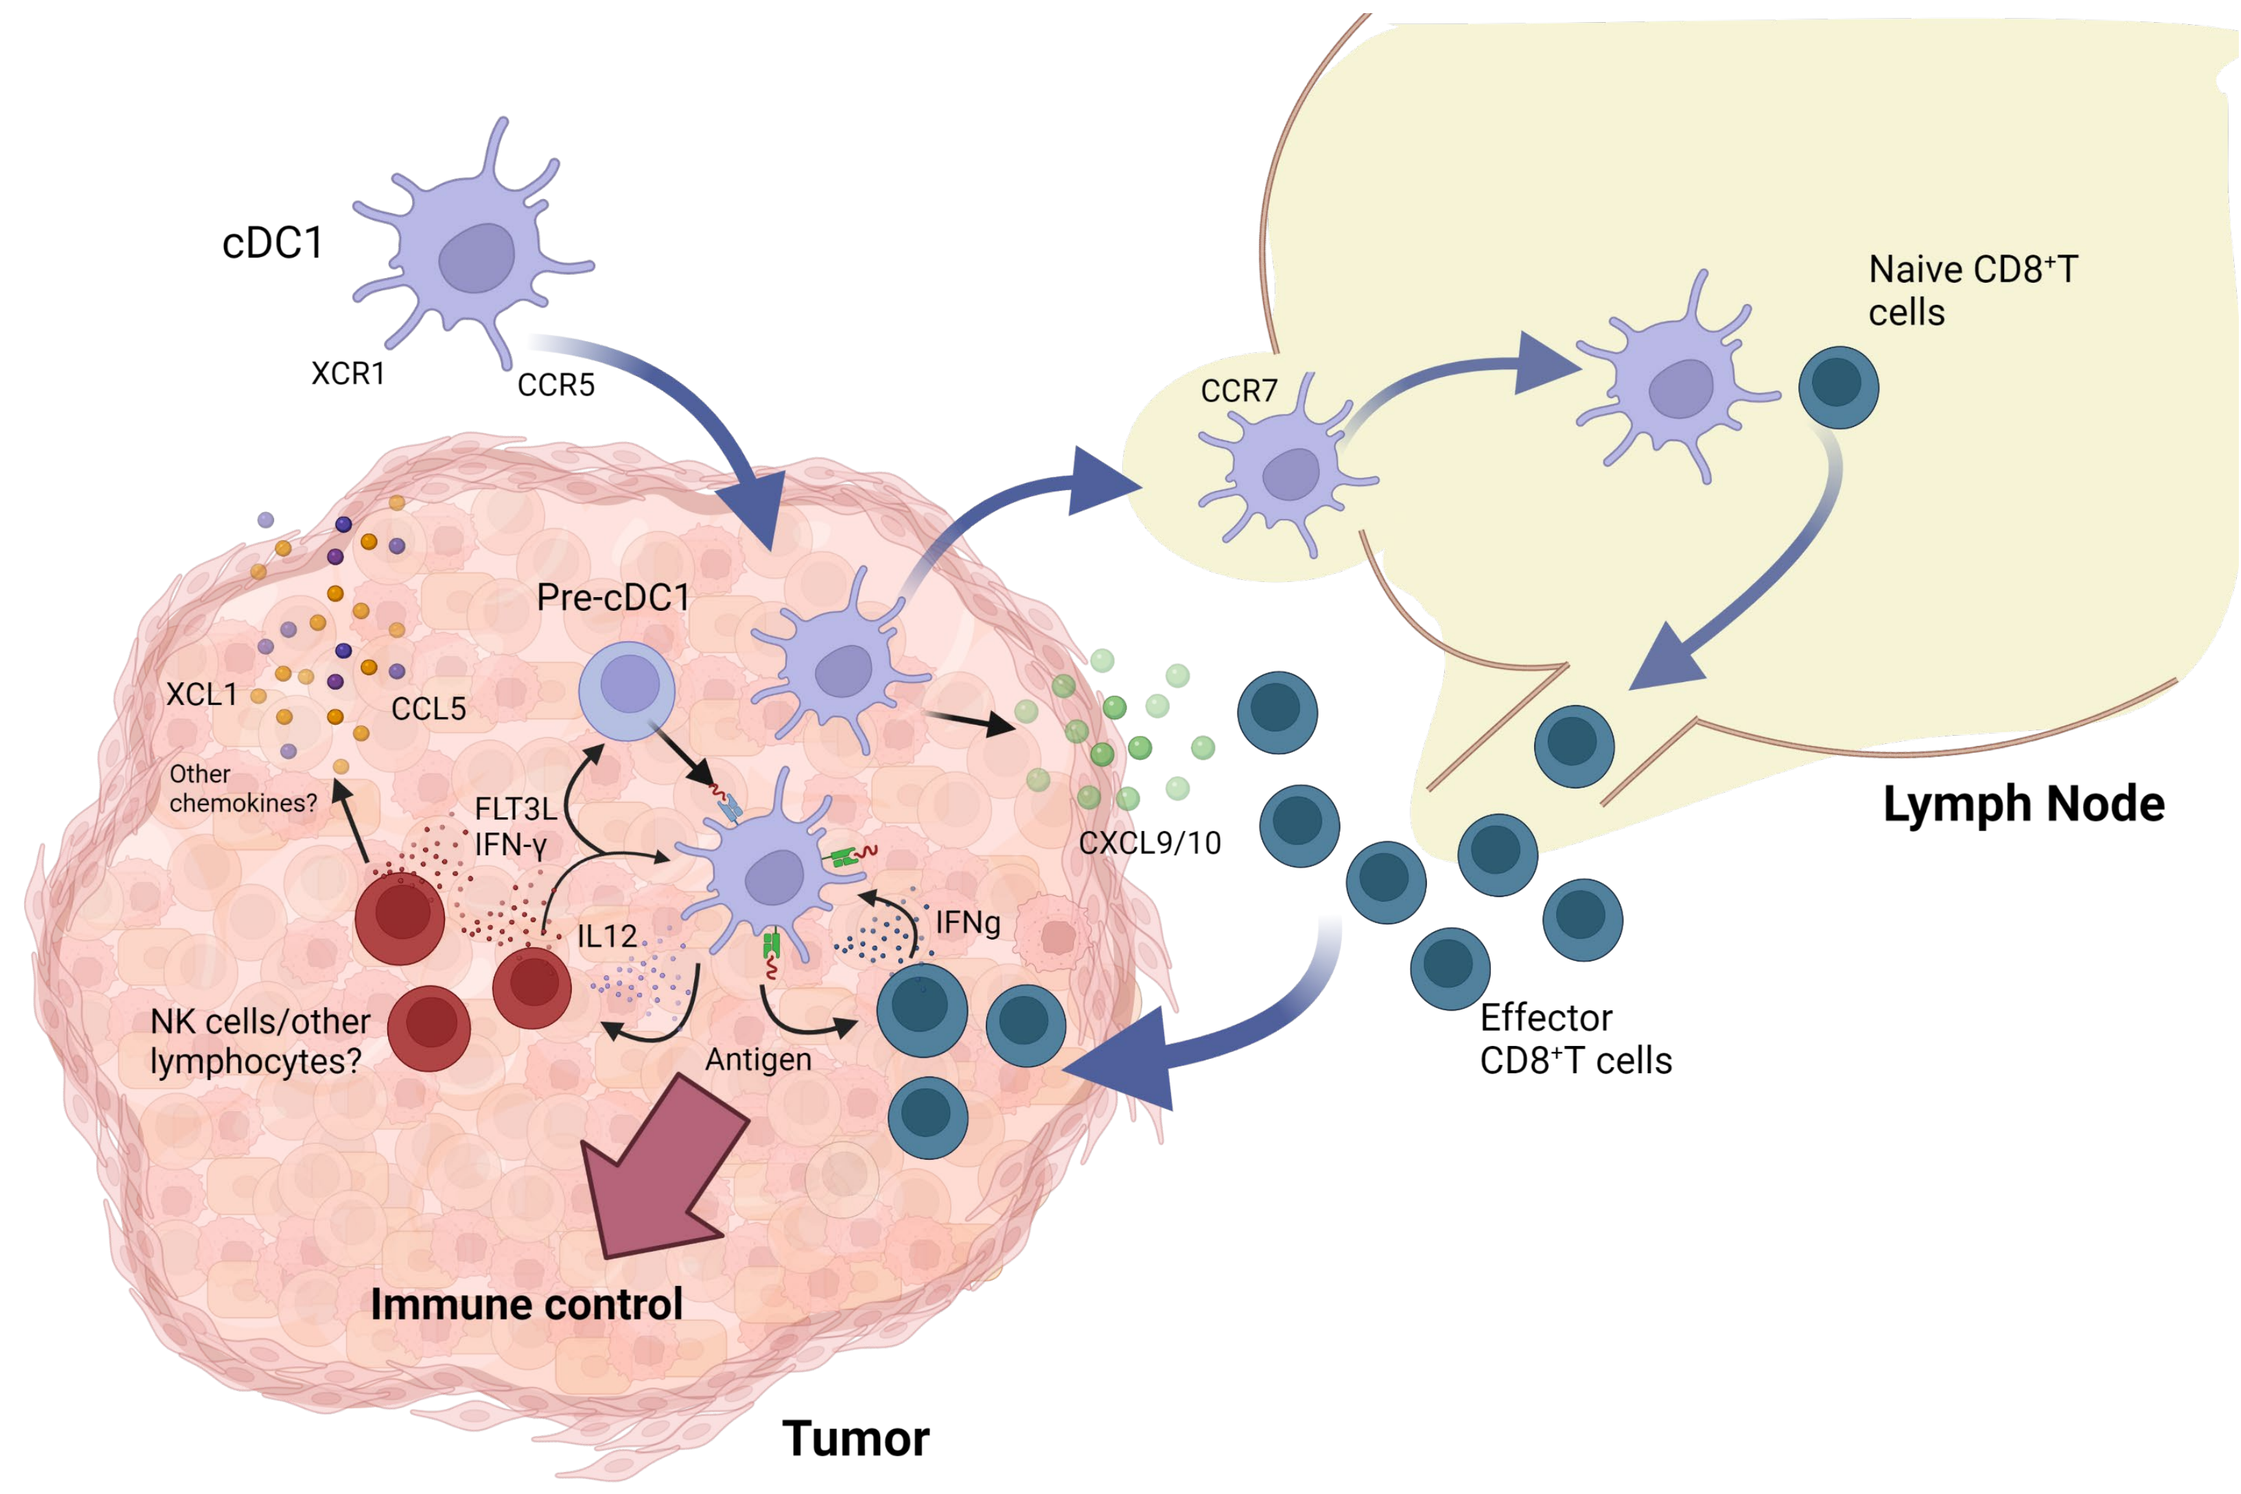
cDC1
XCR1
CCR5
CCR7
Naive CD8⁺T cells
Pre-cDC1
XCL1
CCL5
Other chemokines?
FLT3L
IFN-γ
IL12
IFNg
CXCL9/10
NK cells/other lymphocytes?
Antigen
Lymph Node
Effector CD8⁺T cells
Immune control
Tumor

Figure 1: Adapted from Orchestration of Cancer Immune Control by cDC1 (Böttcher and Reis e Sousa, 2018)[1].

METHODS

We compared two systems that generate a mixed culture containing cDC1 cells from human CD34⁺ hematopoietic stem cells (HSC) isolated from cord blood. In the first system, we developed a scalable in vitro differentiation method that expands and differentiates the CD34⁺ HSC to generate cDC1s over the course of 3.5 weeks. The second system utilized a humanized mouse model; huNOG-EXL mice (CD34⁺ HSC humanized NOG mice, engineered to express human GM-CSF and IL-3 to support the myeloid compartment) were boosted twice with Flt3L-Ig. At day 10 bone marrow was extracted for subsequent culture. Both systems were stimulated with various agonists for 18 h. Cells and supernatant were collected for evaluation of expression and activation.

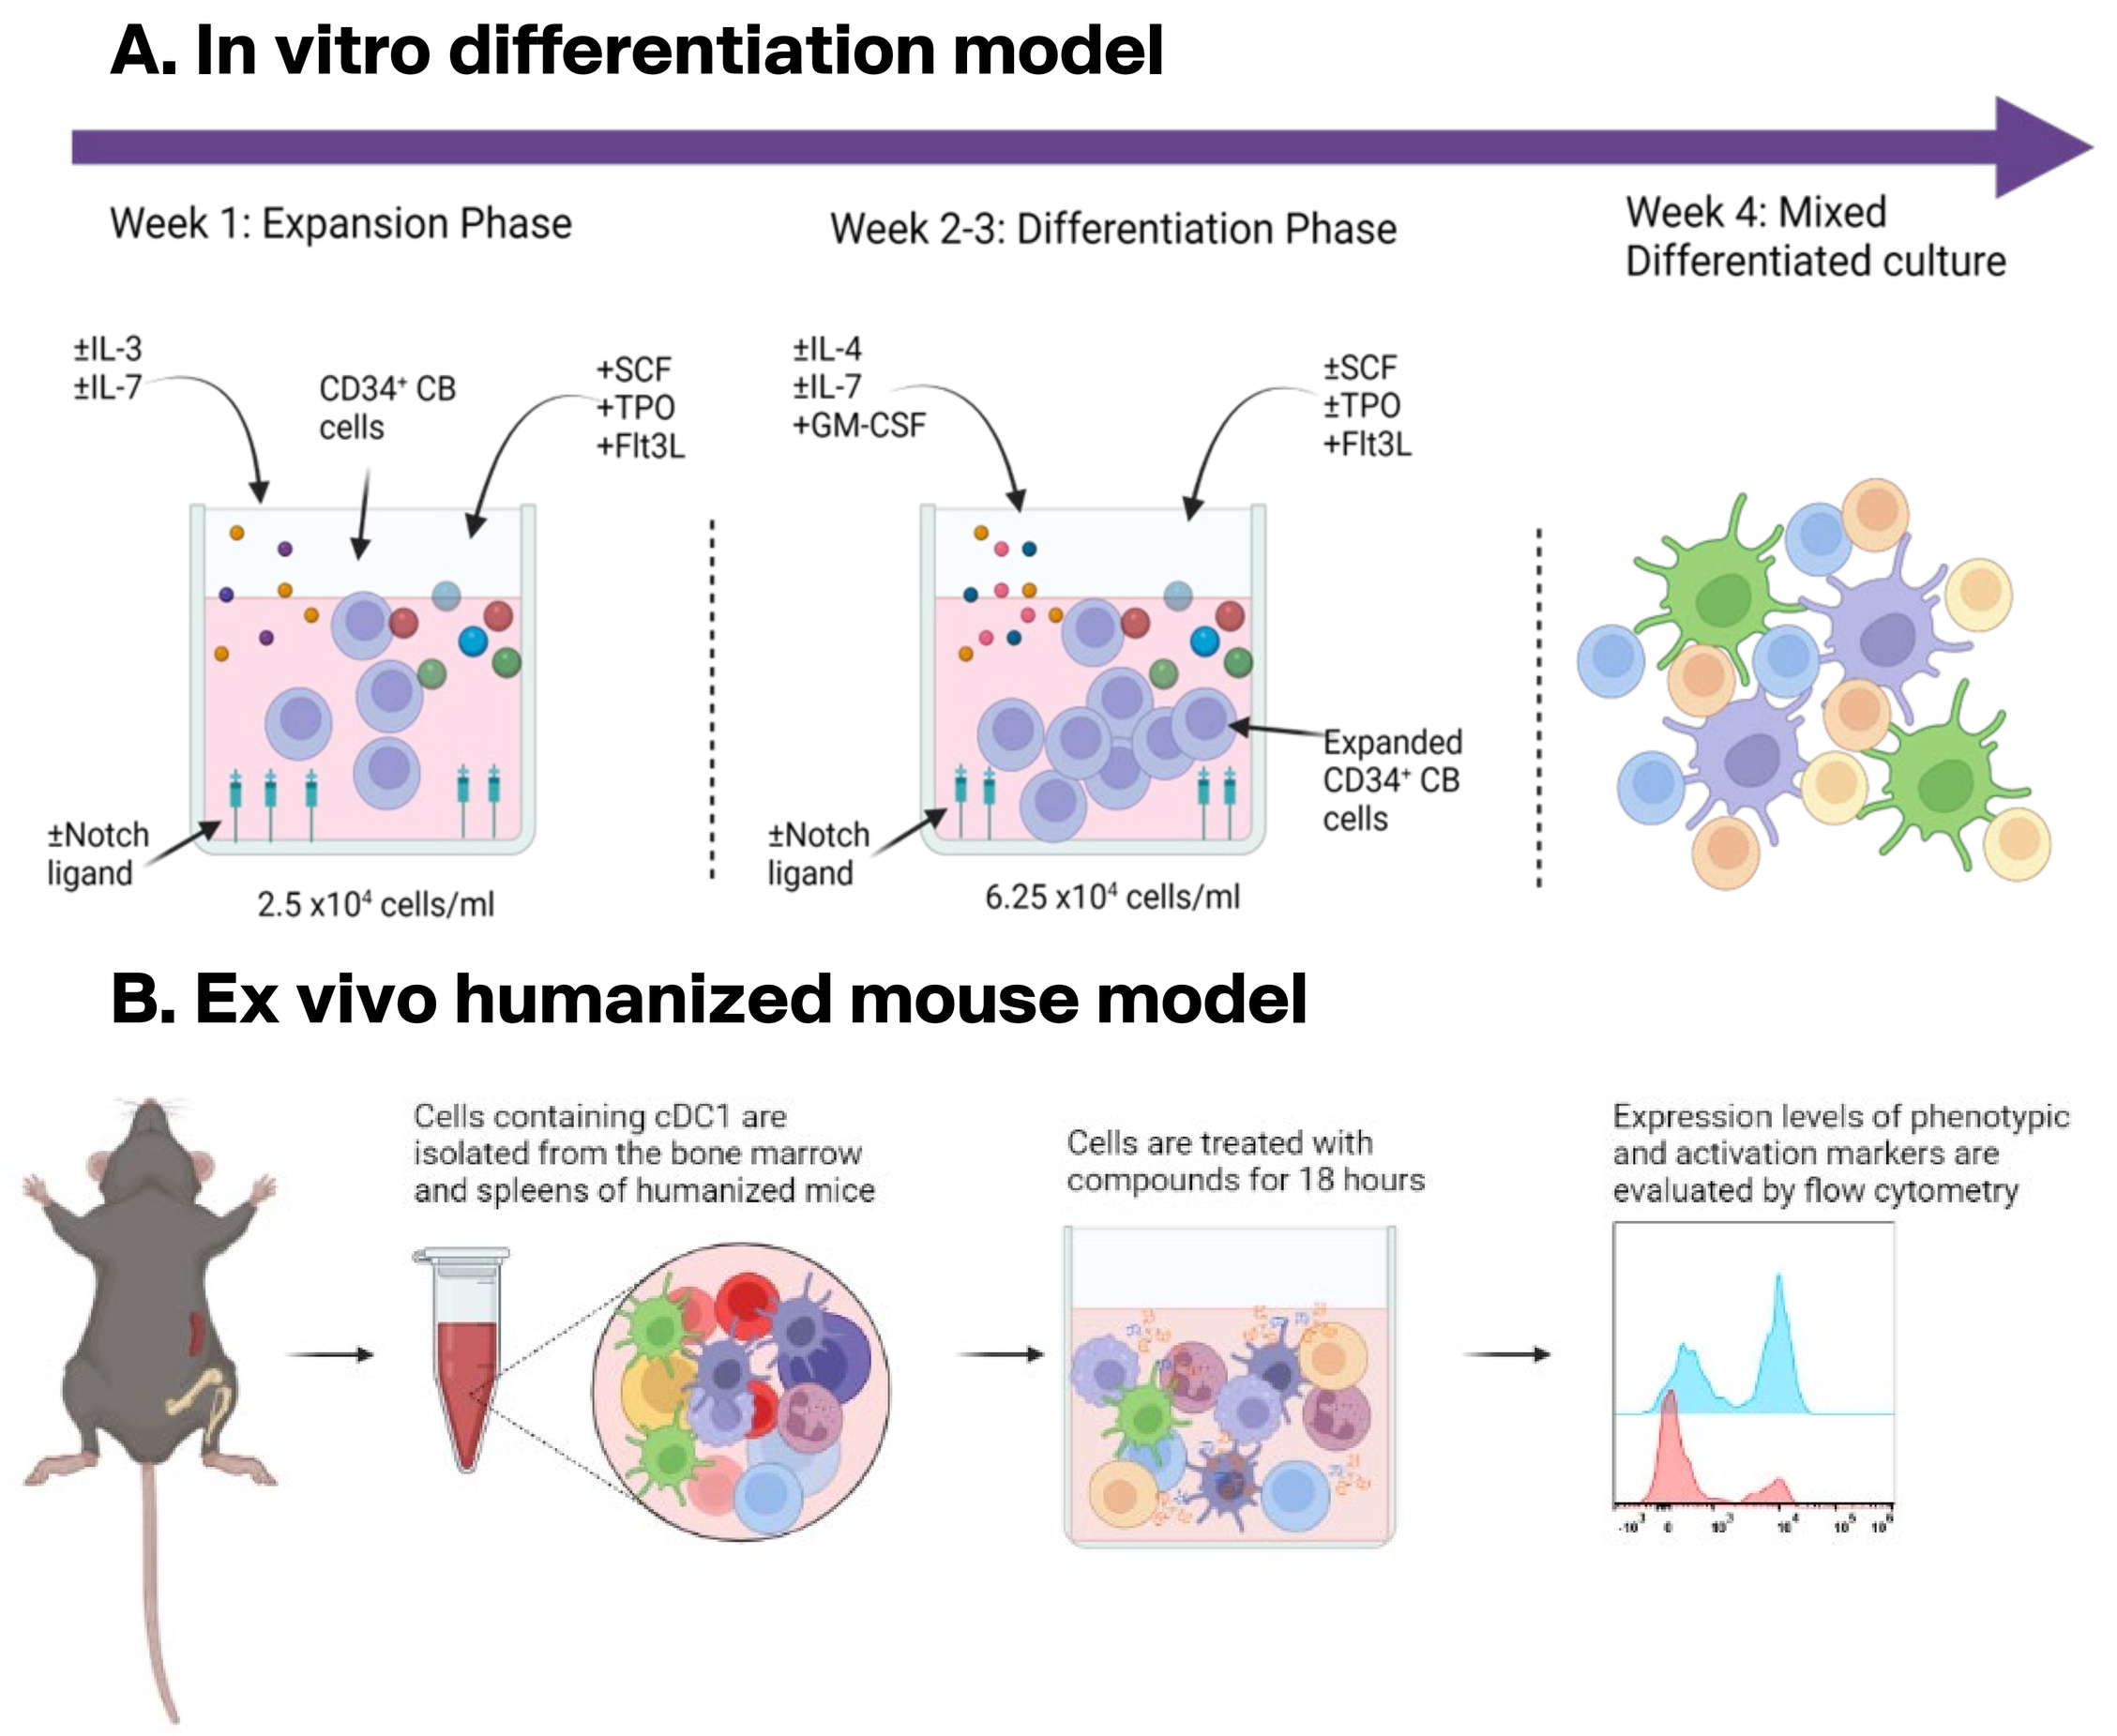
A. In vitro differentiation model
Week 1: Expansion Phase
Week 2-3: Differentiation Phase
Week 4: Mixed Differentiated culture
±IL-3
±IL-7
CD34⁺ CB cells
+SCF
+TPO
+Flt3L
±IL-4
±IL-7
+GM-CSF
±SCF
±TPO
+Flt3L
Expanded CD34⁺ CB cells
±Notch ligand
2.5 x10⁴ cells/ml
±Notch ligand
6.25 x10⁴ cells/ml
B. Ex vivo humanized mouse model
Cells containing cDC1 are isolated from the bone marrow and spleens of humanized mice
Cells are treated with compounds for 18 hours
Expression levels of phenotypic and activation markers are evaluated by flow cytometry

Figure 2: (A) In vitro culture of cDC1 derived from CD34⁺ cord blood cells. Alpha-MEM+Glutamax containing 10%FBS, 1mM Sodium Pyruvate, and 50µM β-mercaptoethanol was used for the expansion phase followed by Alpha-MEM containing 10%FBS for the differentiation phase. Wells were coated with 1µg/mL DLL1 human notch ligand. Cells were cultured at 2.5x10⁴ cells/mL (expansion phase) or 6.25x10⁴ cells/mL (differentiation phase) in media containing specified growth factors. Half media changes were performed 5 days after cells were placed in differentiation media at a 2x cytokine concentration. **(B) Ex vivo culture of cDC1 from humanized mouse.** Human NOG-EXL mice boosted with Flt3L-Ig were used as a source of bone marrow with enriched cDC1s. Bone marrow cells were collected by centrifugation and plated at 0.8x10⁶ cells/mL.

RESULTS

These methods yield an increase in number of cDC1 cells in a mixed cellular population as compared to previous studies using whole blood. The cDC1 cells are phenotypically and functionally similar to those in the blood; they express high levels of CD141 and Clec9a and respond to activating stimuli such as Poly(I:C). The ex vivo cDC1 system using bone marrow from humanized mice is highly reproducible across laboratory operators and with multiple cord blood donors, and it is less laborious than the in vitro differentiation assay (18 hours as compared to the in vitro system which requires 3.5 weeks). Multicolor flow cytometry was used to further phenotype and assess functionality of other cell types present in the ex vivo culture.

Ex vivo and in vitro expression profile of cDC1 cells

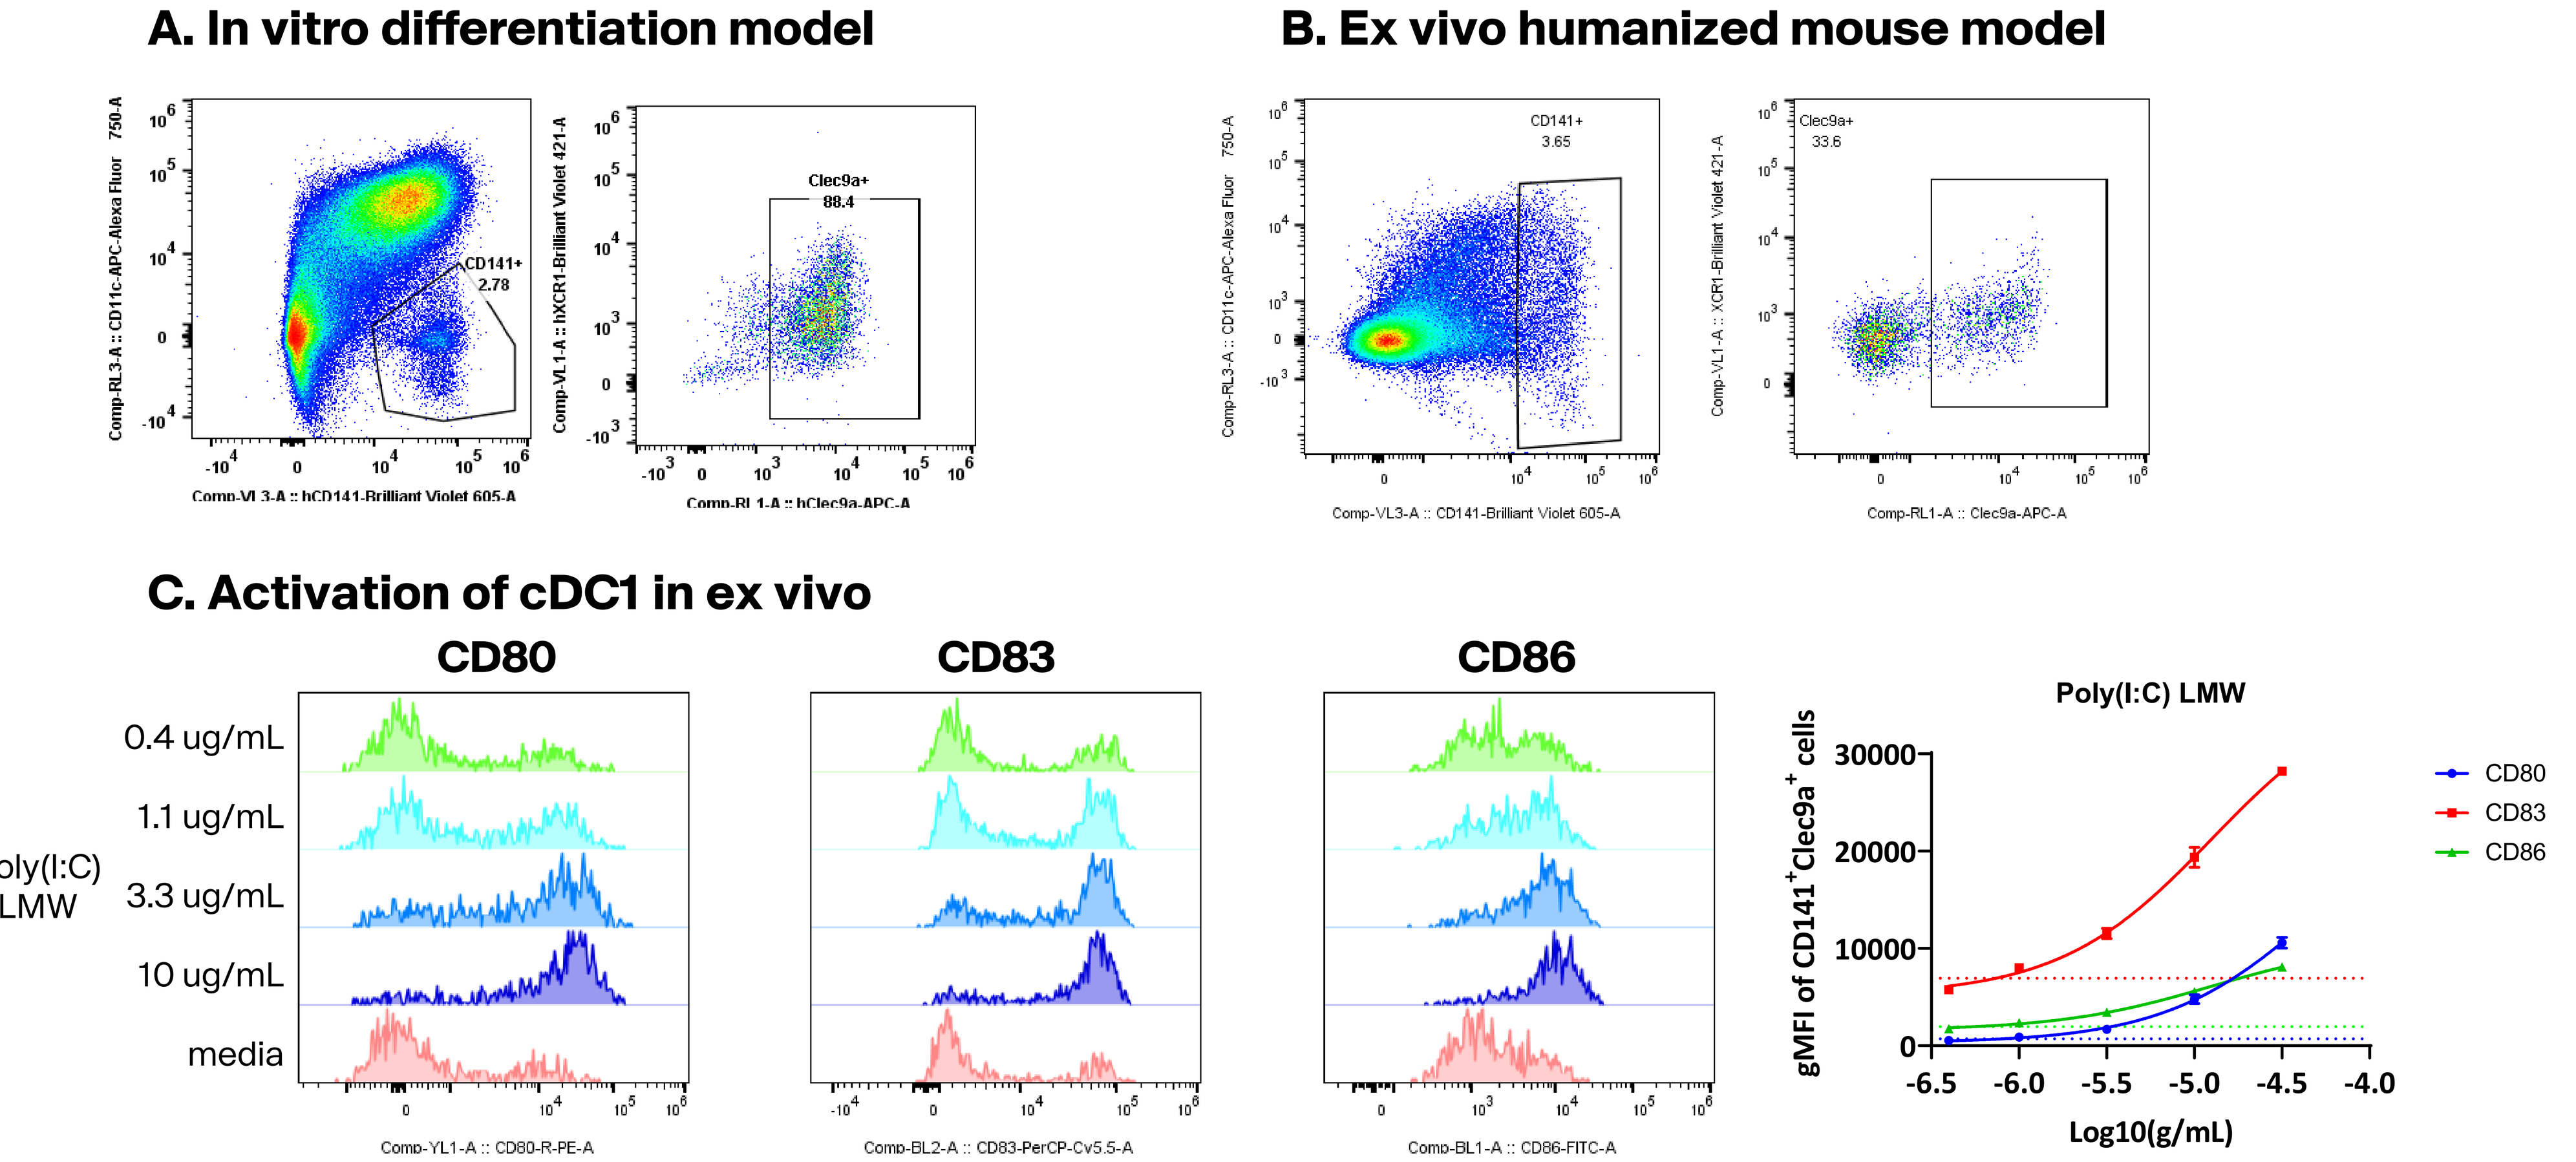
A. In vitro differentiation model
B. Ex vivo humanized mouse model
CD141+ 2.78
Clec9a+ 88.4
CD141+ 3.65
Clec9a+ 33.6
Comp-VL3-A :: hCD141-Brilliant Violet 605-A
Comp-RL1-A :: hClec9a-APC-A
Comp-VL3-A :: CD141-Brilliant Violet 605-A
Comp-RL1-A :: Clec9a-APC-A
C. Activation of cDC1 in ex vivo
CD80
CD83
CD86
Poly(I:C) LMW
0.4 ug/mL
1.1 ug/mL
3.3 ug/mL
10 ug/mL
media
Comp-YL1-A :: CD80-R-PE-A
Comp-BL2-A :: CD83-PerCP-Cv5.5-A
Comp-BL1-A :: CD86-FITC-A
Poly(I:C) LMW
gMFI of CD141⁺Clec9a⁺ cells
Log10(g/mL)
CD80
CD83
CD86

Figure 3: (A, B) Both methods produce cDC1 expressing phenotypic markers CD141 and Clec9a. Cells gated on live, single cell; human CD45⁺ included in gating strategy for mouse model. **(C) Ex vivo cDC1 are functional and respond to agonists**. Activity is defined as upregulation of CD80, CD83, and CD86 over media control baseline in flow cytometry readout. Dotted lines represent media. Data are similar for in vitro assay (not shown).

Characterization of ex vivo myeloid cell populations

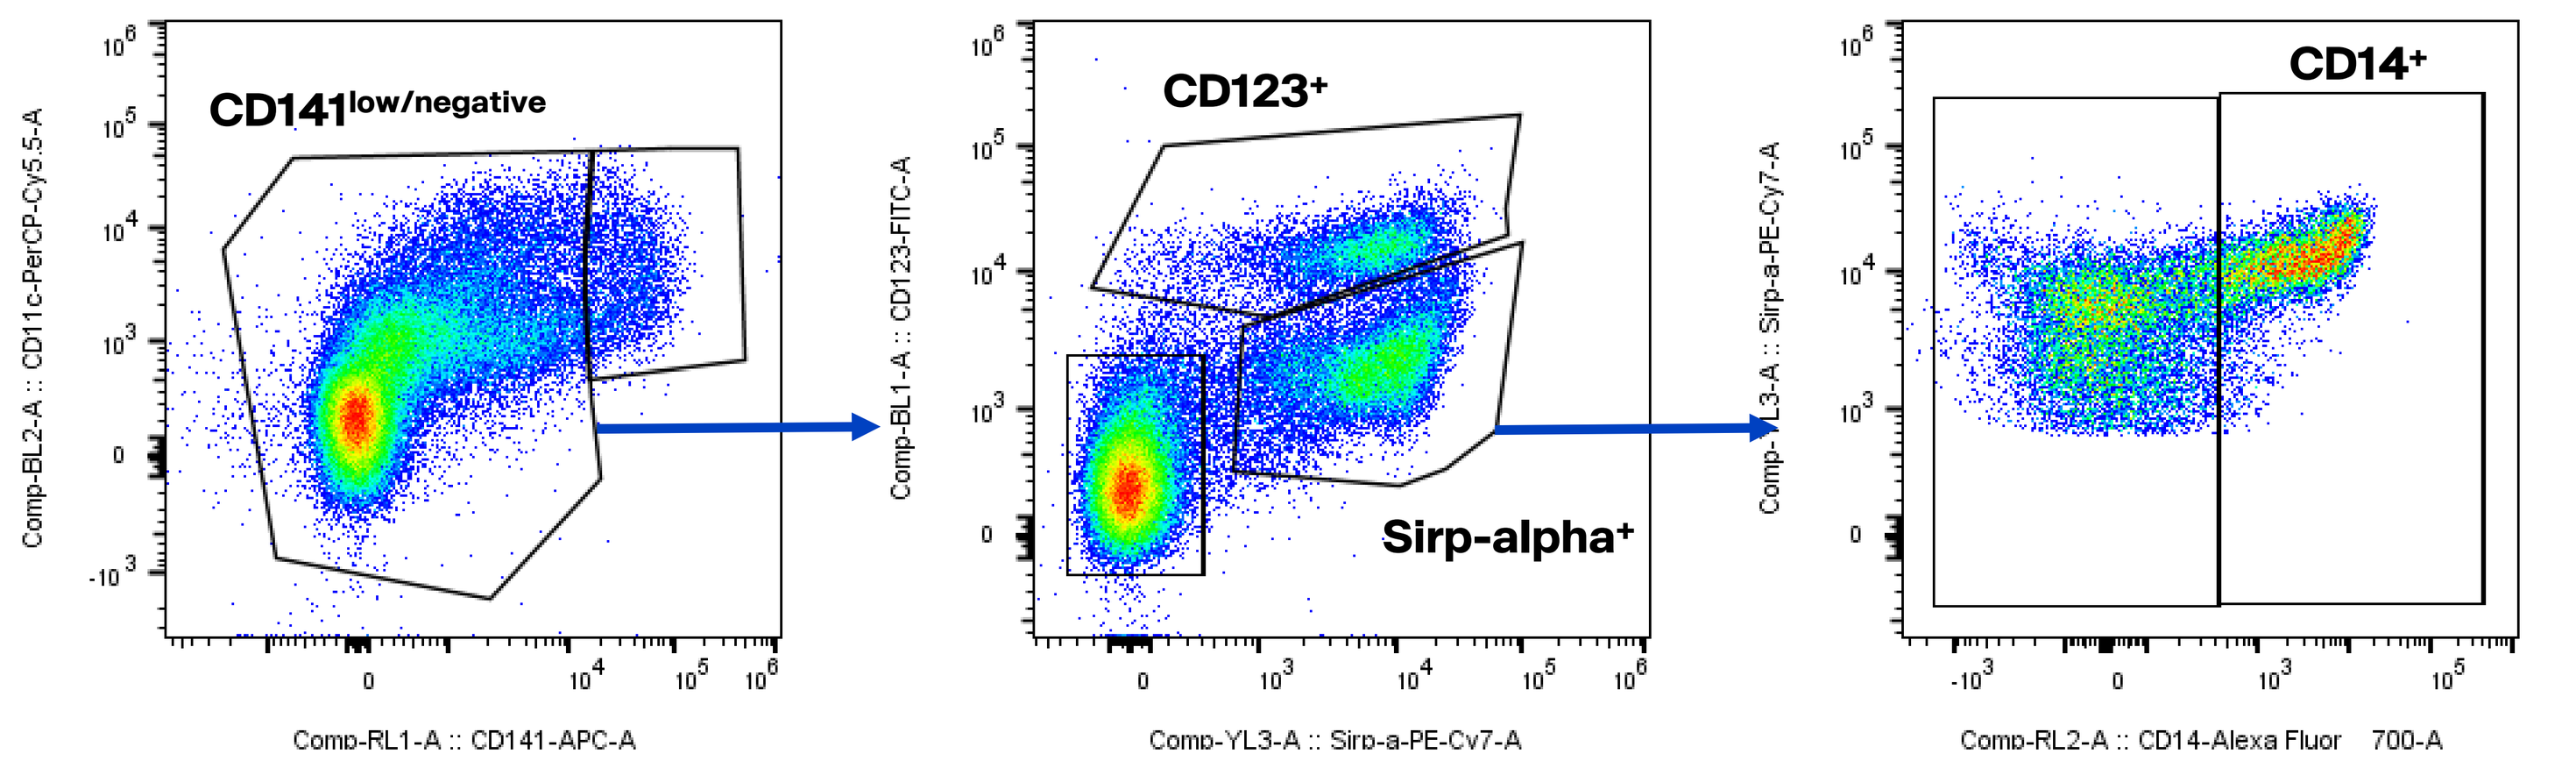
CD141low/negative
CD123⁺
Sirp-alpha⁺
CD14⁺
Comp-RL1-A :: CD141-APC-A
Comp-YL3-A :: Sirp-a-PE-Cv7-A
Comp-RL2-A :: CD14-Alexa Fluor 700-A

Cell type	Marker	TLR expression[2]
cDC1	Clec9a, CD141, BTLA	TLR3 (high expression), TLR9, TLR10
cDC2	sirp-alpha, CD1c	TLR2/6, TLR4, TLR5, TLR8, TLR9
pDC	CD123	TLR7, TLR9

Figure 4: Ex vivo culture was further characterized to demonstrate established myeloid populations. Plasmacytoid DC cells (pDC) are defined as CD123⁺. cDC type II cells (cDC2) are defined as sirp-alpha⁺ CD14⁻.

RESULTS

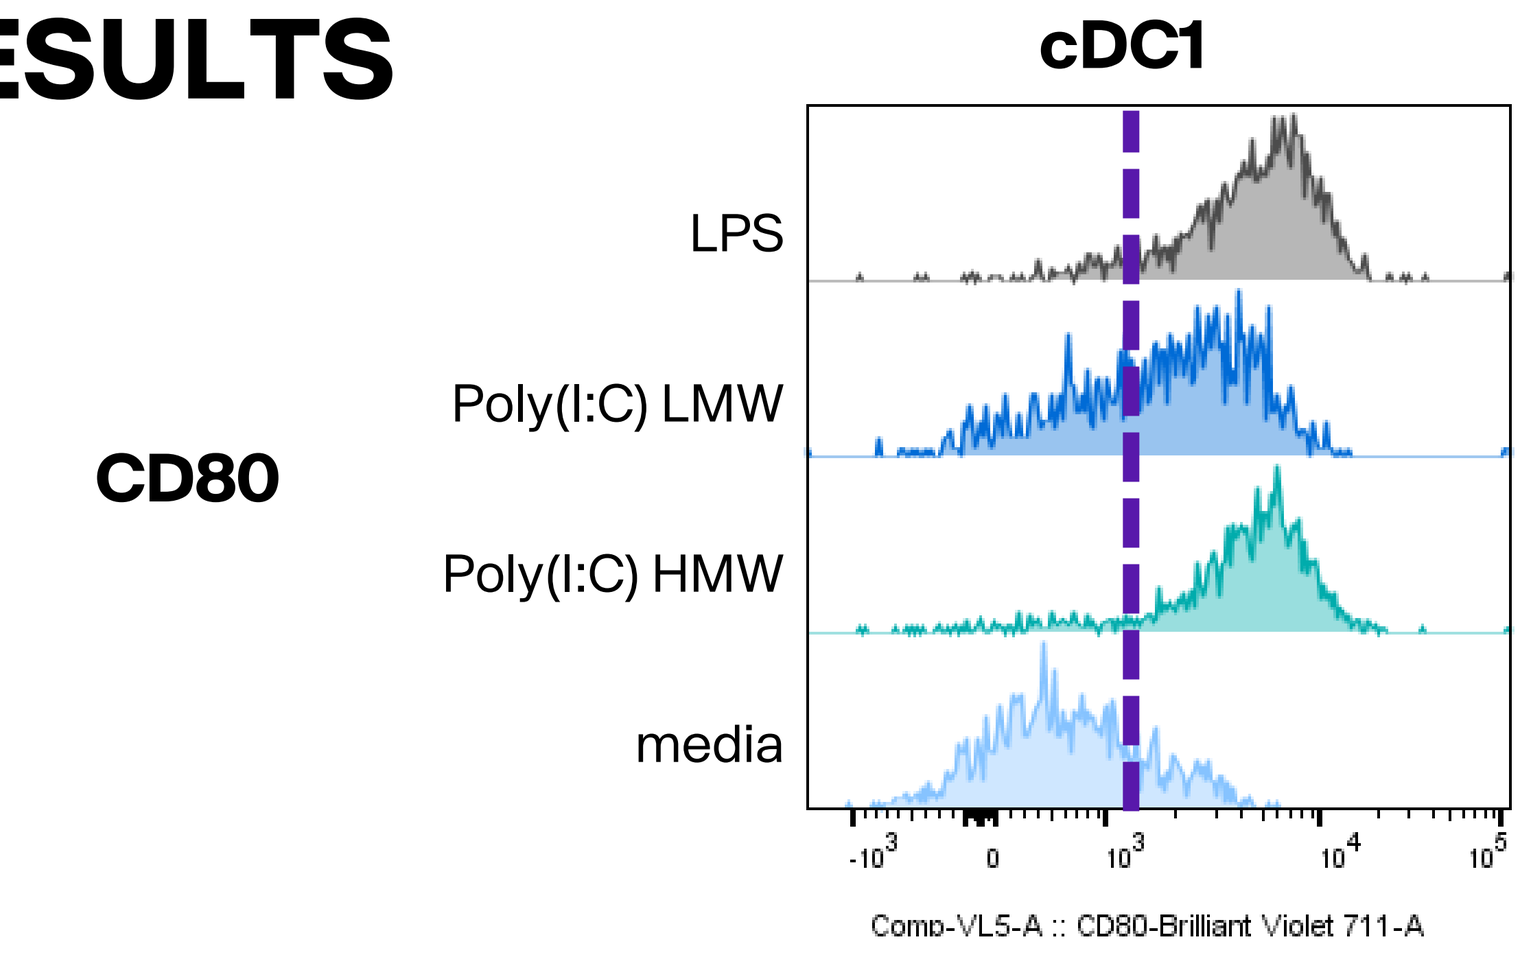
cDC1
pDC
cDC2
CD80
LPS
Poly(I:C) LMW
Poly(I:C) HMW
media
Comp-VL5-A :: CD80-Brilliant Violet 711-A

Figure 5: TLR3 activation differs among DC subtypes. TLR3 strongly activates cDC1 cells, which have high expression of TLR3 (based on literature as compared to other myeloid subsets[3]).

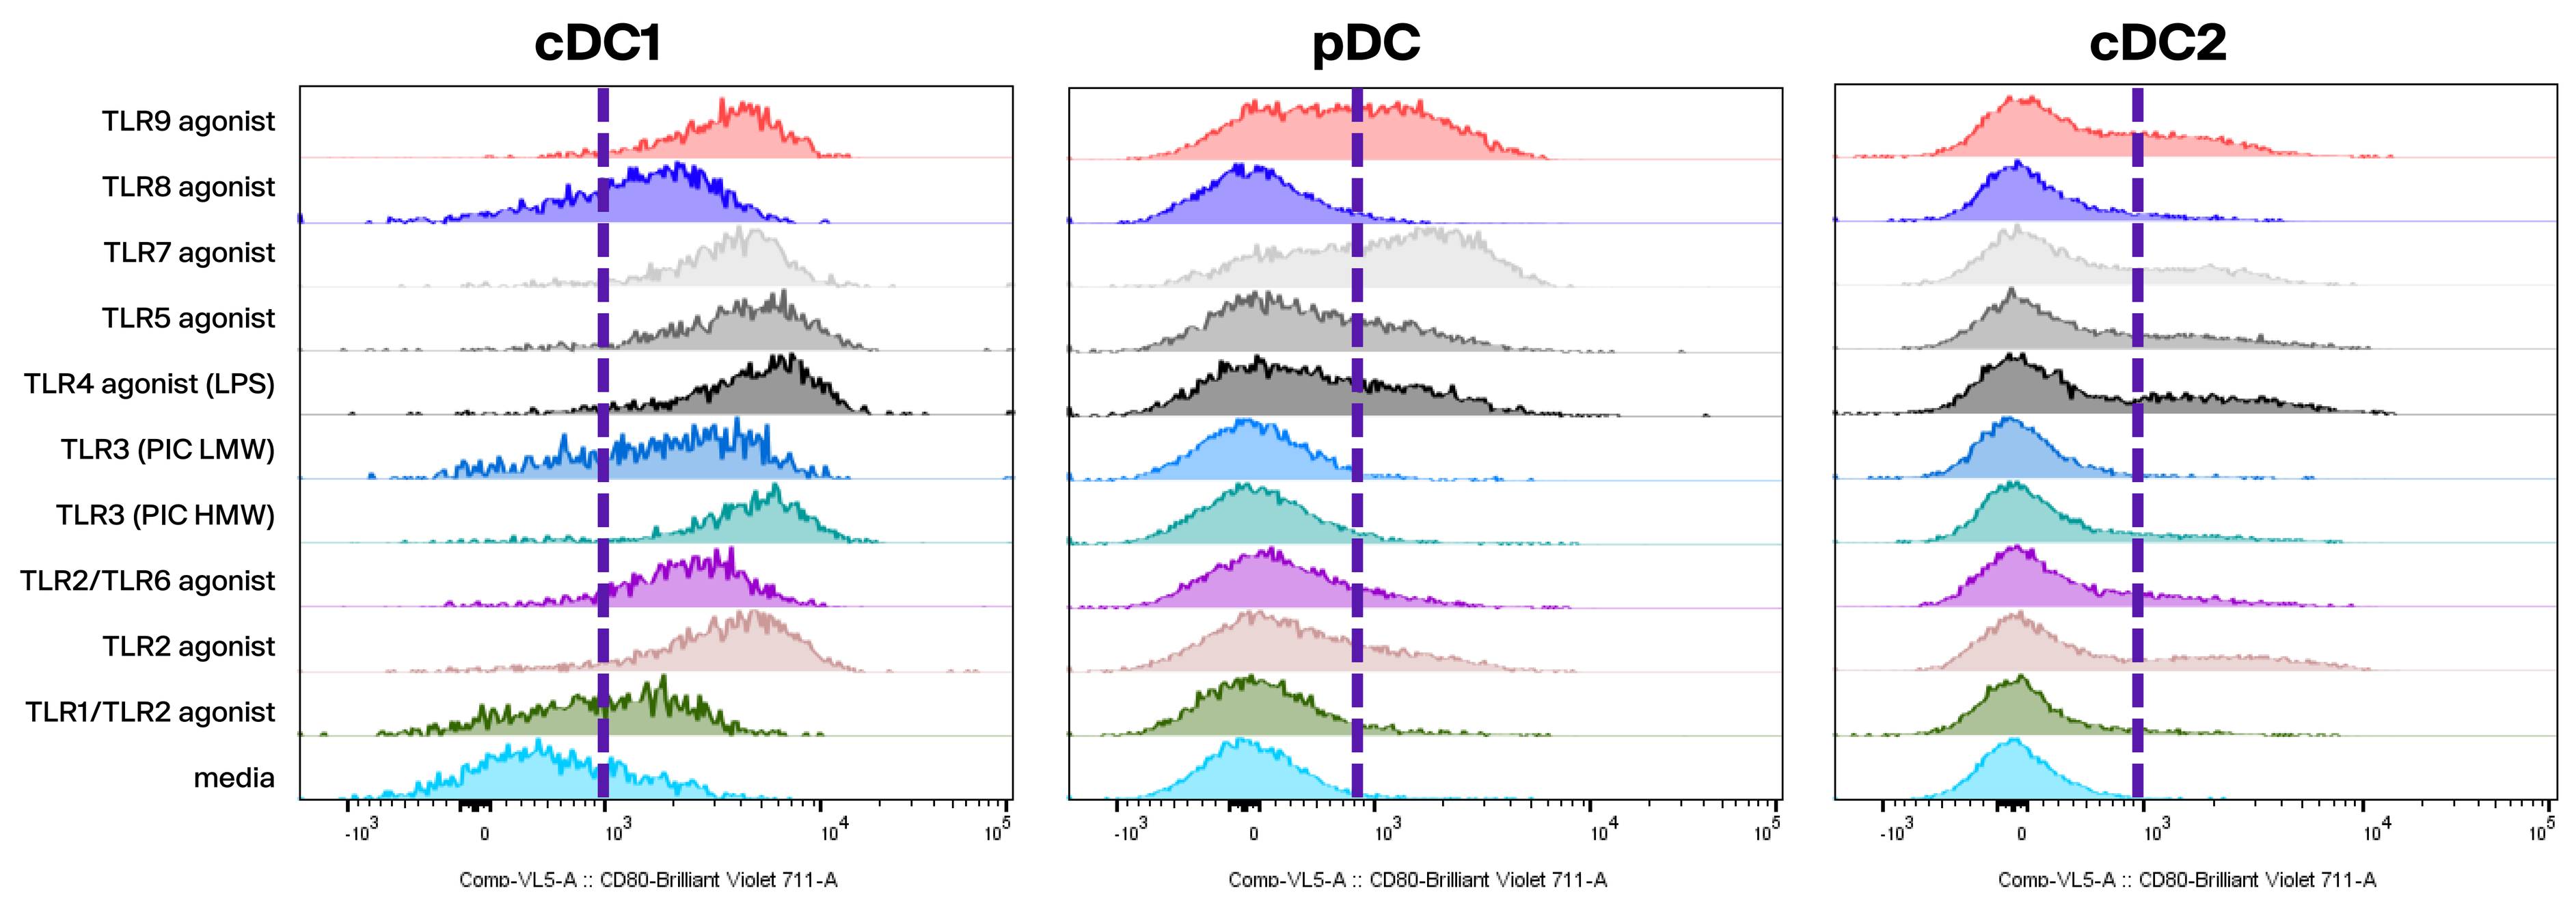
cDC1
pDC
cDC2
TLR9 agonist
TLR8 agonist
TLR7 agonist
TLR5 agonist
TLR4 agonist (LPS)
TLR3 (PIC LMW)
TLR3 (PIC HMW)
TLR2/TLR6 agonist
TLR2 agonist
TLR1/TLR2 agonist
media
Comp-VL5-A :: CD80-Brilliant Violet 711-A

Figure 6: Myeloid cells from ex vivo cultures are functional and respond to various TLR agonists. cDC1 are uniquely and strongly activated by TLR3 agonists. pDCs are activated by TLR7 and TLR9 agonists. cDC2 respond to multiple agonists, especially TLR2/6, TLR5, TLR9. Agonists shown above were obtained from InvivoGen TLR1-9 agonist kit and tested at manufacturer's recommended dose (Catalogue #tlrl-kit1hw).

CONCLUSIONS

Both systems demonstrate that HSC-derived cDC1 cultures can be reliably generated with cDC1 phenotype and functionality and may be used for screening assays in industry settings. The ex vivo system allows for a faster and higher throughput but requires supply of humanized mice. The in vitro model system is lower throughput but is a fully human system that does not require access to humanized mice. The ex vivo model also shows a diverse and competent myeloid repertoire, with specificity for TLR agonists.

ACKNOWLEDGEMENTS

We thank Aditya Murtha, Christine Moussion, Markus Brown, Chris Kemball and Dongping He (Genentech) for guidance and helpful discussions on initiating the culture system and Chinmayee Shah (Bicycle) for artwork.

REFERENCES

1. Böttcher and Reis e Sousa, Trends in Cancer. 4:784-792 (2018).
2. Collin and Bigley. Immunology.154(1):3-20 (2018).
3. Cabeza-Cabrerizo, et al. Annual Review of Immunology. 39:1, 131-166 (2021).
4. Images created with BioRender.com

Bicycle Therapeutics, Inc.
4 Hartwell Place
Lexington, MA
02421-3122, USA
T. +1 617-945-8155

BicycleTx Limited
Portway Building
Granta Park
Cambridge
CB21 6GS, UK
T. +44 (0)1223 261503

bicycletherapeutics.com