Zelenectide pevedotin (BT8009) monotherapy in previously treated metastatic urothelial carcinoma: Update on Duravelo-1

Abstract #

4563

Oscar Reig Torras,[1] Laurence Crouzet,[2] Andrea Necchi,[3] Capucine Baldini,[4] M. Julia Lostes-Bardaji,[5] Louise Carter,[6] Bernard Gaston Doger de Spéville,[7] Antoine Italiano,[8] Loic Verlingue,[9] Valentina Boni,[10] Ignacio Duran,[11] Benjamin Garmezy,[12] Matthew D. Galsky,[13] Gerald Steven Falchook,[14] Leslie DeMars,[15] Gladys Brown,[15] Cong Xu,[16] Elisa Fontana[17]

[1]Hospital Clínic de Barcelona, Barcelona, Spain; [2]Unicancer, Centre Eugène Marquis, Rennes, France; [3]VitaSalute San Raffaele University and IRCCS Ospedale San Raffaele, Milan, Italy; [4]Gustave Roussy Cancer Campus, Villejuif, France; [5]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology, Barcelona, Spain; [6]The University of Manchester and The Christie NHS Foundation Trust, Manchester, United Kingdom; [7]StartMadrid Phase 1 Clinical Unit, START MadridFJDHospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [8]Early Phase Trials Unit, Institut Bergonié, and University of Bordeaux, Bordeaux, France; [9]Phase 1 Clinical Unit, Centre Léon Bérard, Lyon, France; [10]NEXT Madrid, Universitary Hospital Quirónsalud Madrid, Madrid, Spain; [11]Hospital Universitario Marqués de Valdecilla, IDIVAL, Santander, Spain; [12]Sarah Cannon Research Institute, Nashville, TN; [13]Tisch Cancer Institute, Icahn School of Medicine at Mount Sinai, New York, NY; [14]Sarah Cannon Research Institute at HealthONE, Denver, CO; [15]Clinical Development, Bicycle Therapeutics, Cambridge, MA; [16]Biostatistics, Bicycle Therapeutics, Cambridge, MA (affiliation at the time of analysis); [17]Drug Development, Sarah Cannon Research Institute UK, London, United Kingdom

INTRODUCTION

- Additional therapeutic options are needed in locally advanced or metastatic urothelial cancer (la/mUC), as current treatments may be limited by tolerability
 - Current antibody-drug conjugates (ADCs) have high rates of discontinuation due to treatment-related events[1]
- Bicycle® molecules are an innovative therapeutic class under clinical evaluation that offer the manufacturing and pharmacokinetic (PK) properties of a small molecule with the high binding specificity of a biologic,[2-4] making them ideally suited for the targeted delivery of a range of payloads, such as cytotoxins, to solid tumors without the potential adverse events associated with monoclonal antibodies
- Zelenectide pevedotin (zele, formerly BT8009) is a highly selective Bicycle® Drug Conjugate (BDC®) targeting Nectin-4, a protein overexpressed in metastatic urothelial carcinoma (mUC; **Figure 1**)[5-9]
 - Zele has a lower molecular weight (4.2 kDa) and a differentiated PK profile versus available ADCs, with potential to rapidly penetrate tumors and minimize healthy tissue exposure[5,6,10]
- Prior results from the Phase 1/2 Duravelo-1 study (NCT04561362) evaluating zele in patients with mUC who had not previously received enfortumab vedotin (EV) reported no treatment-related Grade ≥3 peripheral neuropathy or skin reactions, suggesting a favorable safety profile in this population[11]

FIGURE 1. ZELENECTIDE PEVEDOTIN STRUCTURE[10]

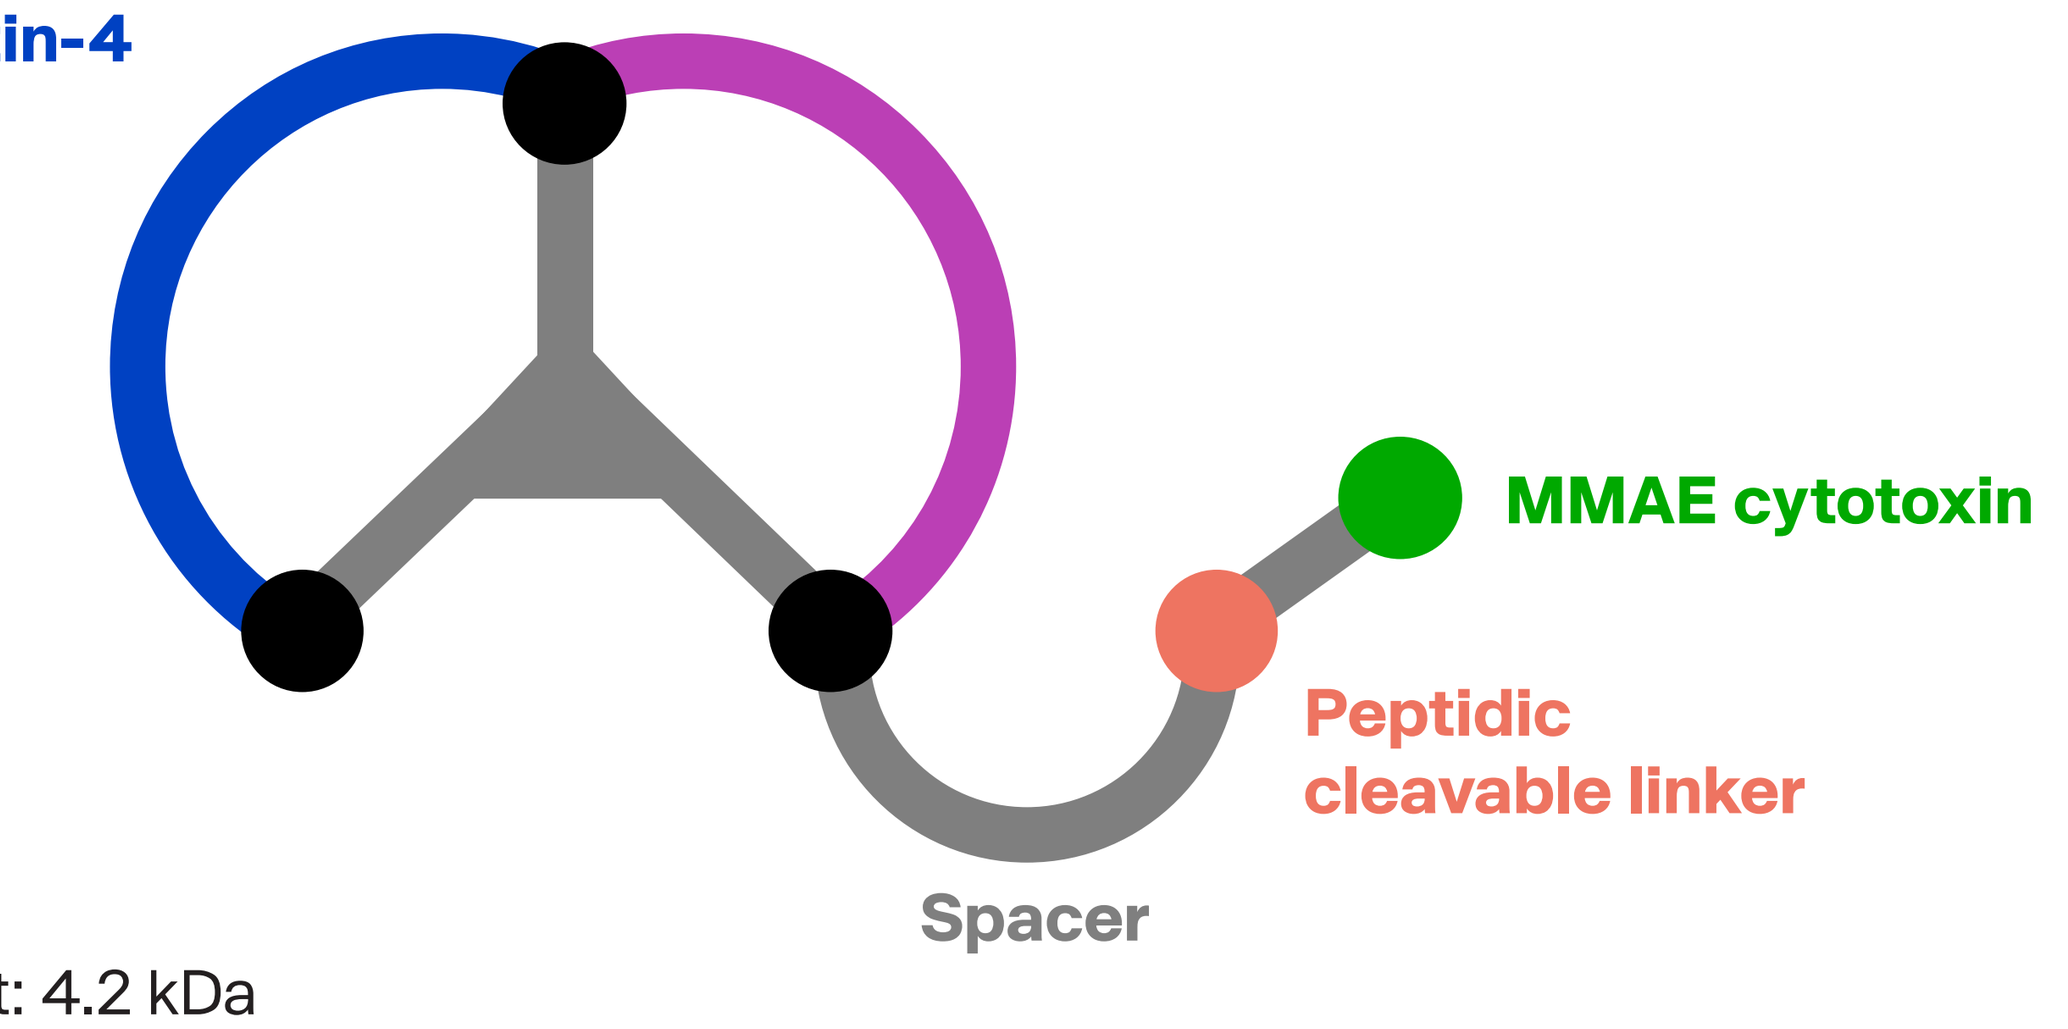
Bicycle® peptide
targeting Nectin-4
MMAE cytotoxin
Peptidic
cleavable linker
Spacer

Molecular weight: 4.2 kDa

OBJECTIVE

- To present updated safety and efficacy results of zele as monotherapy in previously-treated, EV-naïve patients with mUC from the Phase 1/2 Duravelo-1 study (NCT04561362)

METHODS

- Eligible adult patients had recurrent, unresectable mUC, creatinine clearance ≥50 mL/min, prior anti-programmed death-1/programmed death ligand-1 (PD-1/PD-L1) exposure, had disease progression after, or were ineligible for platinum-based chemotherapy, and had received no prior EV
- The primary endpoint for this part of the study was objective response rate (ORR) per RECIST v1.1; secondary endpoints included incidence and severity of treatment-emergent adverse events (TEAEs); duration of response (DoR), clinical benefit rate (CBR), and progression-free survival (PFS); and PK analysis
 - In this study, CBR was defined as the rate of complete response + partial response (PR) + stable disease lasting ≥16 weeks
- All patients with mUC who received the recommended Phase 2 dose of 5 mg/m^2 zele monotherapy once weekly across dose escalation and expansion phases were included for analysis

RESULTS

- As of August 1, 2025, 53 patients were enrolled; the median age was 63 years (range 25–84) and patients had a median of 2 prior lines of therapy (**Table 1**)

TABLE 1. PATIENT DEMOGRAPHICS AND CLINICAL CHARACTERISTICS

Characteristic	Patients (N=53)
Median age, years (range)	**63.0 (25–84)**
Sex, n (%)	
Male	40 (75.5)
Race, n (%)	
White	33 (62.3)
Black or African American	0
Other/missing	20 (37.7)
ECOG PS, n (%)	
0	25 (47.2)
1	28 (52.8)
Median prior lines of therapy (range)	2 (1–7)
Prior therapy, n (%)	
Platinum-based therapy	52 (98.1)
Checkpoint inhibitor	51 (96.2)
Taxane	13 (24.5)
Sacituzumab govitecan	7 (13.2)
FGFR inhibitor	1 (1.9)
Metastatic disease sites,[a] n (%)	
Bone	18 (34.0)
Liver	15 (28.3)
Lung	28 (52.8)

[a]Excludes sites other than bone, liver, and lung. Patients may have metastatic disease in more than one location.

EFFICACY

- Confirmed ORR was 32.1% (n=17) and disease control rate (DCR) was 66%, including 2 confirmed complete responses (**Table 2**)
 - Three additional patients had unconfirmed PR for a total ORR of 38%
 - Among patients who received a prior taxane, 4 of 13 achieved an objective response (31%)
- Median time on treatment was 4.4 months with a maximum time on treatment of 24.9 months
- Median follow-up time was 9.9 months (range 0.5–43.9)
- Median DoR was 10.0 months (95% CI 5.3–16.6) among patients with confirmed responses
- Median PFS was 5.4 months (95% CI 2.3–7.1; **Figure 2**)

TABLE 2. CONFIRMED BEST OVERALL RESPONSE

Best Overall Response	Patients (N=53)
Confirmed ORR, n (%) [95% CI]	**17 (32.1)** [19.9–46.3][a]
Complete response	2 (3.8)
Partial response	15 (28.3)[a]
Stable disease	18 (34.0)
Progressive disease	17 (32.1)
Non-evaluable	1 (1.9)
Median time to confirmed objective response, months (range)	1.8 (0.8–3.7)
DCR, % (95% CI)	66.0 (51.7–78.5)
CBR, %[b] (95% CI)	60.4 (46.0–73.5)
DoR ≥9 months amongst confirmed responses, % (95% CI)	51.8 (26.2–72.4)

[a]3 additional patients had unconfirmed partial responses for a total ORR of 38%; all patients have discontinued treatment. [b]CBR defined as cCR + cPR + SD ≥16 weeks.

FIGURE 2. PFS IN EV-NAÏVE PATIENTS WITH mUC TREATED WITH ZELE (N=53)

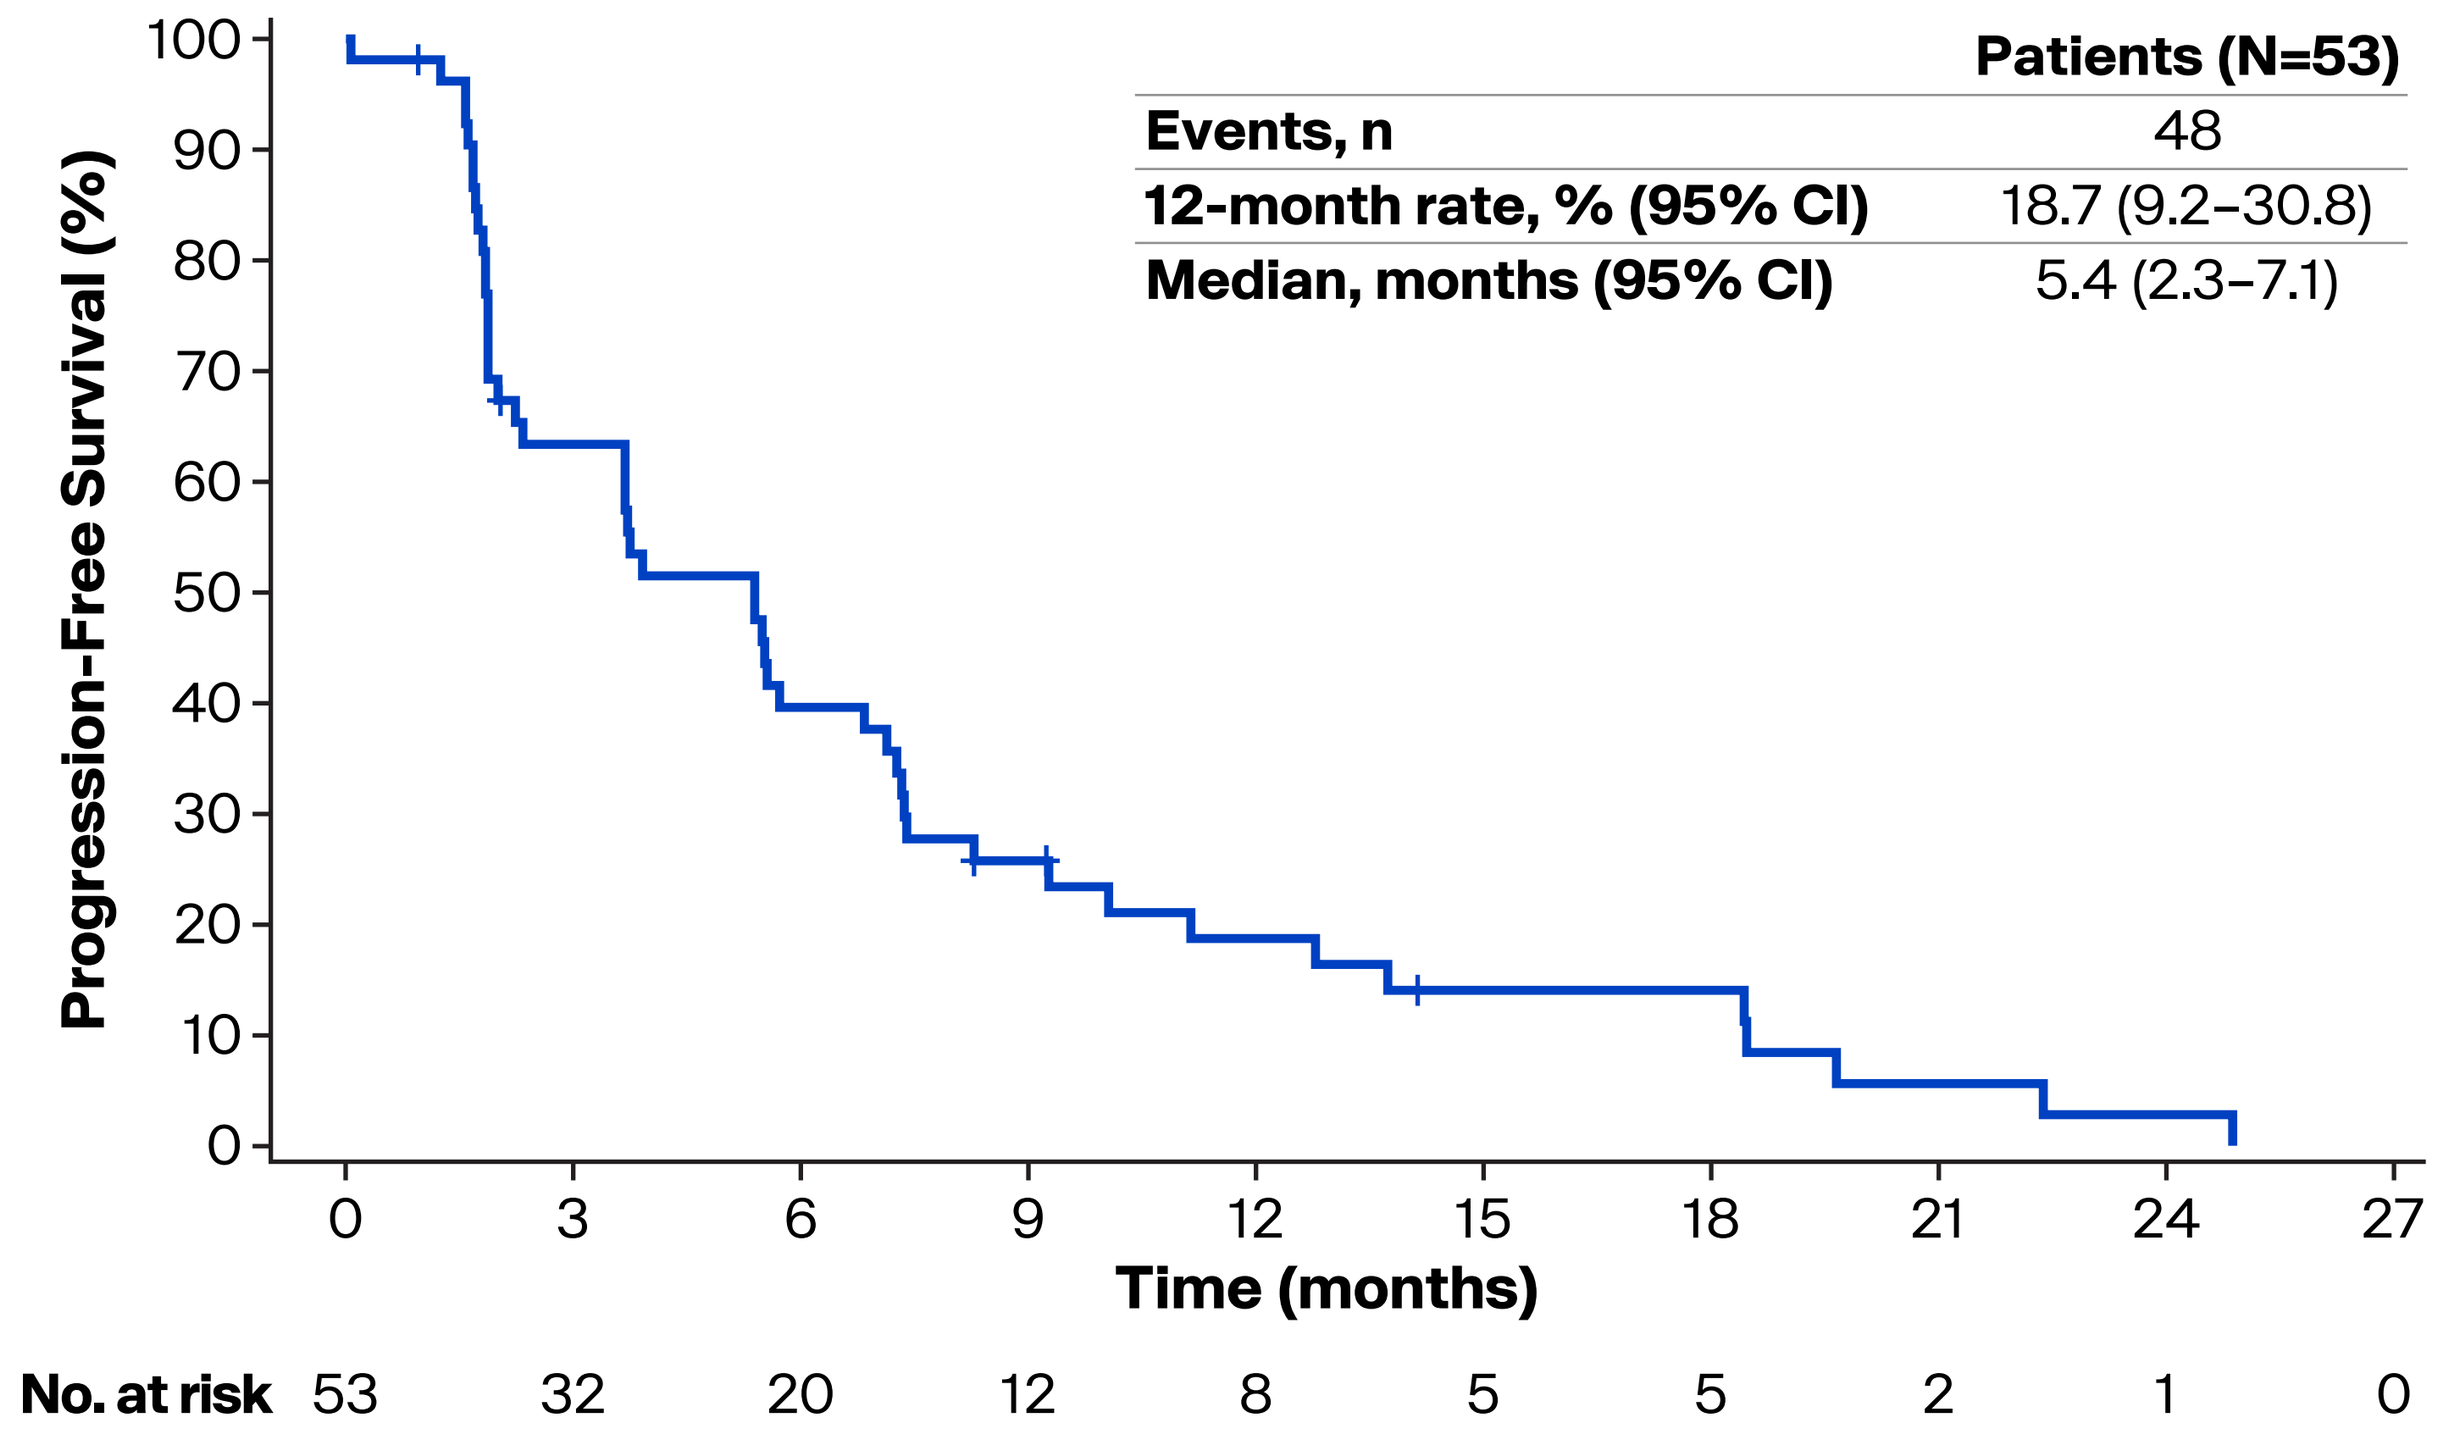
Progression-Free Survival (%)
100
90
80
70
60
50
40
30
20
10
0
Patients (N=53)
Events, n 48
12-month rate, % (95% CI) 18.7 (9.2–30.8)
Median, months (95% CI) 5.4 (2.3–7.1)
0 3 6 9 12 15 18 21 24 27
Time (months)
No. at risk 53 32 20 12 8 5 5 2 1 0

SAFETY

- The most common treatment-related adverse events (TRAEs) were nausea (36%), asthenia (28%), and diarrhea (28%; **Table 3**)
- No Grade 4 or Grade 5 TRAEs were reported
 - One Grade 5 event of hemorrhage/hemorrhagic shock with deterioration occurred and was deemed unrelated to zele
- TRAEs led to dose discontinuation in only 4% of patients (**Table 3**)
 - Only one event of peripheral neuropathy led to treatment discontinuation

TABLE 3. SAFETY SUMMARY OF EV-NAÏVE PATIENTS WITH mUC TREATED WITH ZELE (N=53)

Category, n (%)	Patients (N=53)			
TEAEs	53 (100.0)			
Grade ≥3	31 (58.5)			
TRAEs	47 (88.7)			
Grade ≥3	13 (24.5)			
TRSAEs	5 (9.4)			
Grade ≥3	3 (5.7)			
Dose modifications				
TRAEs leading to dose reduction	14 (26.4)			
TRAEs leading to dose interruption	25 (47.2)			
TRAEs leading to discontinuation	2 (3.8)			
TRAEs occurring in ≥20% or Grade ≥3 TRAEs occurring in ≥5% of patients	**Any Grade**	**Grade 1**	**Grade 2**	**Grade 3**
Nausea[a]	19 (35.8)	14 (26.4)	4 (7.5)	1 (1.9)
Asthenia	15 (28.3)	7 (13.2)	3 (5.7)	5 (9.4)
Diarrhea	15 (28.3)	13 (24.5)	2 (3.8)	0
Alopecia	13 (24.5)	7 (13.2)	6 (11.3)	0
Fatigue	12 (22.6)	5 (9.4)	7 (13.2)	0
Anemia	11 (20.8)	4 (7.5)	5 (9.4)	2 (3.8)
Neuropathy peripheral	11 (20.8)	6 (11.3)	5 (9.4)	0

[a]Prophylactic anti-emetics are prohibited during Cycle 1 of the dose escalation, and use of anti-emetics associated with QT prolongation was prohibited during the study.

- There were no Grade ≥3 TRAEs of peripheral neuropathy (**Table 4**)
- The majority of patients who entered the study with Grade 1 peripheral neuropathy did not develop worsening neuropathy
- The majority of skin reactions were Grade 1 and no events of Stevens-Johnson syndrome, or toxic epidermal necrolysis were reported

TABLE 4. TRAEs OF CLINICAL INTEREST RELATED TO TREATMENT WITH ZELE

	Patients (N=53)					
TRAE of clinical interest, n (%)[a]	**All Grades**	**Grade 1**	**Grade 2**	**Grade 3**	**Grade 4**	**Grade 5**
Peripheral neuropathy[b]	25 (47.2)	13 (24.5)	12 (22.6)	0	0	0
Sensory events	25 (47.2)	13 (24.5)	12 (22.6)	0	0	0
Motor events	0	0	0	0	0	0
Skin reactions[c]	9 (17.0)	7 (13.2)	1 (1.9)	1 (1.9)	0	0
Eye disorders[d]	7 (13.2)	4 (7.5)	3 (5.7)	0	0	0
Hyperglycemia[e]	3 (5.7)	2 (3.8)	0	1 (1.9)	0	0

[a]Patients can have multiple PT within a category. [b]Standardized Medical Dictionary for Regulatory Activities (MedDRA Queries (SMQ) [broad]. [c]Includes the MedDRA SMQ [broad] for Severe Cutaneous Adverse Reactions (SCAR) and the following high-level terms (HLTs): "bullous conditions," "dermatitis and eczema," "rashes, eruptions and exanthems NEC," "erythemas," and "dermatitis ascribed to specific agent"; [d]SOC of eye disorders. [e]Preferred term.

CONCLUSIONS

- This Phase 1/2 study of zele monotherapy at 5 mg/m^2 once weekly continued to demonstrate an encouraging efficacy profile and a generally well-tolerated safety profile, with the potential to differentiate from ADCs among EV-naïve patients with la/mUC
 - Efficacy and safety were preserved in patients with exposure to taxanes
 - No Grade ≥3 treatment-related peripheral neuropathy has been reported
 - Treatment discontinuation resulting from zele-related AEs, including zele-related AEs of clinical interest, was rare
- A Phase 2 study of zele in patients with la/mUC (NCT06225596; Duravelo-2) recently completed an interim analysis for dose optimization

Please visit more presentations about zele at ASCO 2026

- **Abstract #4516:** Loriot Y, et al. Interim Analysis Results from Duravelo-2: Zelenectide Pevedotin + Pembrolizumab in Patients With Previously Untreated Locally Advanced/Metastatic Urothelial Carcinoma. **Rapid Oral presented Monday, June 1, 2026 starting at 8:00 am CDT**
- **Abstract #4564:** Duran I, et al. Zelenectide pevedotin (BT8009) plus pembrolizumab in 1L cisplatin-ineligible locally advanced/metastatic urothelial carcinoma: Update on Duravelo-1 B7
- **Abstract #4566:** Petrylak D, et al. Interim analysis results from Duravelo-2: zelenectide pevedotin (BT8009) in patients with previously treated locally advanced/metastatic urothelial carcinoma
- **Abstract #4567:** Lu Y, et al. Identify optimized dosage for zelenectide pevedotin in locally advanced/metastatic urothelial carcinoma using quantitative analyses

ABBREVIATIONS

ADC, antibody drug conjugate; ASCO, American Society of Clinical Oncology; BDC, Bicycle Drug Conjugate; CBR, clinical benefit rate; cCR, confirmed complete response; cPR, confirmed partial response; CR, complete response; DCR, disease control rate; DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EV, enfortumab vedotin; FGFR, fibroblast growth factor receptor; HLT, high-level term; la/mUC, locally advanced or metastatic urothelial carcinoma; MedDRA, Medical Dictionary for Regulatory Activities; MMAE, monomethyl auristatin E; mUC, metastatic urothelial carcinoma; ORR, objective response rate; PD-1/PD-L1, programmed death-1/programmed death ligand-1; PFS, progression-free survival; PK, pharmacokinetic; PR, partial response; PT, preferred term; QT, electrocardiographic QT interval; RECIST, Response Evaluation Criteria in Solid Tumors; SCAR, severe cutaneous adverse reactions; SD, stable disease; SMQ, standardized MedDRA queries; SOC, system organ class; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event; TRSAE, treatment-related serious adverse event; zele, zelenectide pevedotin.

REFERENCES

1. Powles T, et al. *N Engl J Med.* 2021;384:1125–1135.
2. Eder M, et al. *Cancer Res.* 2019;79(4):841–852.
3. Mudd GE, et al. *J Med Chem.* 2020;63(8):4107–4116.
4. Walsh SJ, et al. *Cancer Res.* 2024;84(6_Suppl):5807–5807.
5. Rigby M, et al. *Mol Cancer Ther.* 2022;21(12):1747–1756.
6. Mudd G, et al. *J Med Chem.* 2022;65(21):14337–14347.
7. Duan X, et al. *Clin Cancer Res.* 2023;29(17):3395–3407.
8. Zhou W, et al. *Mol Cancer Ther.* 2023;22(8):913–925.
9. Challita-Eid PM, et al. *Cancer Res.* 2016;76(10):3003–3013.
10. Kaufman, et al. ESMO 2025. Abstract 628TiP.
11. Reig Torras O, et al. *Ann Oncol.* 2024;35:S515–S516.

ACKNOWLEDGMENTS

Bicycle Therapeutics would like to thank the patients and their families for participating in this study. Without their contributions, this study would not have been possible.

Medical writing support was provided by Danielle Fanslow, PhD, CMPP, of Second City Science, funded by BicycleTx Ltd.

Funding: This study was sponsored by BicycleTx Ltd.

Presenting author: Oscar Reig Torras, MD

Presenting author COI disclosures: ORT reports the following relationships: Honoraria – BMS, Ipsen, Pfizer; Consulting or Advisory Role – Eisai; Speakers' Bureau – Bayer, BMS, Ipsen, Janssen-Cilag, Pfizer, Sanofi; Travel, Accommodations, Expenses – Ipsen.