Interim analysis results from Duravelo-2: zelenectide pevedotin in patients with previously treated locally advanced/metastatic urothelial carcinoma

Abstract #

4566

Daniel P. Petrylak, MD[1]; Mauricio Burotto, MD[2]; Felipe Reyes-Cosmelli, MD[3]; Hernan Cutuli, MD[4]; Robert A. Huddart, MBBS, MRCP, FRCR, PhD[5]; Tamta Makharadze, MD[6]; José Pablo Maroto, MD, PhD[7]; Danish Mazhar, MBBS, PhD[8]; Imanol Martinez Salas, MBBS[9]; Peter Sankey, MBBS, MRCP, FRCR[10]; Ho Kyung Seo, MD, PhD[11]; Cong Xu, PhD[12]; Leslie R. DeMars, MD[13]; Michael Method, MD, MBA, MPH[13]; Yohann Loriot, MD, PhD[14]

[1]Yale School of Medicine, New Haven, CT; [2]Bradford Hill Clinical Research Center, Santiago, Chile; [3]Fundación Arturo López Pérez, Santiago, Chile; [4]Hospital Sirio Libanés, Buenos Aires, Argentina; [5]The Royal Marsden NHS Foundation Trust, London, United Kingdom and The Institute of Cancer Research, London, United Kingdom; [6]LTD High Technology Hospital Medical Center, Batumi, Georgia; [7]Hospital de la Santa Creu i Sant Pau, Barcelona, Spain; [8]Addenbrooke's Hospital, Cambridge University Hospitals NHS Foundation Trust, Cambridge, United Kingdom; [9]University Hospital Fundación Jiménez Diaz, Madrid, Spain; [10]University Hospitals Plymouth NHS Trust, Derriford Hospital, Plymouth, United Kingdom; [11]Center for Urologic Cancer, National Cancer Center, Goyang, Republic of Korea; [12]Bicycle Therapeutics (at the time of analysis), Lexington, MA; [13]Bicycle Therapeutics, Lexington, MA; [14]Gustave Roussy Institute, University Paris-Saclay, Villejuif, France.

INTRODUCTION

- Current antibody-drug conjugate (ADC) regimens with cytotoxic monomethyl auristatin E (MMAE) payload offer targeted and effective therapy for locally advanced or metastatic urothelial carcinoma (la/mUC) yet commonly lead to adverse events (AEs), such as peripheral neuropathy, dermatologic disorders, and eye toxicity[1-3]
 - These AEs lead to reduced tolerability and increased discontinuations, highlighting the unmet need for more tolerable treatments for la/mUC
- Bicycle® molecules are an innovative therapeutic class in development that offer manufacturing and pharmacokinetic (PK) properties of a small molecule with the high binding specificity of a biologic,[4-6] suiting them for the targeted delivery of a range of payloads, including cytotoxins, to solid tumors
- Zelenectide pevedotin (zele; formerly BT8009) is a first-in-class Bicycle® Drug Conjugate, comprising a highly selective Nectin-4–targeted Bicycle® peptide conjugated to MMAE via a cleavable linker (**Figure 1**)[4,7]
 - Nectin-4 is a protein commonly overexpressed in la/mUC, making it a viable target for delivery of MMAE[8-11]
- The Phase 1/2 Duravelo-1 (NCT04561362) dose escalation and expansion study reported a well-tolerated safety profile of zele and preliminary efficacy in 45 patients with previously treated la/mUC receiving 5 mg/m^2 zele monotherapy[12]

FIGURE 1. ZELENECTIDE PEVEDOTIN (ZELE) STRUCTURE[13]

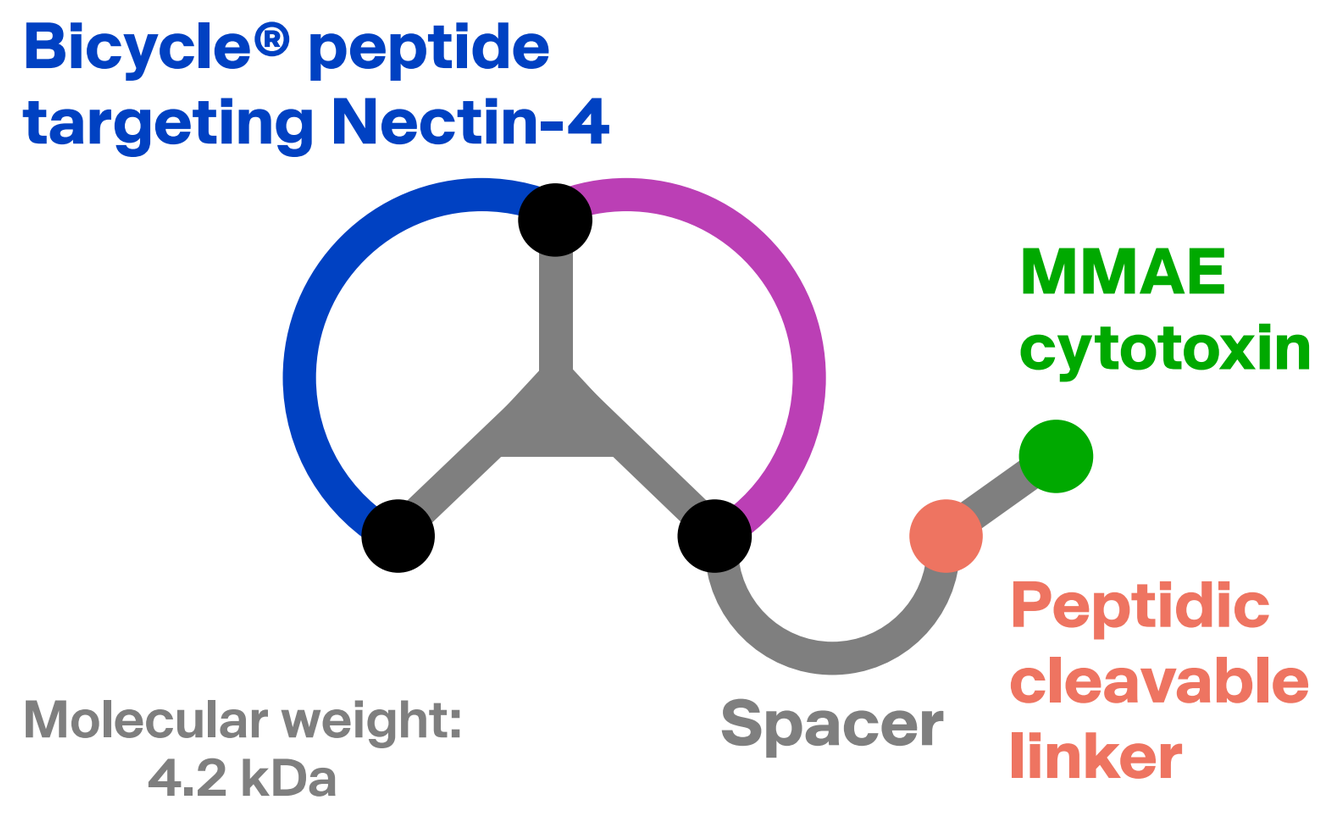

Bicycle® peptide targeting Nectin-4
MMAE cytotoxin
Peptidic cleavable linker
Spacer
Molecular weight: 4.2 kDa

OBJECTIVE

- To report an interim analysis (IA) for zele monotherapy dose optimization from the ongoing randomized Phase 2 Duravelo-2 study (NCT06225596) in patients who have received ≥1 prior systemic therapy for la/mUC

METHODS

- Adults with la/mUC were enrolled into 1 of 2 cohorts: previously untreated patients eligible for platinum-based chemotherapy (Cohort 1) or patients with ≥1 prior systemic therapy (Cohort 2); **here, we report the results of Cohort 2** (**Figure 2**)
 - Participants were stratified by prior treatment with programmed cell death protein 1 and/or programmed death-ligand 1 (PD-1, PD-L1, respectively) inhibitors and presence of liver metastases
- Patients were randomized 1:1 to initially determine the optimized dose of zele monotherapy (**Figure 2**)
 - The 5 mg/m^2 regimen was administered on Day (D) 1, 8, and 15 of each 21-D cycle
 - The 6 mg/m^2 regimen was administered on D 1 and 8 of each 21-D cycle
- Dose optimization was determined by overall data to assess the benefit–risk, including efficacy, safety, utility score, and PK modeling of exposure safety and exposure response
 - Key efficacy endpoints were objective response rate (ORR), disease control rate (DCR), and duration of response (DoR) as evaluated per RECIST v1.1 by blinded independent central review (BICR)
 - Safety endpoints included the frequency of treatment-emergent and treatment-related AEs (TEAEs and TRAEs, respectively) by grade and serious AEs (SAEs)
 - The efficacy-evaluable population includes patients who received any dose of study drug and had measureable disease at baseline

FIGURE 2. DURAVELO-2 STUDY DESIGN FOR ZELE DOSE OPTIMIZATION

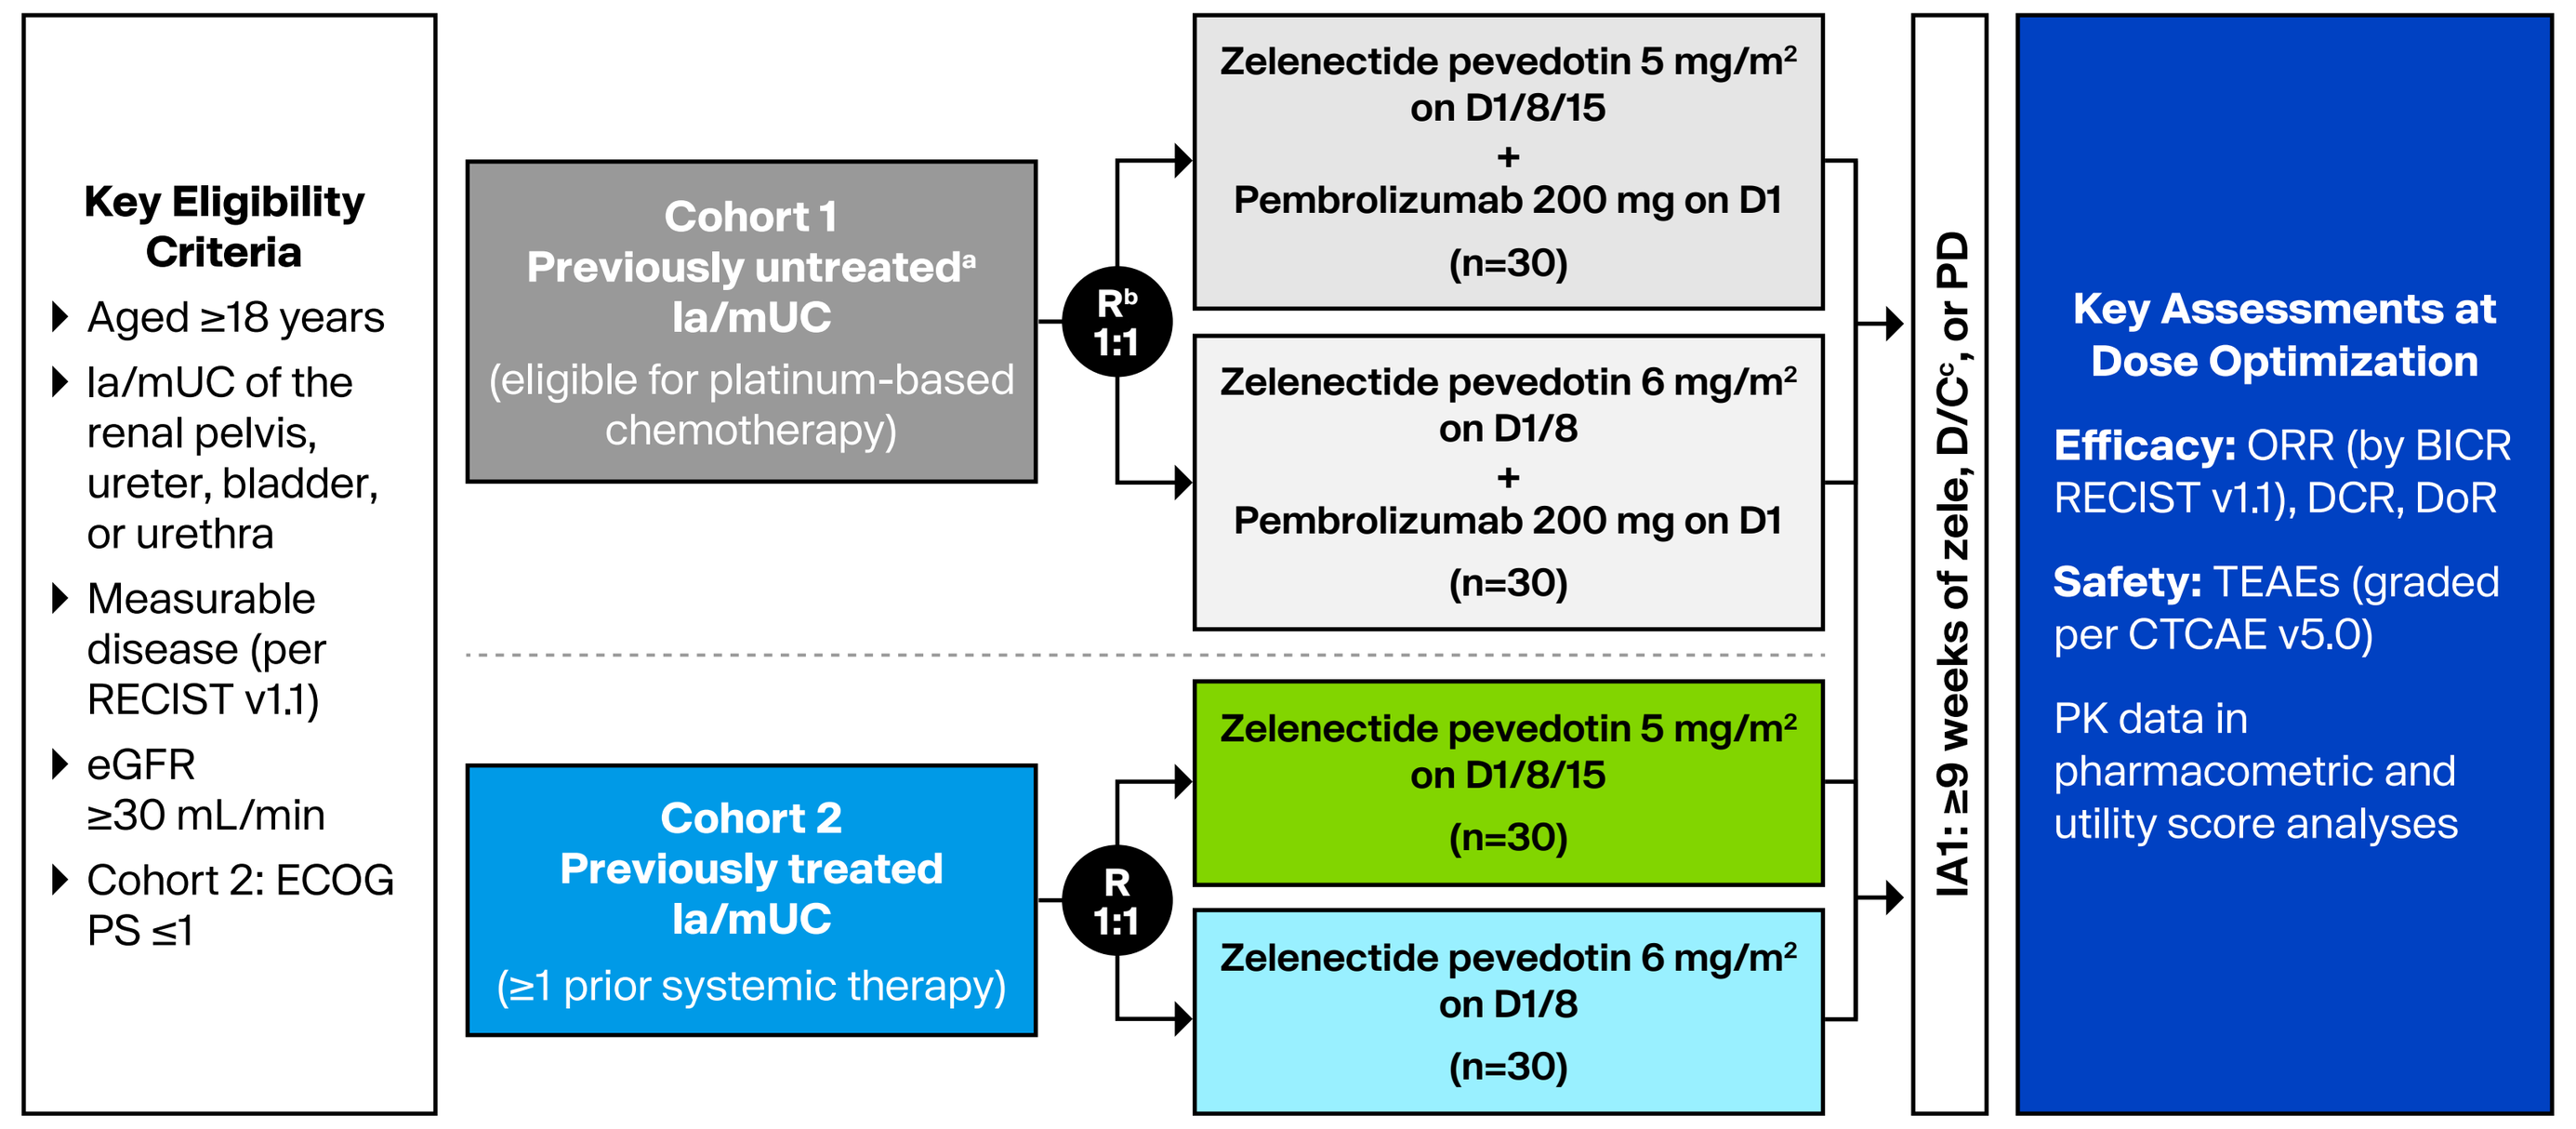

Key Eligibility Criteria
Aged ≥18 years
la/mUC of the renal pelvis, ureter, bladder, or urethra
Measurable disease (per RECIST v1.1)
eGFR ≥30 mL/min
Cohort 2: ECOG PS ≤1
Cohort 1 Previously untreated[a] la/mUC
(eligible for platinum-based chemotherapy)
R[b] 1:1
Zelenectide pevedotin 5 mg/m² on D1/8/15 + Pembrolizumab 200 mg on D1 (n=30)
Zelenectide pevedotin 6 mg/m² on D1/8 + Pembrolizumab 200 mg on D1 (n=30)
Cohort 2 Previously treated la/mUC
(≥1 prior systemic therapy)
R 1:1
Zelenectide pevedotin 5 mg/m² on D1/8/15 (n=30)
Zelenectide pevedotin 6 mg/m² on D1/8 (n=30)
IA1: ≥9 weeks of zele, D/C[c], or PD
Key Assessments at Dose Optimization
Efficacy: ORR (by BICR RECIST v1.1), DCR, DoR
Safety: TEAEs (graded per CTCAE v5.0)
PK data in pharmacometric and utility score analyses

Treatments were based on 21-day cycles. [a]Patients with prior neoadjuvant/adjuvant chemotherapy, MMAE-based therapy, immune checkpoint inhibitor therapy with recurrence >12 months from completion of therapy were allowed. [b]Patients in Cohort 1 were also randomized to a third treatment arm and received 4-6 cycles of gemcitabine + cisplatin/carboplatin ± avelumab maintenance but were not considered for zele dose optimization studies. [c]Discontinuation criteria include planned completion of therapy, progressive disease, and intolerable toxicity.

RESULTS

PATIENT DEMOGRAPHICS AND CHARACTERISTICS

- The IA was performed when the IA1 population had been treated for 27 weeks, progressed, or discontinued the study
- As of 14 June 2025, a total of 60 patients with a median age of 66 (range, 45–83) and previously treated la/mUC were included in Cohort 2 (**Table 1**)
- Median relative dose intensity was 92.9% for the 5 mg/m^2 on D1/8/15 dose group and 85.6% for the 6 mg/m^2 on D1/8 dose group

TABLE 1. BASELINE PATIENT DEMOGRAPHICS AND CLINICAL CHARACTERISTICS

Baseline Patient Characteristic	5 mg/m^2 D1/8/15 (N=30)	6 mg/m^2 D1/8 (N=30)
Median age (range), years	69.5 (46–83)	65 (45–76)
Male sex, n (%)	21 (70.0)	19 (63.3)
Race, n (%)		
Asian	10 (33.3)	7 (23.3)
White	18 (60.0)	21 (70.0)
Other/not reported/unknown	2 (6.7)	2 (6.7)
Primary disease site of origin, n (%)		
Upper urinary	8 (26.7)	7 (23.3)
Lower urinary	22 (73.3)	23 (76.7)
ECOG PS, n (%)		
0	15 (50.0)	15 (50.0)
1	15 (50.0)	15 (50.0)
Median prior lines of therapy (range)	2 (1–3)	2 (1–6)
Prior PD-1/PD-L1 inhibitor, n (%)	22 (73.3)	22 (73.3)
Presence of any metastasis	19 (63.3)	23 (76.7)
Liver metastases	4 (13.3)	6 (20.0)

TABLE 2. CONFIRMED BEST OVERALL RESPONSE IN EFFICACY-EVALUABLE PATIENTS BY ZELE DOSE

Parameter[a]	5 mg/m^2 D1/8/15 (n=29)	6 mg/m^2 D1/8 (n=27)
ORR[b], n (%) [95% CI]	**8 (27.6)** [12.7–47.2]	**8 (29.6)** [13.8–50.2]
Confirmed BOR, n (%)		
Complete response	2 (6.9)	3 (11.1)
Partial response	6 (20.7)	5 (18.5)
Stable disease[c]	14 (48.3)	11 (40.7)
Progressive disease	6 (20.7)	8 (29.6)
Not evaluable	1 (3.4)	0
DCR, n (%)	22 (75.9)	19 (70.4)

[a]BOR with confirmation for a participant is defined as the best confirmed time point response from randomization until documented disease progression, using RECIST v1.1 in the order of CR, PR, SD, PD, and NE. [b]ORR with confirmation (%) is defined as the proportion of participants with BOR with confirmation of CR or PR; 95% CI was calculated by using exact Clopper-Pearson method. [c]SD must be documented as present at least once on or after 6 weeks post first dose for BOR to be SD.

EFFICACY

- Among the 27 patients treated with zele at 6 mg/m^2 D1/8 and with efficacy-evaluable disease, the confirmed ORR was 30% (**Table 2**)
 - At 27 weeks, 8 patients receiving 6 mg/m^2 remained on treatment, including 2 with stable disease (**Figure 3**)
 - The confirmed ORR among patients treated with 5 mg/m^2 of zele on D1/8/15 was 28%
 - Six additional patients had unconfirmed responses: 4 in the 5 mg/m^2 dose group (1 CR, 3 PR; ORR, 41.4%) and 2 in the 6 mg/m^2 dose group (2 PR; ORR, 37.0%)
- Median time on treatment was 4.8 months for patients receiving 6 mg/m^2 on D1/8
- Median duration of follow-up for the 5 mg/m^2 and 6 mg/m^2 doses were 6.7 months (range, 0.5–10.2) and 6.2 months (range, 0.1–9.9), respectively

FIGURE 3. TUMOR SIZE CHANGE (A) AND PATIENT DoR (B) FROM BASELINE IN EFFICACY-EVALUABLE PATIENTS TREATED WITH 6 mg/m² ZELE ON D1/8 BY BICR (n=26[a])

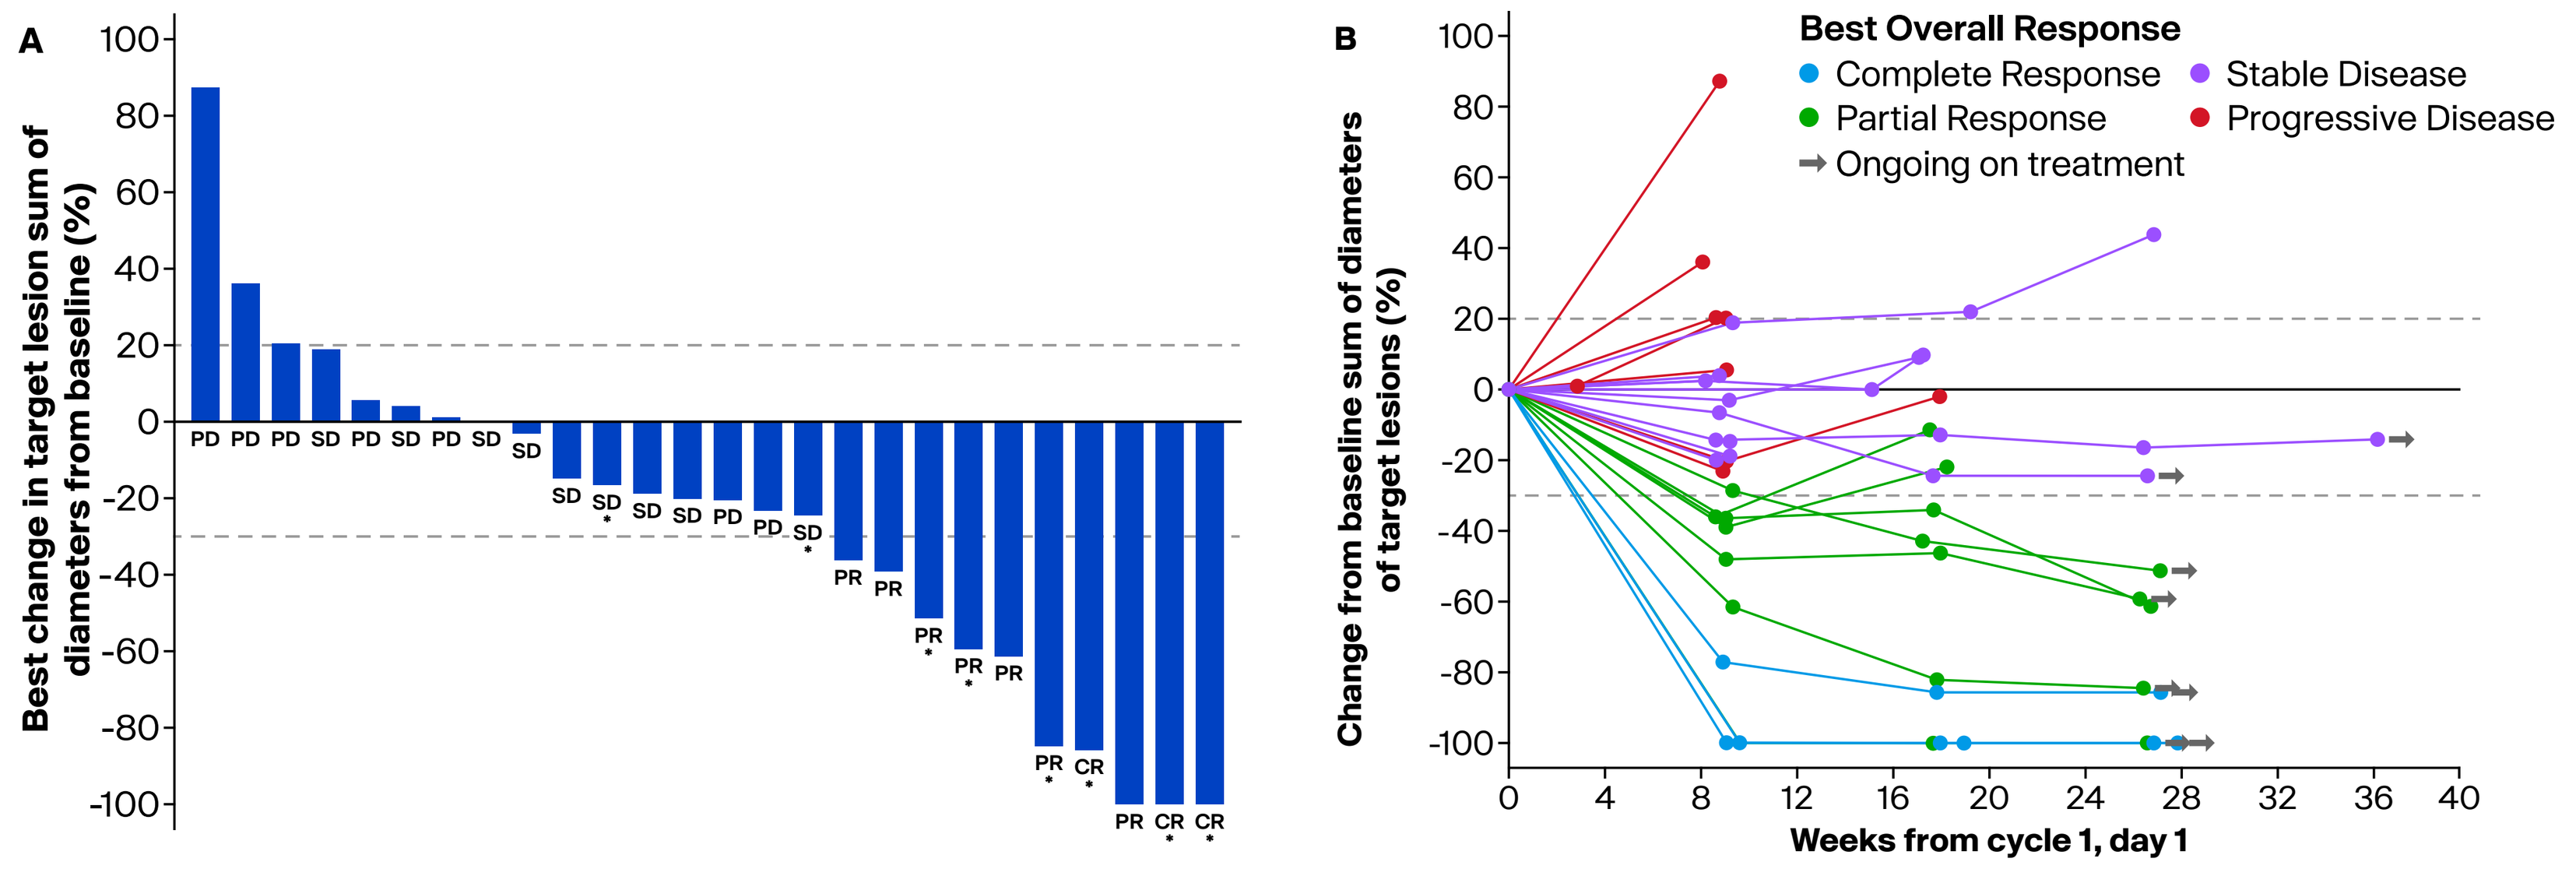

A
Best change in target lesion sum of diameters from baseline (%)
B
Change from baseline sum of diameters of target lesions (%)
Weeks from cycle 1, day 1
Best Overall Response
Complete Response
Partial Response
Stable Disease
Progressive Disease
Ongoing on treatment

*Asterisks and grey arrows indicate patients continuing zele treatment, except patients with PD as their BOR.
[a]Four patients were not included in **Figure 3** based on no measurable disease at baseline (n=2), not receiving zele (n=1), and a lack of adequate post-baseline target lesions (n=1).

SAFETY

TABLE 3. SAFETY SUMMARY BY ZELE DOSE

AEs, n (%)	5 mg/m^2 D1/8/15 (N=30)	6 mg/m^2 D1/8 (n=29[a])
TEAEs of any Grade	30 (100)	29 (100)
Grade ≥3	18 (60.0)	21 (72.4)
TRAEs of any Grade	28 (93.3)	27 (93.1)
Grade ≥3	14 (46.6)	15 (51.7)
SAEs	6 (20.0)	11 (37.9)
Zele-related SAEs	2 (6.7)	4 (13.8)
TRAEs leading to dose modification		
Dose reduction	10 (33.3)	7 (24.1)
Dose interruption	14 (46.7)	17 (58.6)
Dose discontinuation	2 (6.7)	0

[a]The safety-evaluable population treated with 6 mg/m^2 zele excluded n=1 patient who did not receive zele.

- Grade ≥3 TRAEs occurred in 47% and 52% of patients treated with 5 mg/m^2 and 6 mg/m^2, respectively (**Table 3**)
 - No Grade 5 TRAEs were reported in patients treated with 5 mg/m^2
 - No Grade 4 or 5 TRAEs were reported in patients treated with 6 mg/m^2
- No treatment discontinuations occurred in patients who received the 6 mg/m^2 dose

FIGURE 4. COMMON TRAEs ACROSS BOTH ZELE DOSES[a]

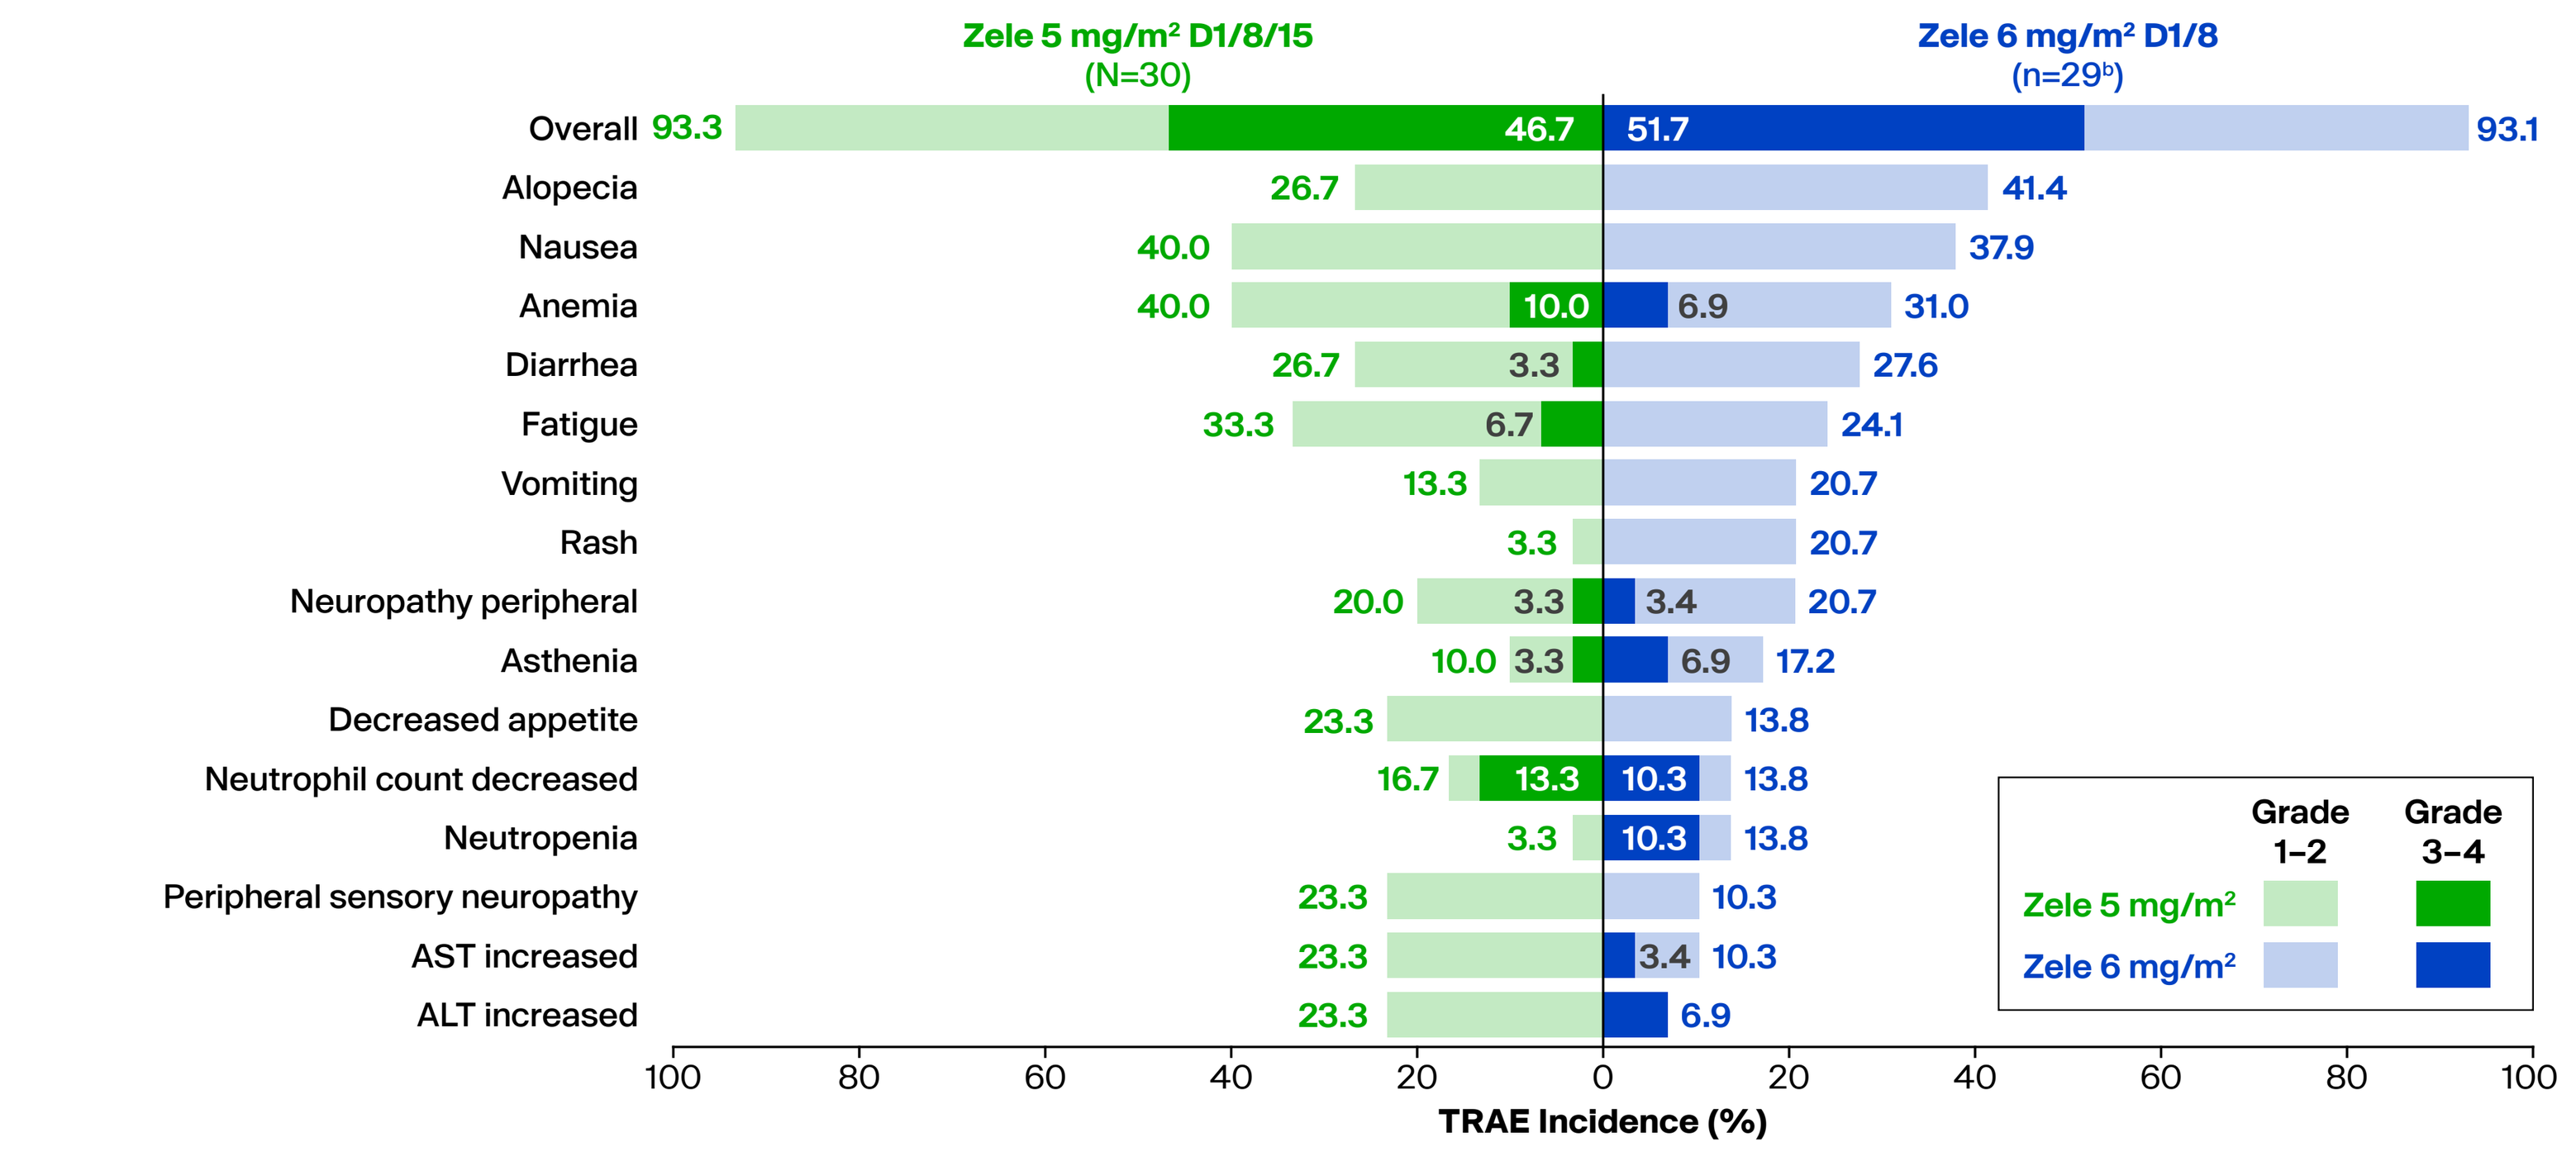

Zele 5 mg/m² D1/8/15 (N=30)
Zele 6 mg/m² D1/8 (n=29[b])
Overall 93.3 46.7 51.7 93.1
Alopecia 26.7 41.4
Nausea 40.0 37.9
Anemia 40.0 10.0 6.9 31.0
Diarrhea 26.7 3.3 27.6
Fatigue 33.3 6.7 24.1
Vomiting 13.3 20.7
Rash 3.3 20.7
Neuropathy peripheral 20.0 3.3 3.4 20.7
Asthenia 10.0 3.3 6.9 17.2
Decreased appetite 23.3 13.8
Neutrophil count decreased 16.7 13.3 10.3 13.8
Neutropenia 3.3 10.3 13.8
Peripheral sensory neuropathy 23.3 10.3
AST increased 23.3 3.4 10.3
ALT increased 23.3 6.9
TRAE Incidence (%)
Grade 1–2
Grade 3–4
Zele 5 mg/m²
Zele 6 mg/m²

[a]Includes AEs of any grade related to zele that occurred in ≥20% of patients or of Grade ≥3 that occurred in ≥5% of patients in either arm. Patients with multiple AEs are counted only once by the worse NCI-CTCAE category within a perferred term. [b]The safety-evaluable population treated with 6 mg/m^2 zele excluded n=1 patient who did not receive zele.

TABLE 4: ZELE-RELATED AECIs IN PATIENTS TREATED WITH 6 mg/m² ZELE ON D1/8

	6 mg/m^2 D1/8 (n=29[b])					
AECIs,[a] n (%)	**Any Grade**	**Grade 1**	**Grade 2**	**Grade 3**	**Grade 4**	**Grade 5**
Peripheral neuropathy[c]	11 (37.9)	9 (31.0)	1 (3.4)	1 (3.4)	0	0
Sensory events	11 (37.9)	9 (31.0)	1 (3.4)	1 (3.4)	0	0
Motor events	0	0	0	0	0	0
Eye disorders[d]	3 (10.3)	3 (10.3)	0	0	0	0
Skin reactions[e]	8 (27.6)	8 (27.6)	0	0	0	0
Hyperglycemia[f]	1 (3.4)	0	0	1 (3.4)	0	0

[a]Patients can have multiple preferred terms within a category. [b]The safety-evaluable population treated with 6 mg/m^2 zele excluded n=1 patient who did not receive zele. [c]MedDRA SMQ [Broad] for peripheral neuropathy. [d]SOC of eye disorders. [e]MedDRA SMQ [broad] for SCAR and high levels terms of “bullous conditions,” “dermatitis and eczema,” “rashes, eruptions and exanthems NEC,” “erythemas,” and “dermatitis ascribed to specific agent.” [f]Preferred term.

- Several common TRAEs of any grade with incidence ≥20% in patients were numerically less in patients treated with 6 mg/m^2 on D1/8 than those treated with 5 mg/m^2 on D1/8/15, including anemia, fatigue, decreased appetite, peripheral sensory neuropathy, and liver-related toxicity (**Figure 4**)
 - Among TRAEs with ≥20% incidence in patients treated with either dose, none were Grade 4 or 5
 - Among TRAEs with ≥5% incidence in patients treated with 6 mg/m^2 zele, none were Grade 4 or 5.
- TRAEs of clinical interest occurring in patients from either dose group were predominantly Grade 1, with no Grade 4 or 5 events reported (**Table 4**)
 - All skin reactions were limited to Grade 1 and no patients reported severe skin reactions
 - Most of the peripheral neuropathy events in 6 mg/m^2 dose group were Grade 1 (82%; 9/11)
 - Grade 3 peripheral neuropathy was limited to 1 patient in each dose group
 - All peripheral neuropathy events were sensory, with no motor events reported in either dose group
 - Eye disorders were predominantly Grade 1
- TRAEs of clinical interest occurring in patients treated with 5 mg/m^2 zele were similar to those treated with 6 mg/m^2 with no incidence of Grade 4 or 5 events
 - Incidence of any-grade and Grade 2 peripheral neuropathy was greater in patients treated with 5 mg/m^2 zele than those treated with 6 mg/m^2

CONCLUSIONS

- The 6 mg/m^2 on D1/8 of a 21-D cycle regimen was identified as the optimized zele monotherapy dose based on the overall benefit–risk profile
- Zele at 6 mg/m^2 demonstrated an encouraging response rate, with 8 patients remaining on treatment at the time of IA
- Based on the safety profile, the 6 mg/m^2 dose of zele meets the need for safer and more tolerable treatments with enhanced potential for combinability and convenience, leading to fewer treatment discontinuations
 - Skin toxicity with zele is low grade and manageable; as of 18 June 2025, no events of Stevens-Johnson syndrome (SJS) or toxic epidermal necrolysis (TEN) have been reported in 595 patients treated with zele, demonstrating a differentiated safety profile compared with currently available MMAE-based ADC regimens[14]

Please visit more presentations about la/mUC at ASCO 2026

- **Abstract #4516:** Loriot Y, et al. Interim analysis results from Duravelo-2: zelenectide pevedotin + pembrolizumab in patients with previously untreated locally advanced/metastatic urothelial carcinoma. **Rapid Oral presented Monday, June 1, 2026, starting at 8:00 am CDT.**
- **Abstract #4563:** Torras OR, et al. Zelenectide pevedotin (BT8009) monotherapy in previously treated metastatic urothelial carcinoma: Update on Duravelo-1.
- **Abstract #4564:** Duran I, et al. Zelenectide pevedotin (BT8009) plus pembrolizumab in first-line cisplatin-ineligible locally advanced/metastatic urothelial carcinoma: Update on Duravelo-1 B7.
- **Abstract #4567:** Lu Y, et al. Identify optimized dosage for zelenectide pevedotin in locally advanced/metastatic urothelial carcinoma using quantitative analyses.

REFERENCES

1. D'Angelo A, et al. *World J Clin Oncol.* 2025;16(9):106646.
2. Chen H, et al. *Front Immunol.* 2024;15:1495137.
3. Powles T, et al. *N Engl J Med.* 2021;384:1125–1135.
4. Eder M, et al. *Cancer Res.* 2019;79(4):841–852.
5. Mudd GE, et al. *J Med Chem.* 2020;63(8):4107–4116.
6. Walsh SJ, et al. *Cancer Res.* 2024;84(6_Suppl):5807.
7. Rigby M, et al. *Mol Cancer Ther.* 2022;21(12):1747–1756.
8. Duan X, et al. *Clin Cancer Res.* 2023;29(17):3395–3407.
9. Mudd GE, et al. *J Med Chem.* 2022;65(21):14337–14347.
10. Zhou W, et al. *Mol Cancer Ther.* 2023;22(8):913–925.
11. Challita-Eid PM, et al. *Cancer Res.* 2016;76(10):3003–3013.
12. Reig Torras O, et al. *Ann Oncol.* 2024;35:S515–S516.
13. Kaufman P, et al. ESMO 2025. Abstract 628TiP.
14. Padcev (enfortumab vedotin-ejfv) [package insert]. Northbrook, IL: Astellas Pharma US, Inc; 2025.

ABBREVIATIONS

ADC, antibody-drug conjugate; AE, adverse event; AECI, adverse event of clinical interest; BICR, blinded independent central review; BOR, best overall response; CI, confidence interval; D/C, discontinuation; DCR, disease control rate; DoR, duration of response; eGFR, estimated glomerular filtration rate; IA, interim analysis; kDa, kilodalton; la/mUC, locally advanced or metastatic urothelial carcinoma; MedDRA, Medical Dictionary for Regulatory Activities; MMAE, monomethyl auristatin E; NE, not evaluable; ORR, objective response rate; PD-1, programmed cell death protein 1; PD-L1, programmed death-ligand 1; PK, pharmacokinetic; R, randomized; SAE, serious adverse event; SJS, Stevens-Johnson syndrome; SMQ, Standardized Medical Dictionary for Regulatory Activities (MedDRA) Queries; TEAE, treatment-emergent adverse event; TEN, toxic epidermal necrolysis; TRAE, treatment-related adverse event; zele, zelenectide pevedotin.

ACKNOWLEDGMENTS

Bicycle Therapeutics would like to thank the patients and their families for participating in this study. Without their contributions, this study would not have been possible.

Medical writing support was provided by Aaron Johnson, PhD, of Second City Science, funded by BicycleTx Ltd.

Funding: This study was sponsored by BicycleTx Ltd.

Presenting author: Daniel Petrylak, MD

Presenting author COI disclosures: Consulting or Advisory Role: Advanced Accelerator Applications, Amgen, Astellas Pharma, AstraZeneca, Bayer, Bicycle Therapeutics, Boehringer Ingelheim, Bristol-Myers Squibb, Clovis Oncology, Exelixis, Gilead Sciences, Incyte, Ipsen, Janssen, Lilly, Mirati Therapeutics, Monopteros Therapeutics, Pfizer, Pharmacyclics, Regeneron, Roche, Seagen, UroGen Pharma. Research Funding: Advanced Accelerator Applications, Agensys, BioXcel Therapeutics, Bristol-Myers Squibb, Clovis Oncology, Eisai, Mirati Therapeutics. Expert Testimony: Celgene, Sanofi.