Identify optimized dosage for zelenectide pevedotin in locally advanced/metastatic urothelial carcinoma using quantitative analyses

Abstract #

4567

Yasong Lu, PhD[1]; Justin Bader, MBA, PharmD[1]; Cong Xu, PhD[2]; Lei Xu, PhD[1]; Leslie DeMars, MD[1]; Michael Method, MD, MBA, MPH[1]; Eric Westin, MD[1]; Hongmei Xu, PhD[1]

[1]Bicycle Therapeutics, Lexington, MA; [2]Bicycle Therapeutics, Lexington, MA (affiliation at the time of analysis)

INTRODUCTION

- Zelenectide pevedotin (zele; BT8009) is a Bicycle® Drug Conjugate (BDC®) comprising a highly selective bicyclic peptide targeting Nectin-4 linked to the cytotoxin monomethyl auristatin E (MMAE) via a cleavable linker (**Figure 1**)[1]
- Zele demonstrated antitumor activity and a generally tolerable safety profile in patients with locally advanced/metastatic urothelial carcinoma (la/mUC) in the Phase 1/2 Duravelo-1 trial (NCT04561362)[2,3]
- In a randomized Phase 2 study, zele is evaluated at 2 dosages: 5 mg/m² on Days (D)1/8/15 and 6 mg/m² on D1/8 of a 21-D cycle, each as monotherapy or combined with pembrolizumab in patients with la/mUC (NCT06225596, Duravelo-2)[4]
 - After ≥27 weeks follow-up in 30 patients per dosage arm (N=120 total), an interim analysis was conducted to select the optimized dosage[5]

FIGURE 1. ZELENECTIDE PEVEDOTIN STRUCTURE[6]

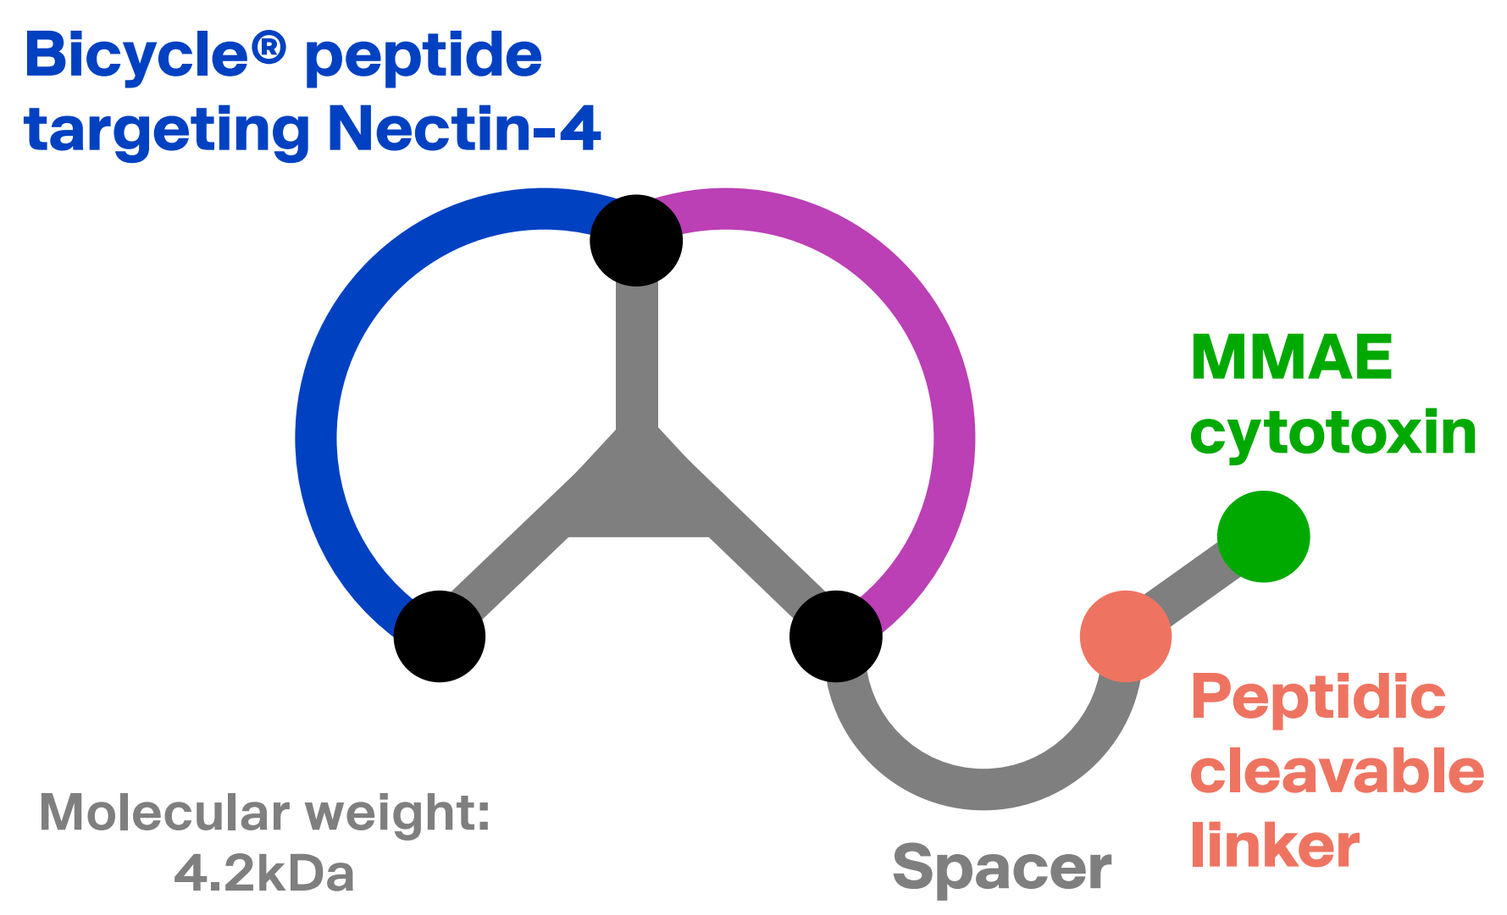
Bicycle® peptide targeting Nectin-4
MMAE cytotoxin
Peptidic cleavable linker
Spacer
Molecular weight: 4.2kDa

OBJECTIVE

- To comprehensively quantify the benefit-risk profile of each zele dosage by pharmacometric (PMx) and statistical analyses for optimized dosage selection for patients with la/mUC

METHODS

PMx MODELING

- Clinical data (pharmacokinetics [PK], efficacy, safety, laboratory values, and demographic data) from Duravelo-1 and Duravelo-2 were pooled and analyzed by using population PK (PopPK) and exposure-response modeling (**Figure 2**)
 - **PopPK:** PopPK models for zele and MMAE were developed by using plasma PK concentrations
 - **Exposure-Efficacy:** confirmed overall response rate (ORR), assessed by investigator and/or blinded independent central reviewer (BICR), was modeled using logistic regression in MMAE-naive patients with la/mUC who were treated with zele, had ≥1 adequate post-baseline response assessment, and whose response was not non-evaluable
 - **Exposure-Safety:** Grade ≥3 zele-related adverse events (TRAEs), Grade ≥3 neutropenia, Grade ≥2 gastrointestinal disorders, Grade ≥2 peripheral neuropathy, any-grade skin reactions, and dose modifications were modeled using logistic regression analysis
- Simulations for ORR and adverse event (AE) probabilities were generated to compare dosages with a balance of relevant patient characteristics and therapy types (monotherapy or combination with pembrolizumab)

UTILITY SCORE ANALYSIS

- The utility score analysis was mainly based on Grade ≥3 TRAEs and ORR by BICR regardless of confirmation in Duravelo-2, with sensitivity analyses based on alternative safety and/or efficacy endpoints (confirmed ORR by BICR)

FIGURE 2. PMx ANALYSIS WORKFLOW

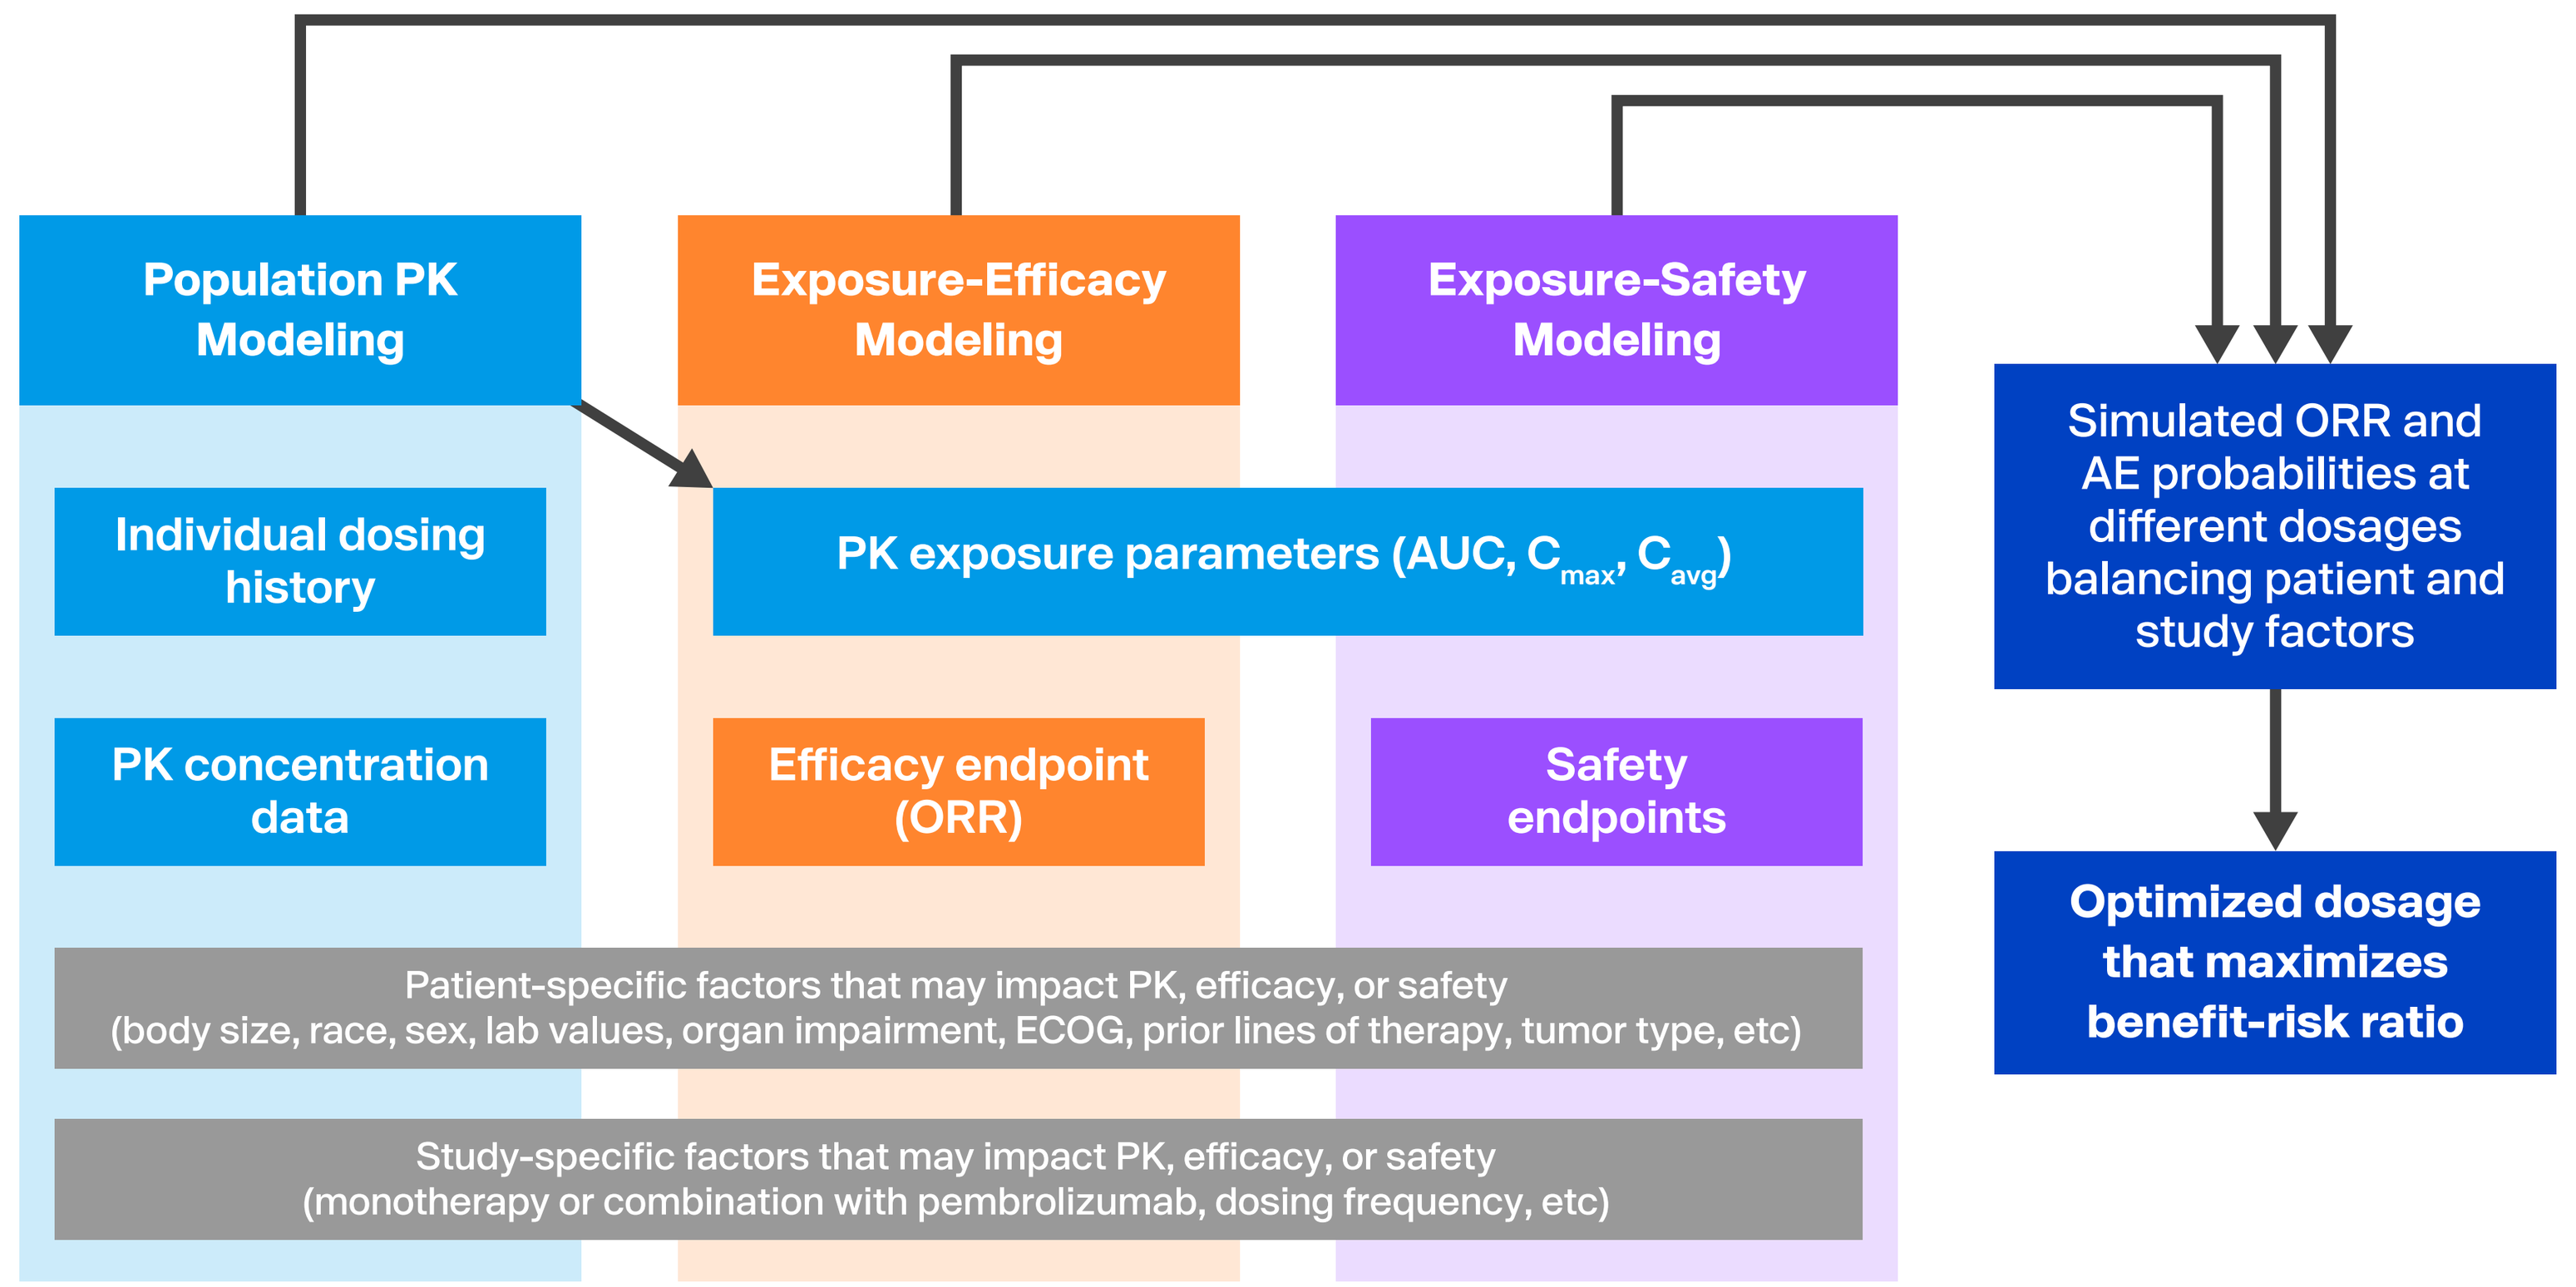
Population PK Modeling
Exposure-Efficacy Modeling
Exposure-Safety Modeling
Individual dosing history
PK exposure parameters (AUC, C_{max}, C_{avg})
PK concentration data
Efficacy endpoint (ORR)
Safety endpoints
Patient-specific factors that may impact PK, efficacy, or safety
(body size, race, sex, lab values, organ impairment, ECOG, prior lines of therapy, tumor type, etc)
Study-specific factors that may impact PK, efficacy, or safety
(monotherapy or combination with pembrolizumab, dosing frequency, etc)
Simulated ORR and AE probabilities at different dosages balancing patient and study factors
Optimized dosage that maximizes benefit-risk ratio

RESULTS

TABLE 1. BASELINE CHARACTERISTICS OF PATIENTS IN SAFETY ANALYSIS POPULATION

Characteristic	Zele Monotherapy (N=305)	Combination With Pembrolizumab (N=89)
Median age (range), years	63 (25–84)	71 (29–87)
Sex, n (%)		
Male	144 (47.2)	63 (70.8)
Female	161 (52.8)	26 (29.2)
Race, n (%)		
White	162 (53.1)	68 (76.4)
Asian	22 (7.2)	13 (14.6)
Other/unknown	121 (39.7)	8 (9.0)
ECOG PS, n (%)		
0	129 (42.3)	40 (44.9)
1	176 (57.7)	38 (42.7)
2	0	11 (12.4)
BSA, median (range), m²	1.80 (1.32–2.54)	1.86 (1.35–2.31)
Liver metastases, n (%)	89 (29.2)	20 (22.5)

DATASET AND PATIENT CHARACTERISTICS

- Data were collected for the PopPK analysis from 430 patients from Duravelo-1 and Duravelo-2 trials
 - PK data from Duravelo-1 was available for 313 patients
 - PK data from Duravelo-2 was available for 117 patients
- Data for exposure-efficacy modeling was available for 203 patients (investigator assessed) and 114 patients (BICR)
- Data for exposure-safety modeling was available for 394 patients
- Data were collected over a wide dose range of 2.5 mg/m² to 10 mg/m² given QW-Q2W, with most data from 5 mg/m² QW and 6 mg/m² on D1/8 (Duravelo-2 doses were limited to 5 mg/m² QW and 6 mg/m² on D1/8)
- Baseline characteristics in the safety analysis dataset are reported in **Table 1**
 - Most patients had urothelial or bladder cancer (60.2%, n=237); other cancer types were all from Duravelo-1 (17.5% [n=69] breast cancer, 15.7% [n=62] non-small cell lung cancer, and 6.6% [n=26] other)

TABLE 2. CONFIRMED ORR IN EXPOSURE-EFFICACY ANALYSIS DATASET

Study	Assessment	Zele Monotherapy, n/N (ORR %)		Combination With Pembrolizumab, n/N (ORR %)	
		5 mg/m² QW	6 mg/m² D1/8	5 mg/m² QW	6 mg/m² D1/8
Duravelo-1	**Investigator**	17/52 (32.7)	–	13/23 (56.5)	–
Duravelo-2	**Investigator**	11/29 (37.9)	7/29 (24.1)	19/28[a] (67.9)	17/28[b] (60.7)
	BICR	8/29 (27.6)	9/29 (31.0)	16/28[a] (57.1)	16/28[b] (57.1)

[a]One patient who was not administered with zele and one with best overall response of NE were excluded. [b]Two patients with best overall response of NE were excluded from the exposure-efficacy analysis.

TABLE 3. TRAE RATES IN PATIENTS TREATED WITH ZELE

Endpoints, n/N (%)	Zele Monotherapy		Combination With Pembrolizumab	
	5 mg/m² QW	6 mg/m² D1/8[a]	5 mg/m² QW	6 mg/m² D1/8
Grade ≥3 TRAE	**62/187 (33.2)**	37/84 (44.0)	30/59 (50.8)	**12/30 (40.0)**
Grade ≥2 GI disorder	27/187 (14.4)	12/84 (14.3)	14/59 (23.7)	**4/30 (13.3)**
Grade ≥3 neutropenia	**19/187 (10.2)**	17/84 (20.2)	9/59 (15.3)	5/30 (16.7)
Grade ≥2 peripheral neuropathy	36/187 (19.3)	**2/29 (6.9)**	22/59 (37.3)	**5/30 (16.7)**
Any-grade skin reactions	**28/187 (15.0)**	23/84 (27.4)	22/59 (37.3)	**8/30 (26.7)**
Dose modifications due to AEs	130/187 (69.5)	**47/84 (56.0)**	53/59 (89.8)	**21/30 (70.0)**

Bold values indicate the favored dose between the 2 options in each category. [a]Due to immaturity of Part B-8 and B-9 results from Duravelo-1, safety data were excluded if patient was ongoing but treated for less than 18 weeks. B-8 and B-9 are excluded from assessment of peripheral neuropathy.

PMx MODELING & SIMULATIONS

- No significant exposure-response relationship was identified for ORR (**Figure 3**)
 - A slight positive trend in ORR vs MMAE area under the curve (AUC) and average concentration (C_{avg}) was observed (P<0.1)
- Simulated ORR was similar between 5 mg/m² and 6 mg/m² dosages (**Table 4**)
- Significant exposure-safety relationships were identified for all the analyzed TRAEs; the majority of AEs were correlated with C_{avg} of zele and MMAE (**Figure 4**)
 - Safety favored 6 mg/m² D1/8 as its predicted AE probabilities were mostly lower and associated with greater magnitudes of risk reduction in endpoints where it was favored
 - For AEs favoring 6 mg/m² D1/8, predicted probabilities were lower by 2.6% to 15.5% as monotherapy, 3.3% to 19.1% in combination (**Table 4**)

FIGURE 3. NO SIGNIFICANT EXPOSURE-ORR RELATIONSHIP IDENTIFIED FROM LOGISTIC MODELING

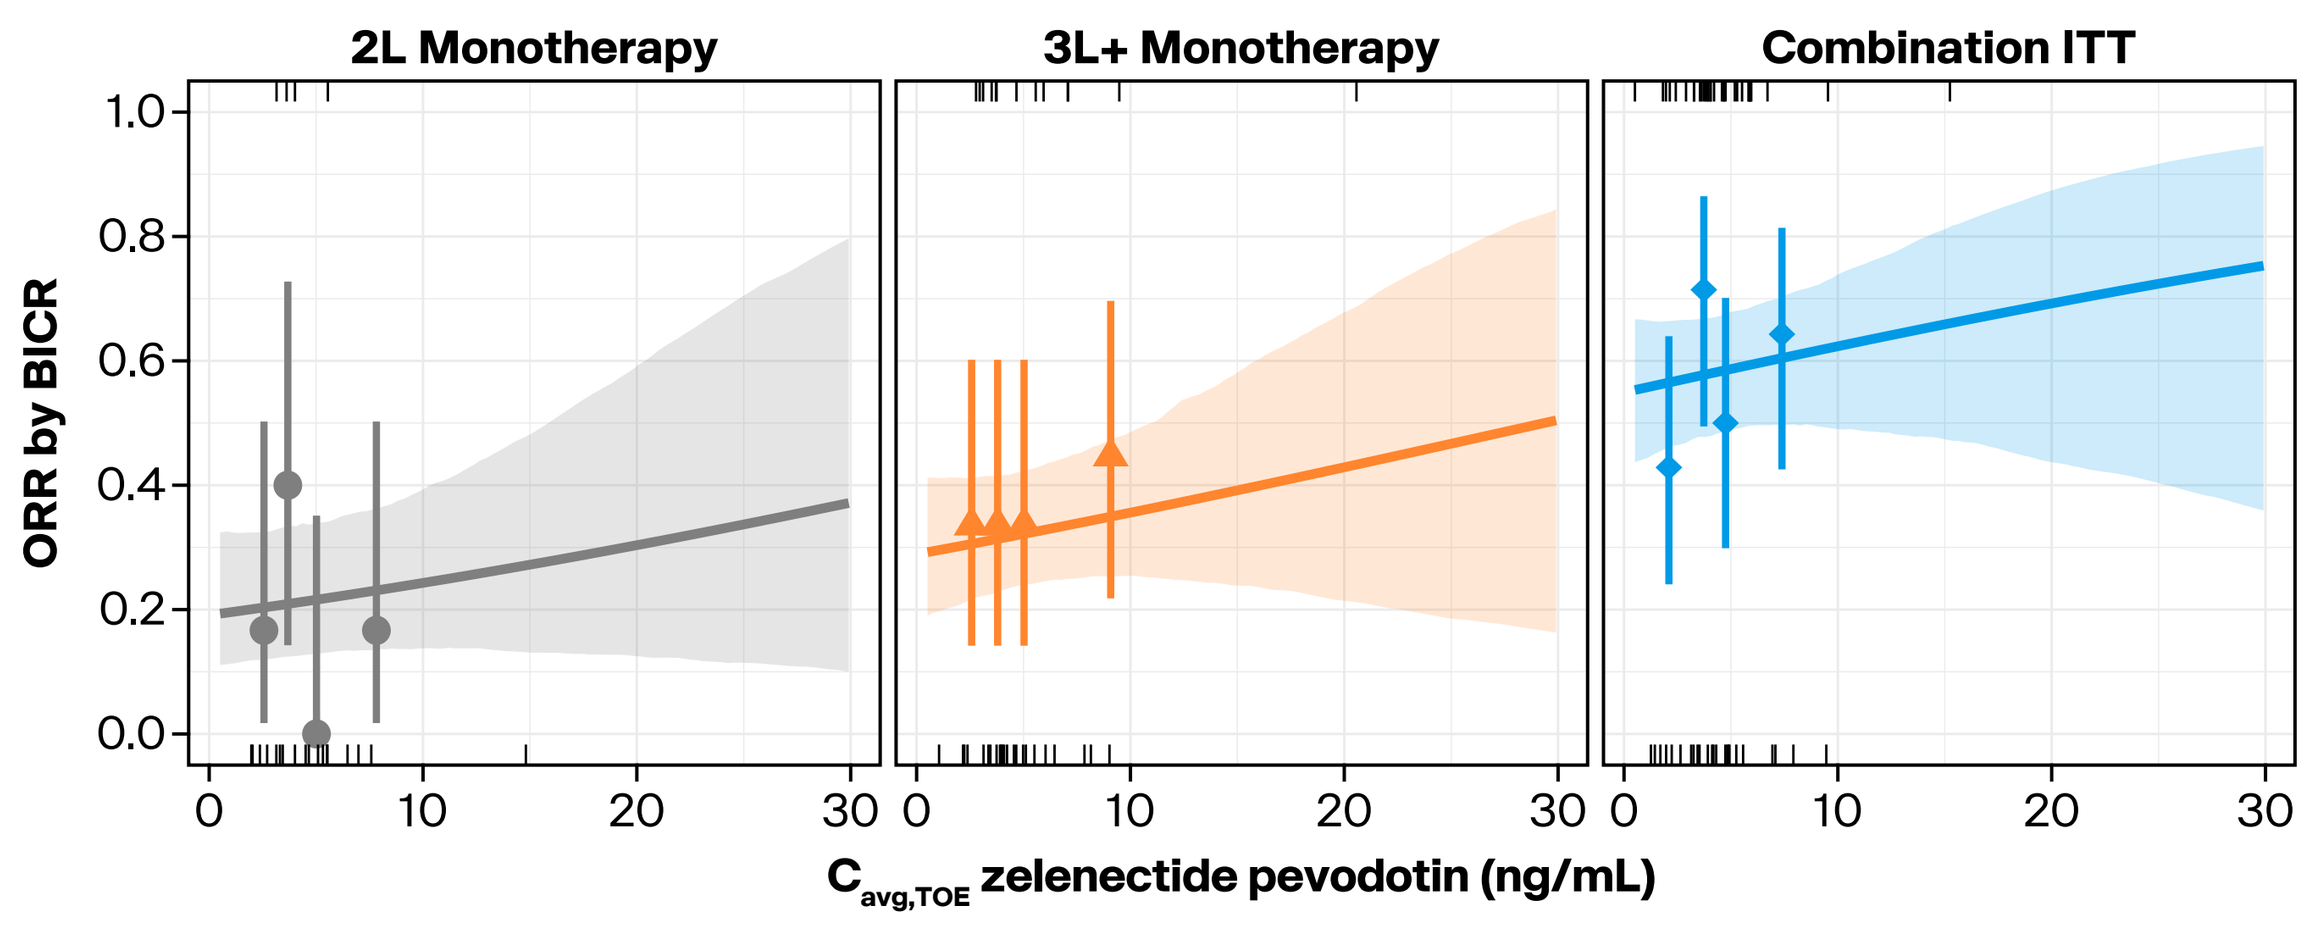
2L Monotherapy
3L+ Monotherapy
Combination ITT
ORR by BICR
1.0
0.8
0.6
0.4
0.2
0.0
0
10
20
30
$C_{avg,TOE}$ zelenectide pevodotin (ng/mL)

Plot shows response probability versus exposure, with the data stratified into exposure quartiles. Symbols present response rates per exposure quartile plotted at the median exposure of the quartile. Vertical bars are 90% CIs of the rates. Colored lines and bands present logistic regression fitting and corresponding 90% CI. Black ticks represent the exposures of responders (coded as 1) and nonresponders (coded as 0).

FIGURE 4. EXPOSURE-SAFETY RELATIONSHIP IDENTIFIED FOR GRADE ≥3 TRAE

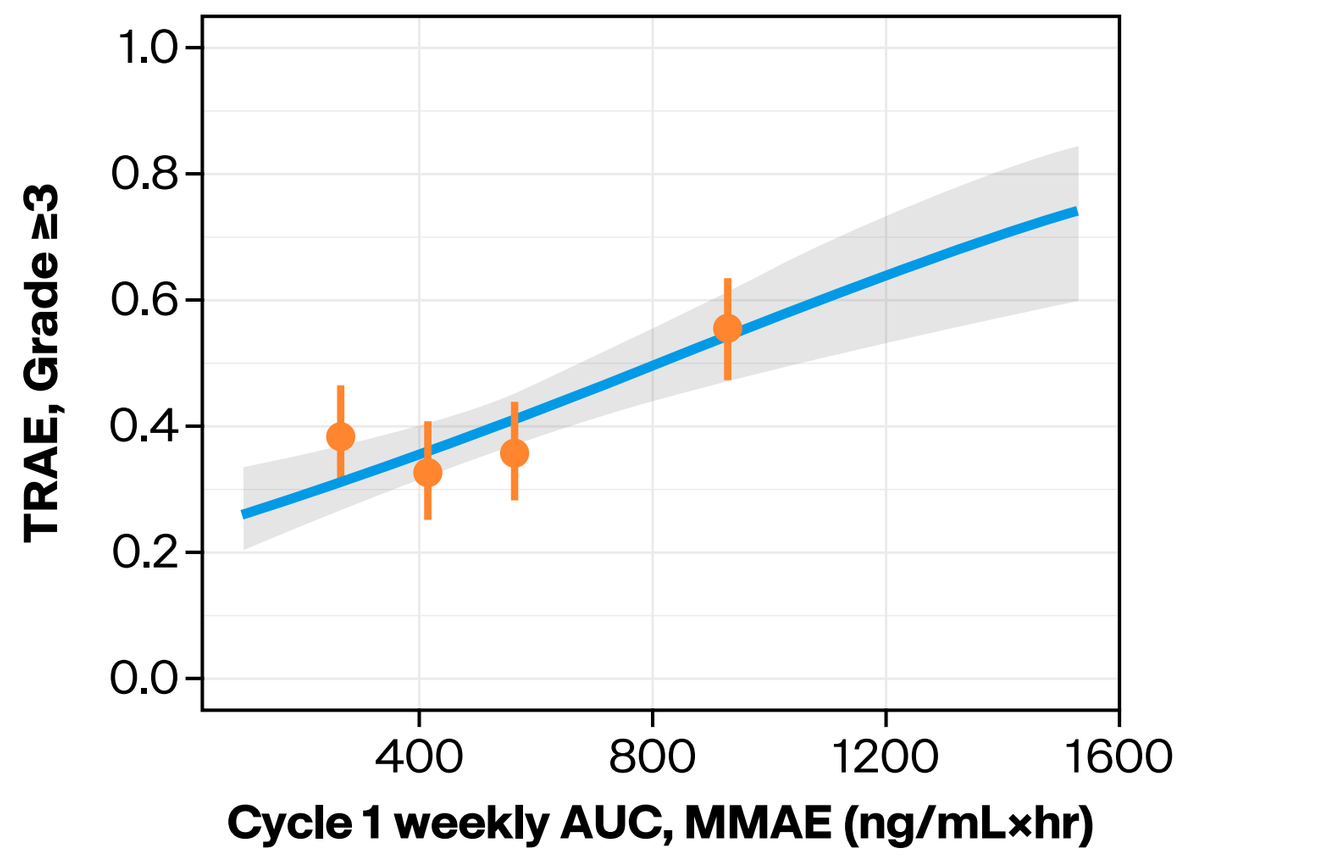
TRAE, Grade ≥3
1.0
0.8
0.6
0.4
0.2
0.0
400
800
1200
1600
Cycle 1 weekly AUC, MMAE (ng/mL×hr)

Plot shows event probability versus exposure, with the data stratified into exposure quartiles. Symbols present observed event rates per exposure quartile plotted at the median exposure of the quartile. Vertical bars are 90% CIs of the rates. Colored line and band represent logistic regression fitting and corresponding 90% CI.

TABLE 4: MODEL-SIMULATED ORR AND TRAE PROBABILITIES BY DOSAGE

Predicted probability of event (%) by zele dosage[a]	Zele Monotherapy			Combination With Pembrolizumab		
	5 mg/m² QW	6 mg/m² D1/8	Difference (6 mg/m²–5 mg/m²)	5 mg/m² QW	6 mg/m² D1/8	Difference (6 mg/m²–5 mg/m²)
Confirmed ORR	**32.5**	30.5	-2.0	**60.9**	58.8	-2.1
Grade ≥3 TRAE	**40.8**	44.5	3.7	**46.6**	50.4	3.8
Grade ≥2 GI disorder	15.3	**12.7**	-2.6	20.8	**17.6**	-3.3
Grade ≥3 neutropenia	**12.3**	17.1	4.7	**13.9**	19.1	5.1
Grade ≥2 peripheral neuropathy	20.6	**8.9**	-11.7	37.8	**18.7**	-19.1
Any-grade skin reactions	**18.1**	18.3	0.2	32.2	32.5	0.3
Dose modification	73.9	**58.4**	-15.5	87.0	**76.5**	-10.5

Bold values indicate the favored dose between the 2 options in each category. [a]Predicted event rates are based on pooled data from Duravelo-1 and Duravelo-2.

UTILITY SCORE ANALYSIS

- The utility score analysis favored 6 mg/m² D1/8 consistently for zele combined with pembrolizumab and remained neutral between the dosages for monotherapy (**Table 5**)

TABLE 5. UTILITY SCORE ANALYSIS OF DURAVELO-2

	Zele 5 mg/m² QW		Zele 6 mg/m² D1/8	
	Cohort 1 (Combination With Pembrolizumab) N=30	Cohort 2 (Monotherapy) N=30	Cohort 1 (Combination With Pembrolizumab) N=30	Cohort 2 (Monotherapy) N=30
Primary approach based on objective response regardless of confirmation and Grade ≥3 TRAE				
Objective responses by BICR=Yes, Grade ≥3 TRAE=No, n (%)	11 (36.7)	4 (13.3)	9 (30.0)	4 (13.3)
Objective responses by BICR=Yes, Grade ≥3 TRAE=Yes, n (%)	7 (23.3)	8 (26.7)	10 (33.3)	7 (23.3)
Objective responses by BICR=No, Grade ≥3 TRAE=No, n (%)	5 (16.7)	12 (40.0)	9 (30.0)	11 (36.7)
Objective responses by BICR=No, Grade ≥3 TRAE=Yes, n (%)	7 (23.3)	6 (20.0)	2 (6.7)	8 (26.7)
Desirability based on utility score assignment,[a] *U*	58.68	**42.66**	**62.64**	39.28
Sensitivity analysis based on confirmed objective response and Grade ≥3 TRAE				
Confirmed objective responses by BICR=Yes, Grade ≥3 TRAE=No, n (%)	10 (33.3)	3 (10.0)	8 (26.7)	4 (13.3)
Confirmed objective responses by BICR=Yes, Grade ≥3 TRAE=Yes, n (%)	6 (20.0)	5 (16.7)	8 (26.7)	5 (16.7)
Confirmed objective responses by BICR=No, Grade ≥3 TRAE=No, n (%)	6 (20.0)	13 (43.3)	10 (33.3)	11 (36.7)
Confirmed objective responses by BICR=No, Grade ≥3 TRAE=Yes, n (%)	8 (26.7)	9 (30.0)	4 (13.3)	10 (33.3)
Desirability based on utility score assignment,[a] *U*	53.30	32.02	**54.72**	**34.00**
Meet safety and efficacy criteria[b]	Yes	Yes	Yes	Yes

Bold values indicate the favored dose between the 2 options in each category. [a]Utility score assignment: 100, 80, 20, 0 (more weights on efficacy over safety 80 vs 20). [b]Criteria are as follows: probability (true grade ≥3 TRAE incidence <75% | observed IA data) >80%; Cohort 1: probability (true ORR by BICR >25% | observed IA data) >80%. Cohort 2: probability (true ORR by BICR >18% | observed IA data) >80%.

CONCLUSIONS

- The 6 mg/m² D1/8 dosage was identified as the optimized zele dosage for monotherapy and in combination with pembrolizumab based on the overall benefit-risk informed by
 - PMx analyses of data from Duravelo-1 and Duravelo-2
 - Utility-based analysis from Duravelo-2
 - Other clinical factors
- Zele at 6 mg/m² D1/8 offers similar efficacy and improved safety versus 5 mg/m² QW and is more convenient for patients, which may enhance treatment compliance

Please visit more presentations about zele at ASCO 2026

- **Abstract #4516:** Loriot Y, et al. Interim analysis results from Duravelo-2: zelenectide pevedotin + pembrolizumab in patients with previously untreated locally advanced/metastatic urothelial carcinoma. **Rapid Oral presented Monday, June 1, 2026, starting at 8:00 am CDT.**
- **Abstract #4563:** Reig Torras O, et al. Zelenectide pevedotin (BT8009) monotherapy in previously treated metastatic urothelial carcinoma: update on Duravelo-1.
- **Abstract #4564:** Duran I, et al. Zelenectide pevedotin (BT8009) plus pembrolizumab in first-line cisplatin-ineligible locally advanced/metastatic urothelial carcinoma: update on Duravelo-1 B7.
- **Abstract #4566:** Petrylak D, et al. Interim analysis results from Duravelo-2: zelenectide pevedotin (BT8009) in patients with previously treated locally advanced/metastatic urothelial carcinoma.

REFERENCES

1. Rigby M, et al. *Mol Cancer Ther*. 2022;21(12):1747–1756.
2. Reig Torras O, et al. ESMO 2024. Abstract 652P.
3. Giannatempo P, et al. ASCO 2025. Abstract 4567.
4. Loriot Y, et al. ASCO 2024. Abstract TPS4619.
5. Loriot Y, et al. ASCO 2026. Abstract 4516.
6. Kaufman, et al. ESMO 2025. Abstract 628TiP.

ABBREVIATIONS

2L, second-line; 3L+, third-line or later; AE, adverse event; AUC, area under the curve; BDC, Bicycle Drug Conjugate®; BICR, blinded independent central reviewer; BSA, body surface area; C_{avg}, average concentration; $C_{avg,TOE}$, average concentration to time of event; C_{max}, maximum concentration; D, day; ECOG PS, Eastern Cooperative Oncology Group performance status; GI, gastrointestinal; ITT, intent-to-treat; la/mUC, locally advanced/metastatic urothelial carcinoma; MMAE, monomethyl auristatin E; NE, not evaluable; ORR, overall response rate; PK, pharmacokinetic; PMx, pharmacometric; popPK, population PK; TRAE, treatment-related adverse event; QW, once weekly; Q2W, every 2 weeks; zele, zelenectide pevedotin.

ACKNOWLEDGMENTS

Bicycle Therapeutics would like to thank the patients and their families for participating in this study. Without their contributions, this study would not have been possible. Medical writing support was provided by Danielle Fanslow, PhD, CMPP, of Second City Science, and funded by BicycleTx Ltd.

Funding: This study was sponsored by BicycleTx Ltd.

Presenting author: Hongmei Xu, PhD

Presenting author COI disclosures: employee of BicycleTx Ltd; stock and other ownership Interests in AstraZeneca, Bicycle Therapeutics, and Karyopharm Therapeutics; patents, royalties, other intellectual property in Bicycle Therapeutics.